Cardiovascular (CV) Safety and Pharmacokinetics (PK) of Serdexmethylphenidate (SDX), a Prodrug of d-Methylphenidate (d-MPH) Compared to Ritalin and Ritalin LA in a Single-Dose Crossover Study in Healthy Volunteers

Poster no. 6

Rene Braeckman, Sven Guenther, Travis C. Mickle, Adam Smith
Zevra Therapeutics, Celebration, FL, USA

BACKGROUND

Serdexmethylphenidate (SDX), a prodrug of d-methylphenidate (d-MPH), is a central nervous system stimulant in clinical development for the treatment of sleep disorders with excessive daytime sleepiness (idiopathic hypersomnia and narcolepsy) and stimulant use disorder.

OBJECTIVE

To assess cardiovascular safety and pharmacokinetics in healthy volunteers after single oral doses of SDX compared to Ritalin.

METHODS

- Phase 1, open-label, single dose, 4-treatment, 4-period, randomized, crossover PK and cardiovascular effect study of single oral doses of SDX, Ritalin®, and Ritalin LA®.
- The study consisted of a screening period, an in-clinic treatment phase, and a follow-up phone call.
- 15 healthy male subjects received single oral doses of 80 mg and 200 mg SDX, 80 mg Ritalin® (immediate release; 2 × 40 mg, 5 hours apart), and 80 mg Ritalin LA® (long acting) with 7 days washout between treatments.
- Multiple blood PK samples and cardiovascular safety parameters (heart rate, blood pressure, ECG) were collected after each administration.
- The PK/PD analysis consisted of an evaluation of the relationship between changes from predose in heart rate and blood pressure, and the systemic exposure of d-MPH at multiple timepoints postdose.

RESULTS

Demographics

- 15 subjects were enrolled and received at least 1 dose of study drug. The mean age was 34.8 years. Most subjects were White (11 [73.3%]) and not Hispanic or Latino (12 [80.0%]).

PK

- d-MPH plasma concentrations after SDX peaked later (6-8 hours) compared to after Ritalin (Figure 1, left panel: 2-4 hours; first of 2 peaks).
- Although the SDX doses were equivalent (80 mg) or 2.5-fold higher (200 mg) on a molar d-MPH basis, the mean maximum exposure to d-MPH (C_{max}) after 80 mg and 200 mg SDX was approximately 25% and 50% lower compared to Ritalin and Ritalin LA, respectively (Table 1 and Figure 3).

Cardiovascular PD

- The mean heart rate change from baseline E_{max} and $AUE_{0\text{-}24}$ was lowest for the SDX 80 mg treatment (Figure 3).
- The mean heart rate change from baseline for the SDX 200 mg treatment was comparable with both Ritalin treatments (Figure 1, right panel and Figure 2).
- The median heart rate TE_{max} for the SDX 200 mg treatment was longer compared to the other treatments.

PK/PD

- Across all treatments, a positive correlation was observed between the mean heart rate change from baseline and the plasma concentration of d-MPH for the first 10 hours (Figures 1 and 2).
- There were no clinically meaningful changes or trends for the PR interval, RR interval QRS duration, QT interval, QTcF interval, and QTcB interval in relation to changes in d-MPH concentration.

Safety

- Most AEs reported were typical for central nervous system stimulants (Table 2).
- Peak heart rate changes after SDX were lower to equal to those after Ritalin.

CONCLUSION

At an SDX dose (200 mg) 2.5-fold higher than the molar-equivalent Ritalin doses (80 mg), the peak exposure to d-MPH occurred later and was lower. SDX (both doses) was generally better tolerated compared to Ritalin. Fewer subjects experienced cardiovascular adverse events after SDX compared to Ritalin. From an overall cardiovascular safety perspective, vital signs after a single oral dose of 200 mg SDX (the highest dose tested) were comparable to Ritalin IR 80 mg and Ritalin LA 80 mg.

