Poster ID 094

Arimoclomol Safety Profile in the Treatment of NPC in a Real-World Setting: Long-Term Data From an Expanded Access Program in the United States

Can Ficicioglu[1], Elizabeth M. Berry-Kravis[2], Walla Al-Hertani[3,4], Raymond Wang[4], Marc Patterson[5], Loren Pena[6], Kristina Julich[7], Damara Ortiz[8], Paula Schleifer[9], Paul Hillman[10], Caroline Hastings[11], Ronan O'Reilly[12], Christine Dali[12], Daniel Gallo[12]

[1]Children's Hospital of Philadelphia, PA, USA, [2]Rush University Medical Center, IL, USA, [3]Boston Children's Hospital, Harvard Medical School, MA, USA, [4]Children's Hospital of Orange County, CA, USA, [5]Mayo Clinic Children's Center, MN, USA, [6]Cincinnati Children's Hospital, OH, USA, [7]Dell Medical School, The University of Texas at Austin, TX, USA, [8]UPMC Children's Hospital, PA, USA, [9]Nicklaus Children's Hospital, FL, USA, [10]Department of Pediatrics, Division of Medical Genetics, McGovern Medical School, University of Texas Health Science Center at Houston (UTHealth Houston) and Children's Memorial Hermann Hospital, Houston, TX, [11]UCSF Benioff Children's Hospitals, CA, USA, [12]Zevra Therapeutics, Celebration, FL, USA

WORLD*Symposium* February 3-7, San Diego, California

BACKGROUND AND OBJECTIVE

- Niemann-Pick disease type C (NPC) is an ultra-rare, progressive neurodegenerative lysosomal storage disease with persisting unmet medical need.
- Arimoclomol, an orally available small molecule, is the first FDA-approved treatment for NPC when used in combination with miglustat.
- The US arimoclomol Expanded Access Program (EAP), initiated in June 2020 (NCT04316637), provides access to eligible NPC patients.
- Here we present safety data from pediatric and adult NPC patients treated longitudinally in the US EAP with arimoclomol.
- Optional real-world data (RWD) collected in an ongoing protocol-driven EAP was initiated to expand the understanding of NPC, including populations not previously studied in randomized clinical trials, for those patients consenting to collection is presented on poster 032.

METHODS

- The US EAP is an ongoing, prospective real-world study designed to provide expanded access to arimoclomol for NPC patients who were not eligible for or unable to participate in clinical trials.
- The 15-site protocol driven US EAP was designed to include broad geographic site representation, telemedicine consultations, and local laboratory testing.
- Routine clinical care, including miglustat, was maintained throughout the US EAP.
- Adverse event (AE) experience, including serious adverse events (SAEs) and reasons for withdrawal were captured.
- All adverse event experience was summarized and is presented as event counts and percentages of patients.
- Results are presented with the current data as of May 8, 2024.

METHODS AND RESULTS

Table 1: EAP Eligibility and Ineligibility Criteria*

Eligibility Criteria
Confirmed NPC diagnosis and at least 1 neurological symptom
Age ≥2 years
Permanent US resident
If taking miglustat (Zavesca®), the patient must have been on the target dose for the past 6 weeks
Sexually active females of childbearing potential must agree to use highly effective contraception
Confirmed negative pregnancy test for sexually active females
Sexually active male patients with female partners of child-bearing potential agree to use a condom in addition to birth control used by their partners
If history of seizures, the condition must be adequately controlled, i.e., seizure activity must be stable, and patient must be on stable dose and regimen of antiepileptic Medication during 1 month prior to screening
Written informed consent (patient or parent/guardian)
For participants in CT-ORZY-NPC-002 clinical trial: The treating physician confirms a positive benefit risk assessment for the patient at the end of the trial

Ineligibility Criteria
Severe liver disease
Kidney disease
Known or suspected allergy or intolerance to arimoclomol
Pregnancy, planning to become pregnant (during the EAP) or currently breastfeeding
Plans for treatment with other investigational drug during the EAP or in the 4 weeks prior to arimoclomol treatment start
The patient is both eligible and able to participate in or is currently participating in an active interventional clinical trial within the indication
The patient, in the opinion of the clinician, is unable to comply with the treatment or has a medical condition that would potentially increase the risk to the patient by participation
The patient has a medical condition which hinders the clinician's assessment of arimoclomol safety and efficacy (eg, certain epileptic conditions or severe sudden loss of consciousness)