Table 1. PK parameters for d-MPH

Parameter	Statistic	SDX 80 mg (N=14)	SDX 200 mg (N=14)	Ritalin IR, 80 mg (N=14)	Ritalin LA, 80 mg (N=14)
C_{max} (ng/mL)	Mean (%CV)	6.06 (22.6)	13.7 (36.2)	27.7 (33.8)	27.6 (23.4)
T_{max} (h)	Median (min,max)	6.00 (4.0, 24.1)	8.00 (4.9, 30.0)	6.50 (1.0, 8.0)	4.00 (1.5, 7.0)
$AUC_{0\text{-}24}$ (h*ng/mL)	Mean (%CV)	89.0 (31.2)	210 (34.4)	220 (31.1)	219 (24.8)
AUC_{inf} (h*ng/mL)	Mean (%CV)	139 (32.1)	331 (41.0)	226 (31.9)	224 (25.6)
$T_{1/2}$ (h)	Mean (%CV)	9.68 (39.2)	7.81 (36.2)	3.90 (16.3)	3.99 (14.7)

Figure 1. Mean d-MPH concentration and heart rate change from baseline

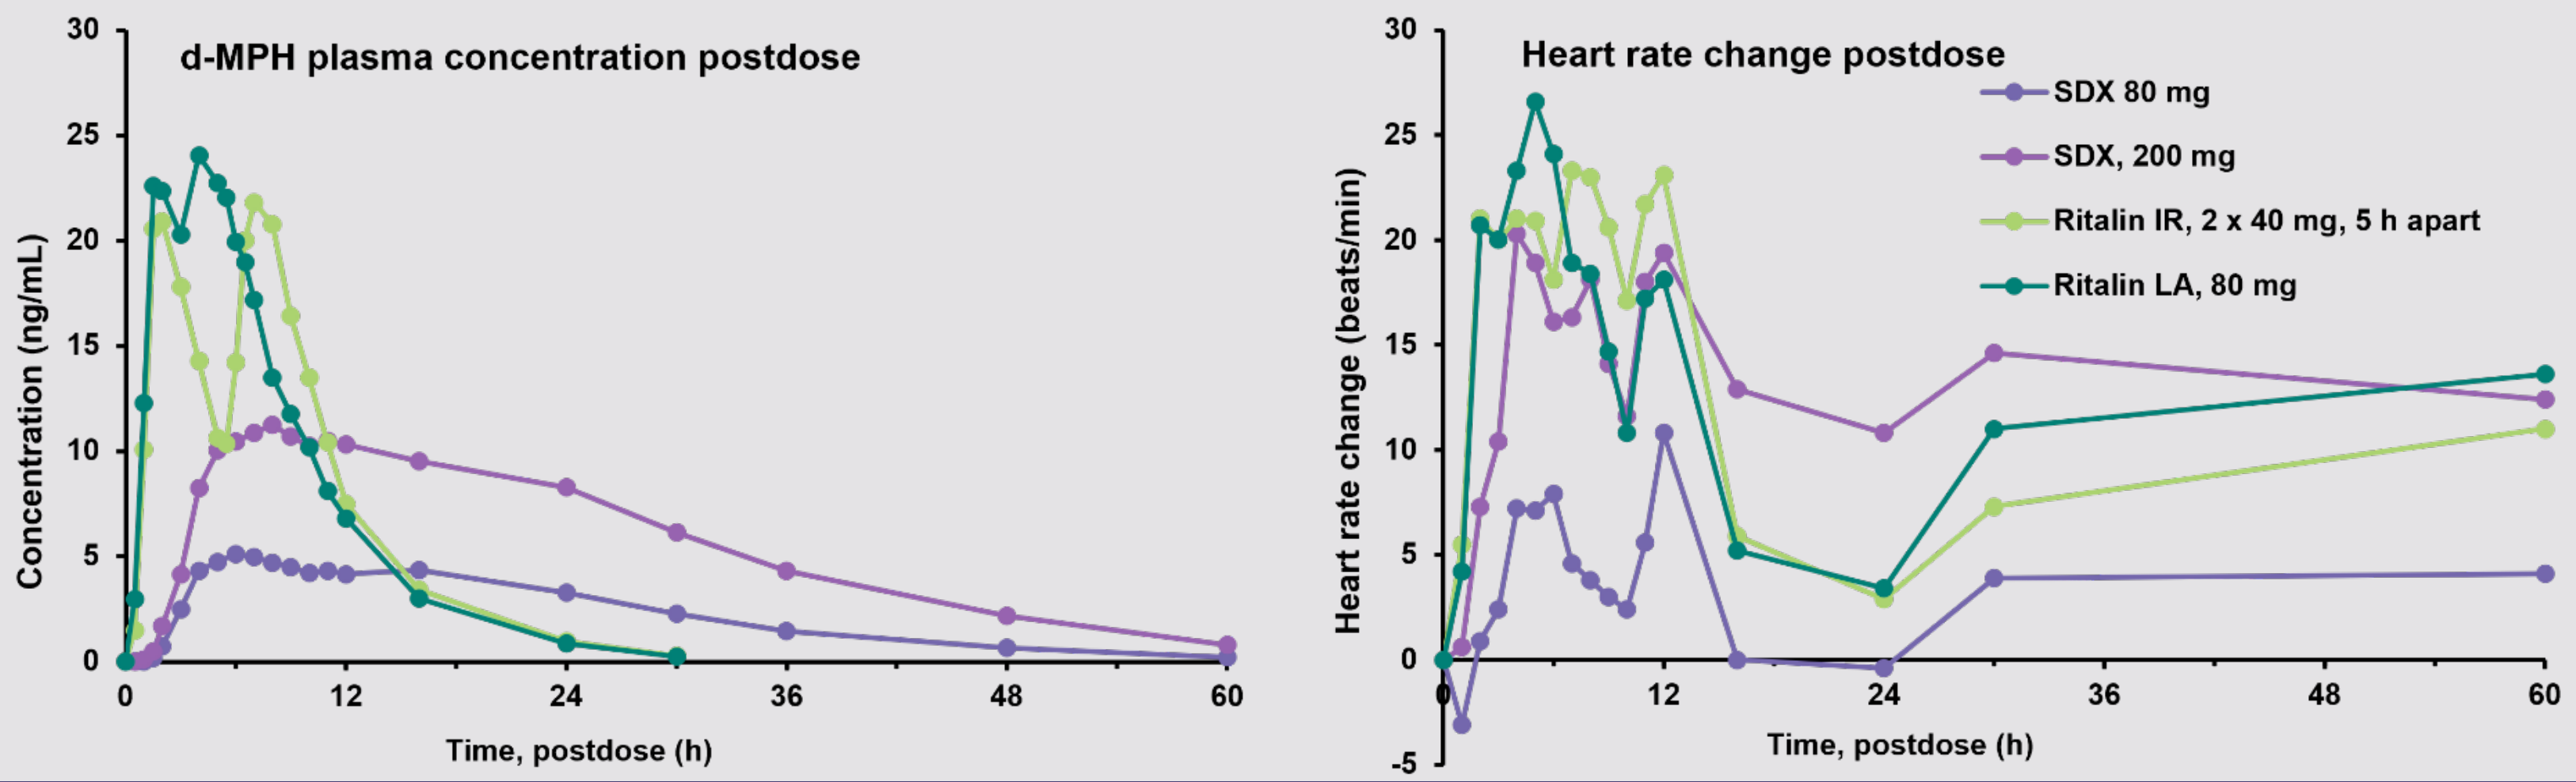
d-MPH plasma concentration postdose
Concentration (ng/mL)
Time, postdose (h)
Heart rate change postdose
Heart rate change (beats/min)
SDX 80 mg
SDX, 200 mg
Ritalin IR, 2 x 40 mg, 5 h apart
Ritalin LA, 80 mg