*- Eligibility and ineligibility criteria described are not inclusive of all criteria. Refer to NCT04316637 for complete criteria

Figure 1: US EAP Spans 15 Sites

14 out of 15 sites have enrolled patients and 13 sites are active

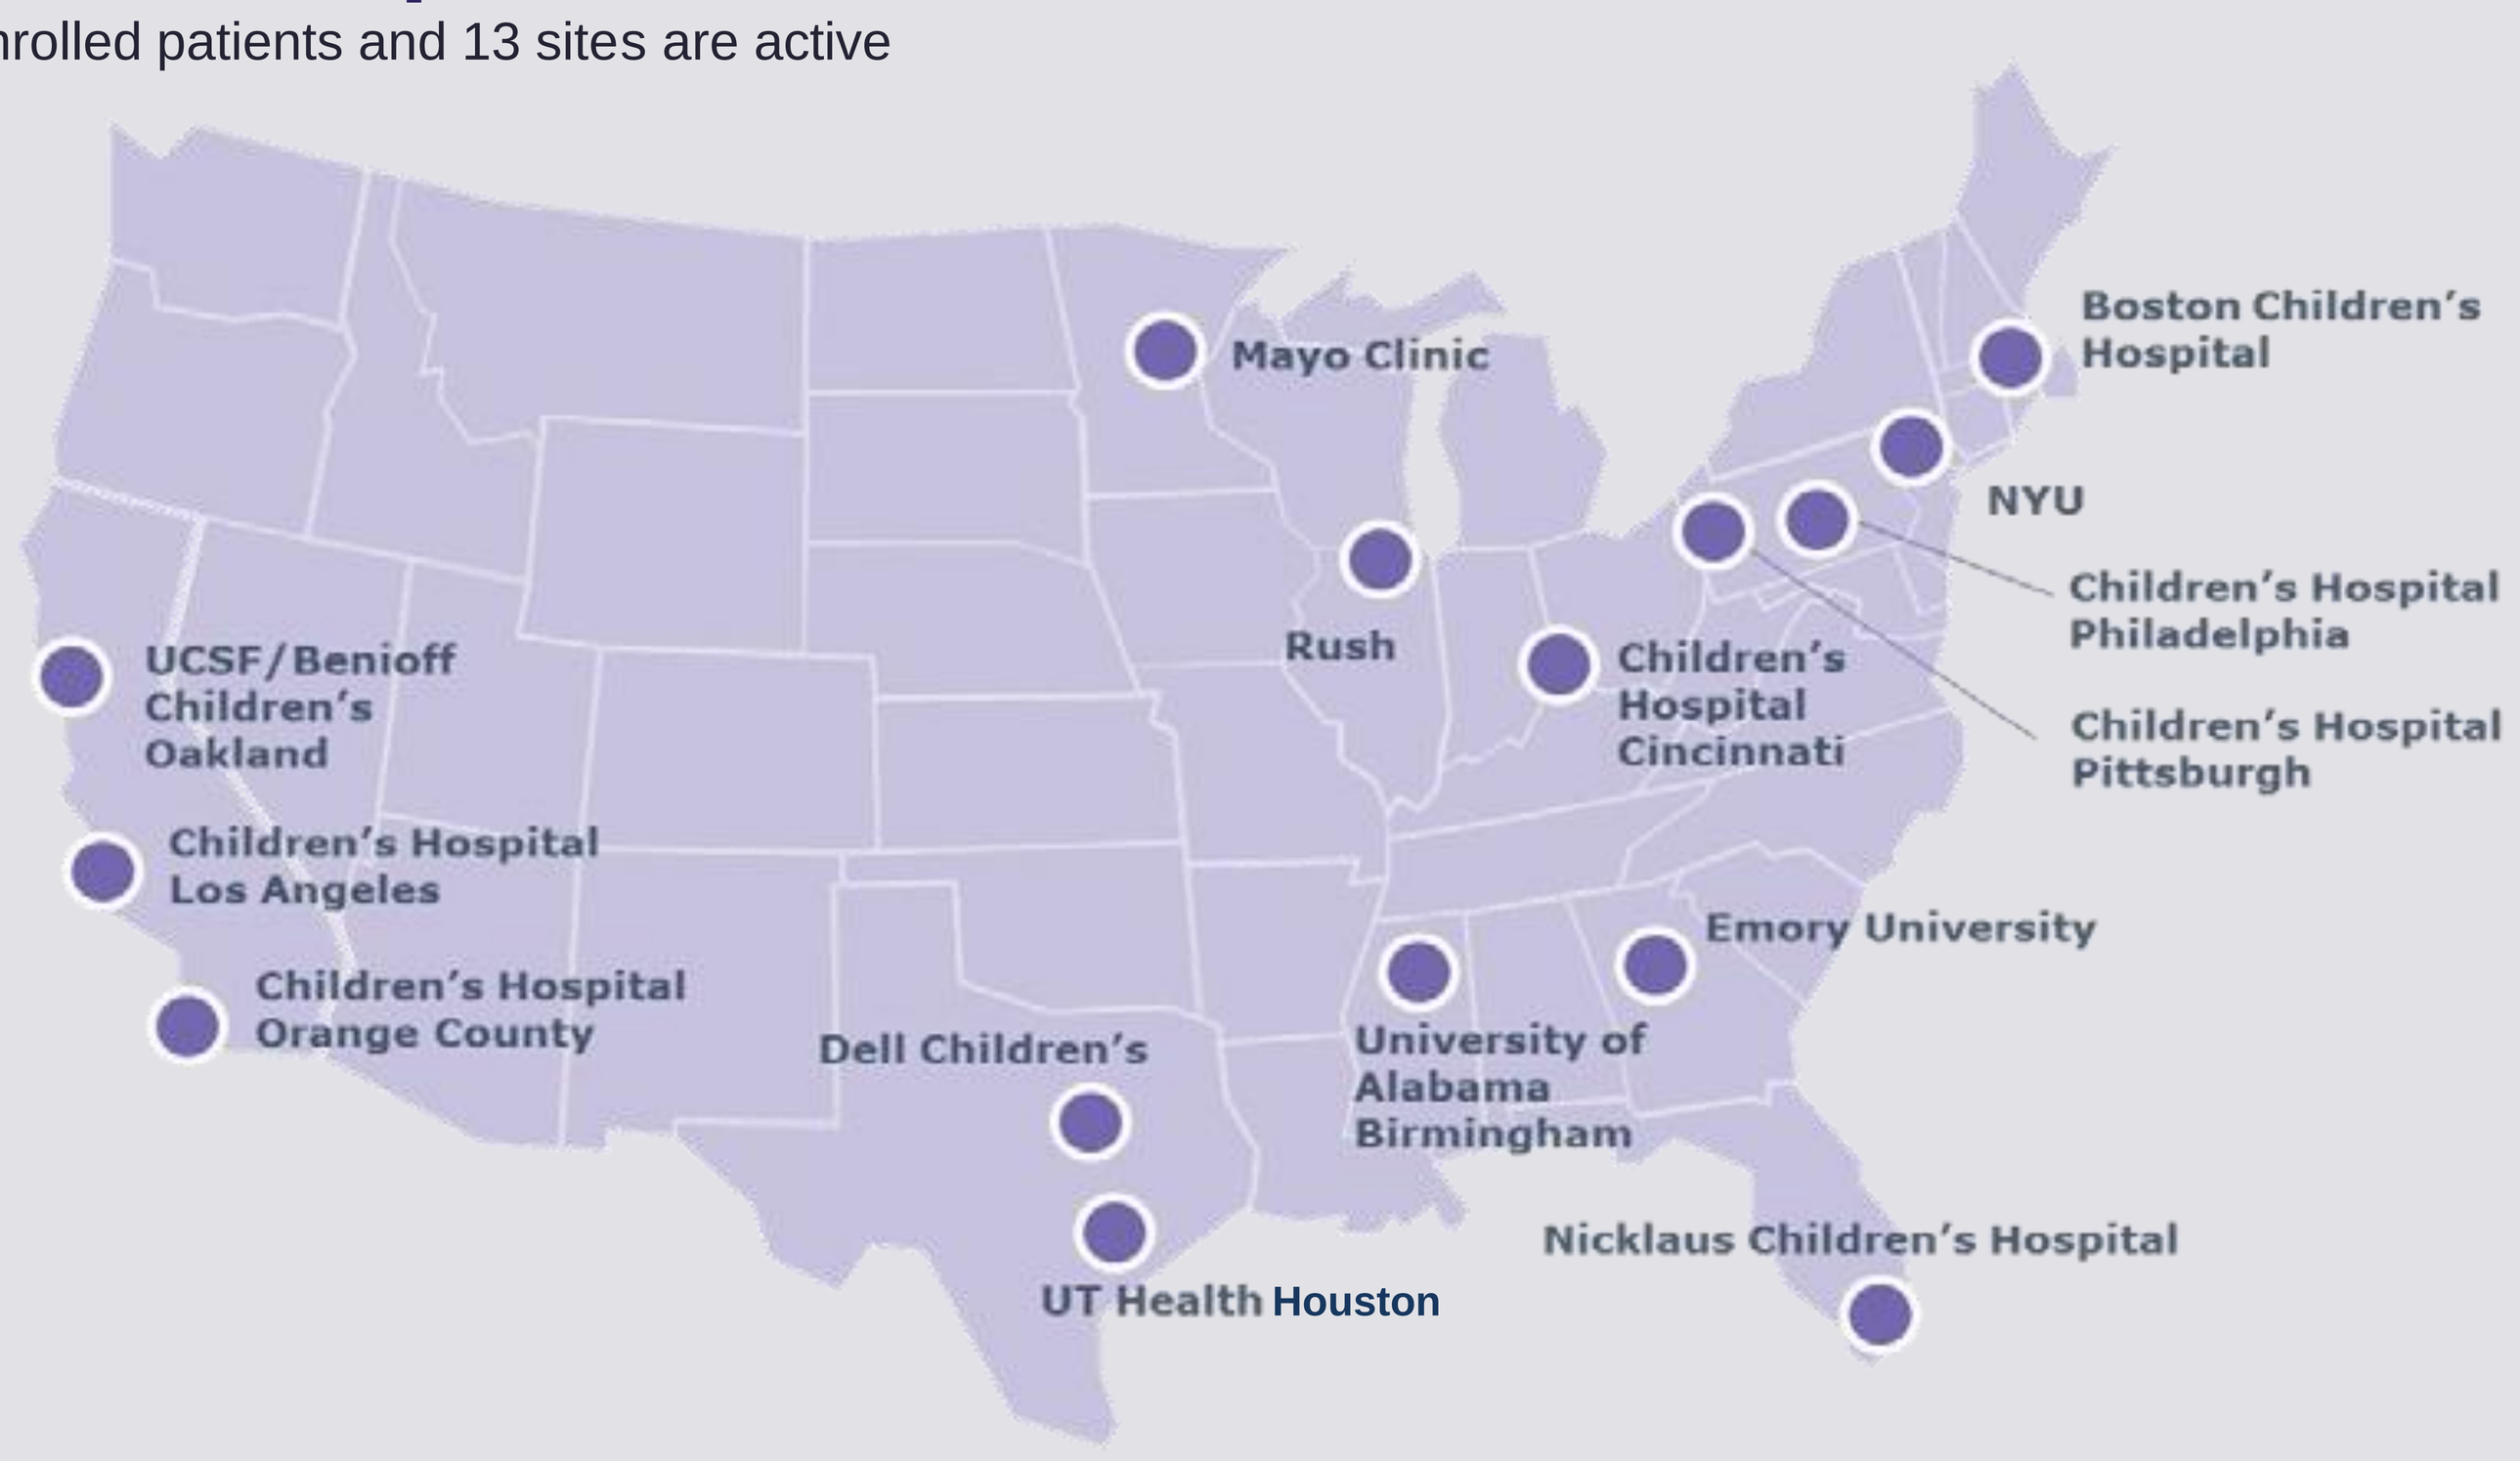

Mayo Clinic
Boston Children's Hospital
NYU
Children's Hospital Philadelphia
Children's Hospital Pittsburgh
Rush
Children's Hospital Cincinnati
UCSF/Benioff Children's Oakland
Children's Hospital Los Angeles
Children's Hospital Orange County
Dell Children's
University of Alabama Birmingham
Emory University
UT Health Houston
Nicklaus Children's Hospital

RESULTS

- 94 participants enrolled in the US EAP and were included in the safety analysis (**Table 2**).
- A total of 201 adverse events were reported for 62 patients (66%) while adverse events were not reported for 32 patients (34%) (**Table 3**).
- Pneumonia (14 events, 11.7% of patients), corona virus infection (13 events, 11.7% of patients), and diarrhea (8 events, 7.4% of patients) were the most commonly reported adverse events (**Table 3**).
- The adverse event profile was similar for patients with and without miglustat use at any point during the US EAP (**Table 3**).
- 70 serious adverse events were reported for 28 Patients of which 9 patients (9.6%) had events with a fatal outcome (**Tables 3 & 4**). No serious adverse events were determined to be related to arimoclomol.
- 23 patients (25%) have discontinued arimoclomol treatment (**Table 4**).