Figure 2. Mean d-MPH plasma concentration and heart rate change from baseline

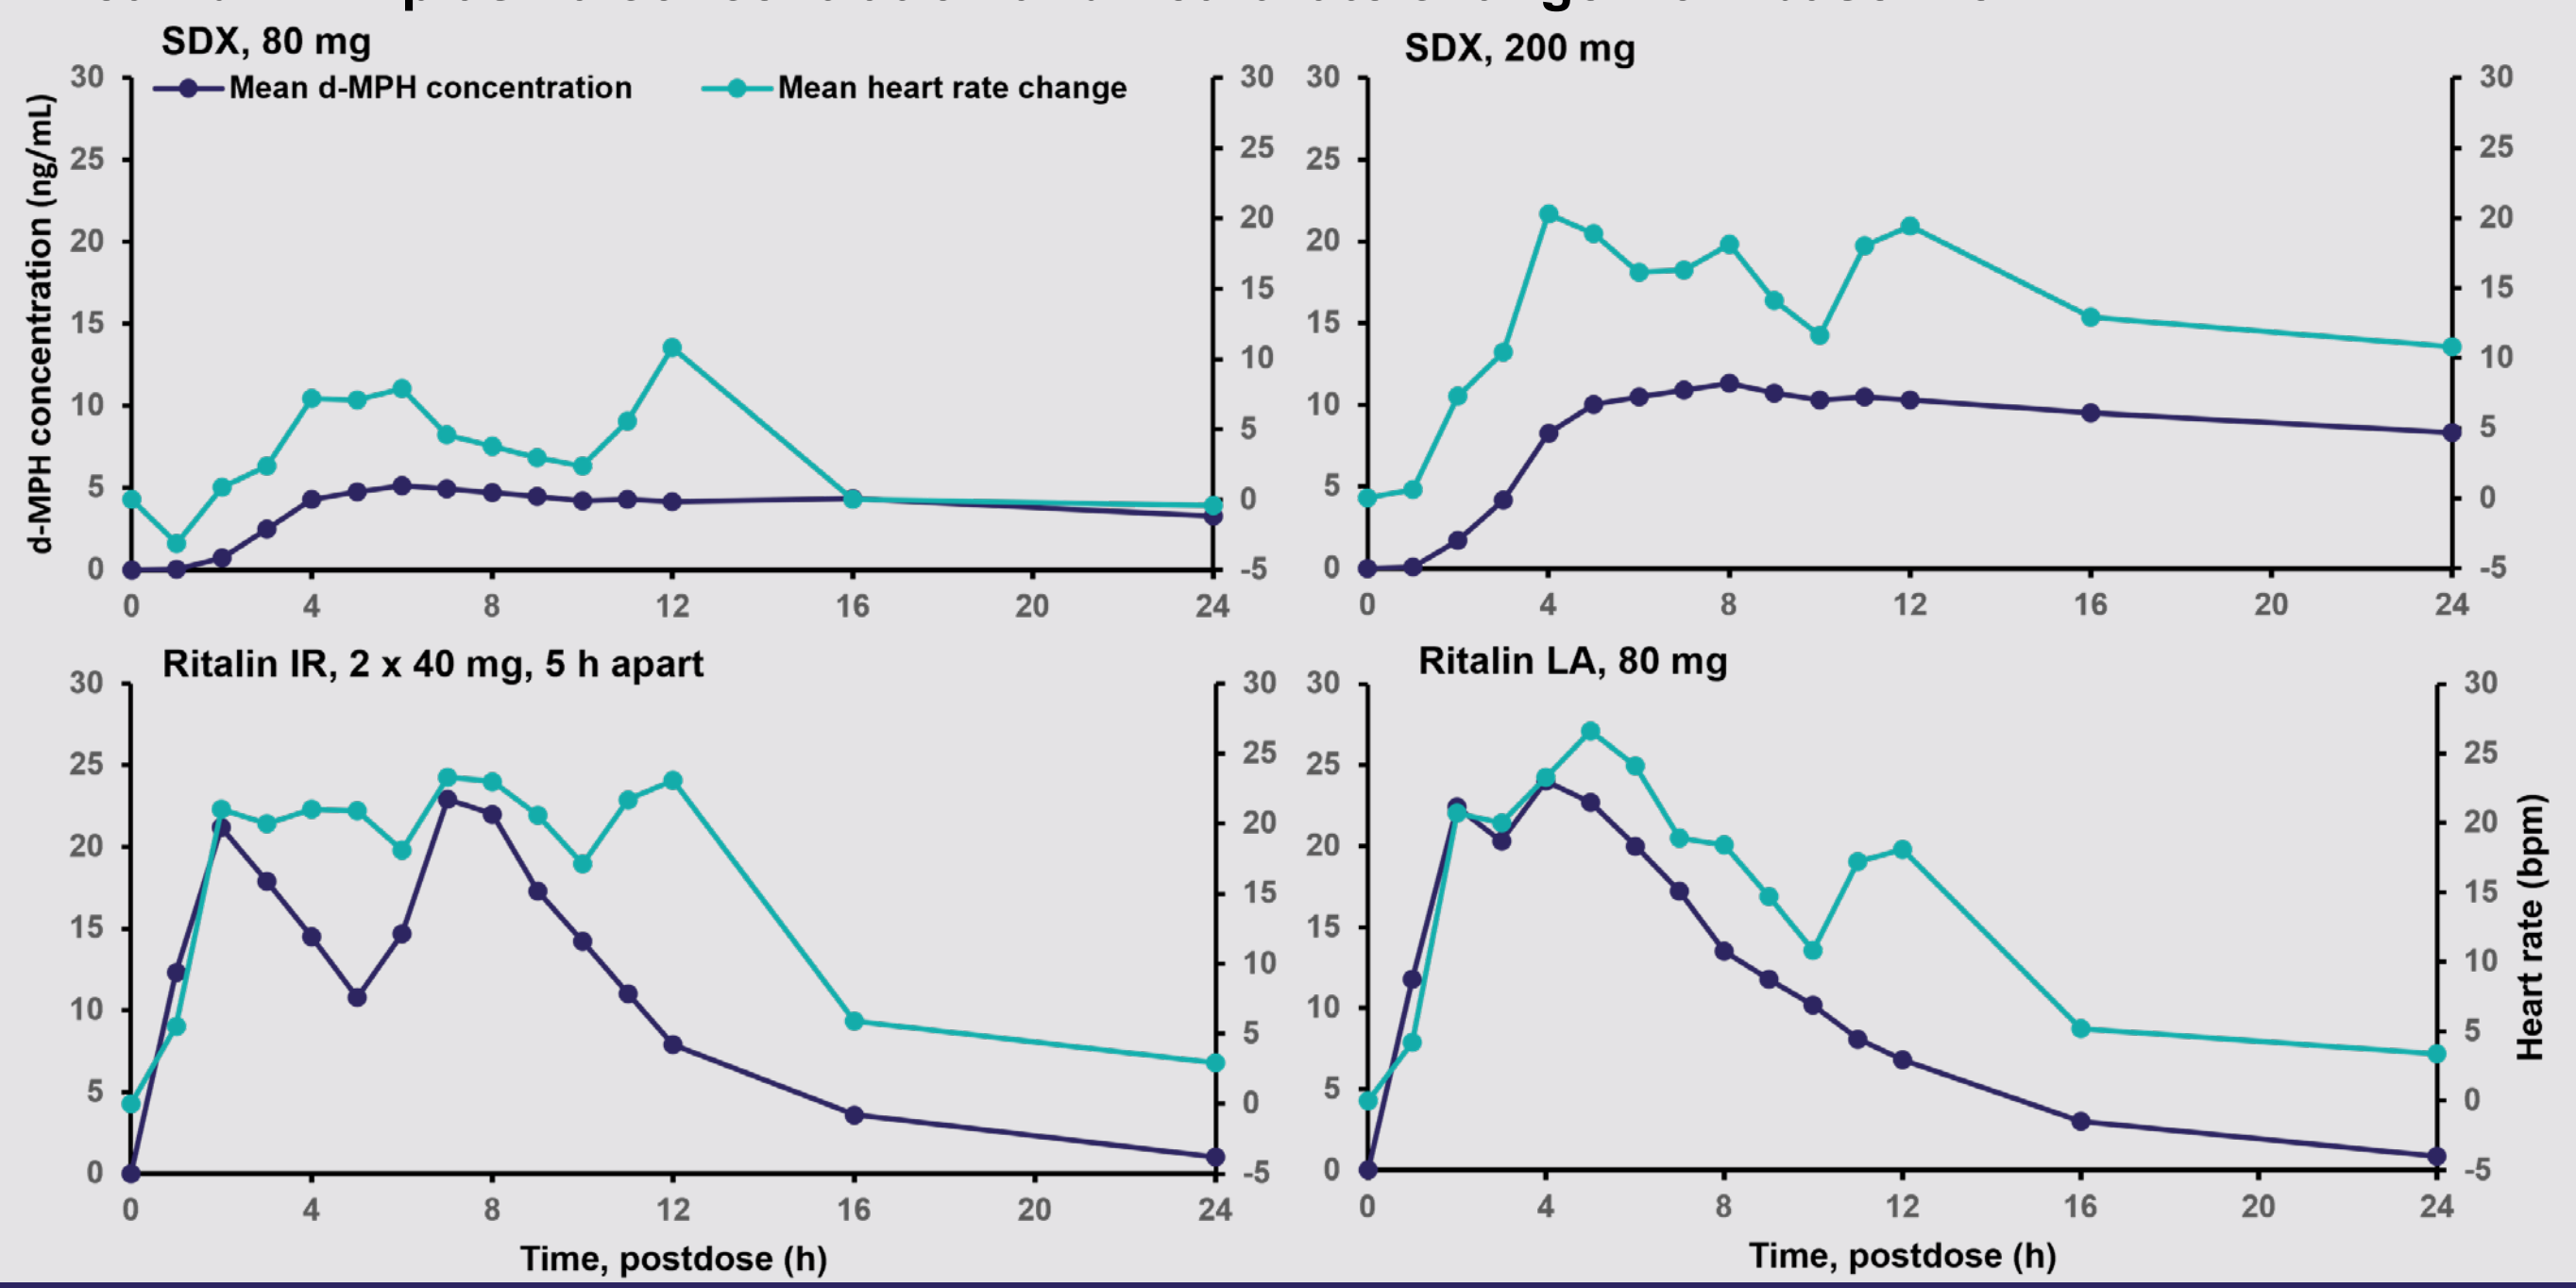
SDX, 80 mg
Mean d-MPH concentration
Mean heart rate change
SDX, 200 mg
Ritalin IR, 2 x 40 mg, 5 h apart
Ritalin LA, 80 mg
d-MPH concentration (ng/mL)
Heart rate (bpm)
Time, postdose (h)