Table 2: US EAP Safety Reporting Patient Characteristics

	Patients Initiated to Treatment	Arimoclomol	Arimoclomol + Miglustat as Part of Routine Clinical Care
Number of Participants, n(%)	94 (100 %)	32 (34%)	62 (66%)
Age at Registration (Years)			
Mean (SD)	20.2 (12.9)	21.9 (14.62)	19.3 (11.93)
Median (Range)	18 (2 – 66)	18.0 (2 – 66)	19.0 (2 – 43)
Arimoclomol Treatment Duration (Months)			
Mean (SD)	25.0 (14.89)	19.68 (15.82)	27.75 (13.73)
Median (Range)	29.5 (0.03 – 46.87)	14.43 (0.03 – 46.87)	31.87 (2.77 – 46.0)

Table 2: US EAP patient characteristics & demographics for the 94 patients included in the safety analysis.

Table 3: US EAP Adverse Event (AE) Experience

	Patients treated with Arimoclomol (N = 94)	Arimoclomol (N = 32)	Arimoclomol + miglustat as part of routine clinical care (N = 62)
Any, n events (% of patients)	201 (65.9%)	64 (56.3%)	137 (70.9%)
By Seriousness, n events (% of patients)			
Non-Serious AE	131 (51.1%)	43 (50.0%)	88 (51.6%)
Serious AE	70 (29.8%)	21 (18.8%)	49 (35.5%)
Fatal Serious AE	11 (9.6 %)	3 (6.3%)	8 (11.3%)
By Type of Adverse Event (reported by ≥3 patients), n events (% patients)			
Pneumonia	14 (11.7%)	3 (6.3%)	11 (14.4%)
Coronavirus infection	13 (11.7%)	3 (6.3%)	10 (14.4%)
Diarrhea	8 (7.4%)	2 (6.3%)	6 (8.1%)
Urinary Tract Infection	7 (3.2%)	7 (9.4%)	0 (0%)
Fall	6 (6.4%)	1 (3.1%)	5 (8.1%)
Upper Respiratory Tract Infection	6 (4.3%)	4 (6.3%)	2 (3.2%)

Table 3: Safety event reporting overall and by seriousness for the US EAP reported as number of events and % of patients. Type of adverse event summary of the number of events and % of patients experiencing the event. The type of adverse event summary includes only events that were reported for > 5 % of the total population. No serious adverse events were determined to be related to treatment with arimoclomol.

Table 4: Withdrawals From the US EAP

	All Patients (N = 94)
Withdrew from Study or Discontinued Treatment with Arimoclomol, n events (% of patients)	23 (24.5%)
Reason for Withdrawal, n events (% of patients)	
Death*	9 (9.6%)
Pneumonia	4
Acute respiratory failure	2
Choking	1
Coronavirus infection	1
Disease progression	1
Influenza	1
Sepsis	1
Other Reason**	5 (5.3%)
Adverse Event	5 (5.3%)
Lost to Follow-Up	1 (1.1%)
Reason Unknown	3 (3.2%)

Table 4: Reasons for study withdrawal or discontinuation. There were a total of 9 deaths related to events of pneumonia (4), acute respiratory failure (2), choking (1), COVID-19 (1), disease progression (1), influenza (1), and sepsis (1). 2 patients each experienced 2 fatal serious adverse events. *None of the deaths were determined to be related to treatment with arimoclomol. **-other reason included withdrawal of consent (4 patients) and disease progression (1 patient)

CONCLUSIONS

- **The US EAP for arimoclomol for NPC has demonstrated a safety profile consistent with the published clinical trial experience of arimoclomol in NPC.[1]**
- **Over more than 3 years of treatment, arimoclomol was generally well tolerated throughout the study duration.**

References: 1. Mengel E et al. J Inherit Metab Dis. 2021 Nov;44(6):1463-1480

Disclosures: Poster was prepared by Zevra Therapeutics.
Zevra would like to thank the research sites including all study coordinators and staff involved in the US EAP.