Figure 3. Mean Values of C_{max}, $AUC_{0\text{-}24}$, E_{max} and $AUE_{0\text{-}24}$ by Treatment

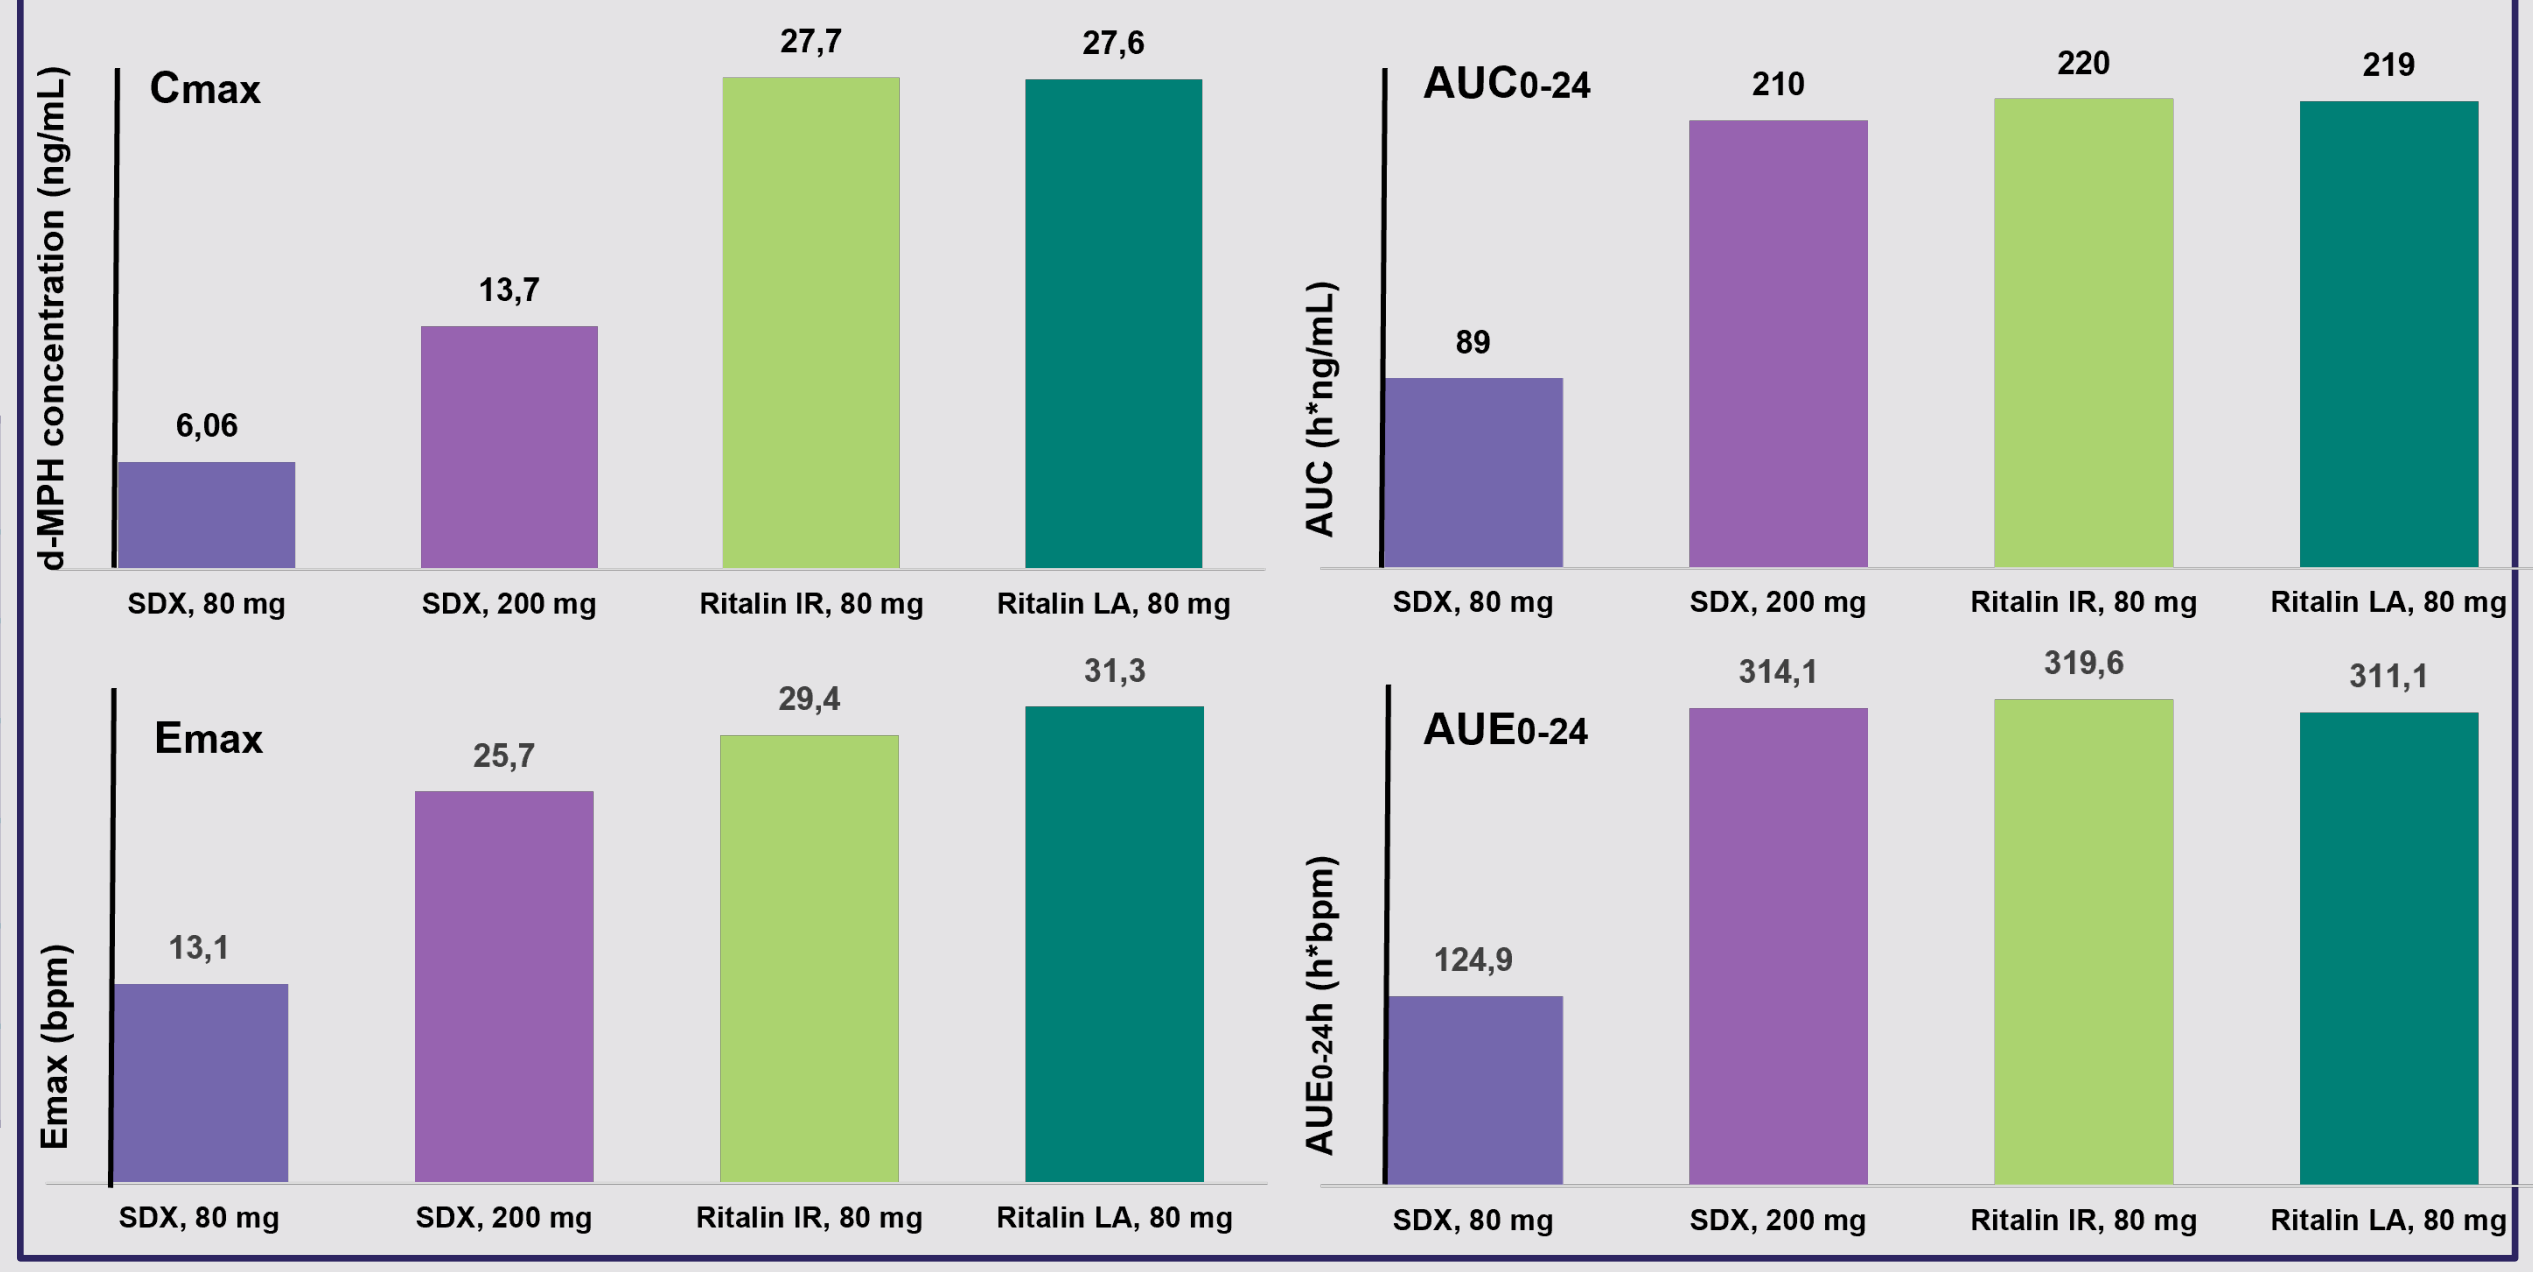
Cmax
d-MPH concentration (ng/mL)
6,06
13,7
27,7
27,6
AUC0-24
AUC (h*ng/mL)
89
210
220
219
Emax
Emax (bpm)
13,1
25,7
29,4
31,3
AUE0-24
AUE0-24h (h*bpm)
124,9
314,1
319,6
311,1
SDX, 80 mg
SDX, 200 mg
Ritalin IR, 80 mg
Ritalin LA, 80 mg

Table 2. Summary of treatment emergent adverse events, cardiovascular events and most frequently reported AEs

	SDX, 80 mg (N=14)	SDX, 200 mg (N=14)	Ritalin IR, 80 mg (N=15)	Ritalin LA, 80 mg (N=14)
Total TEAEs	10	15	32	16
Subjects with ≥1 TEAE, n (%)	5 (35.7)	6 (42.9)	9 (60.0)	11 (78.6)
-mild	4 (28.6)	6 (42.9)	4 (26.7)	10 (71.4)
-moderate	1 (7.1)	0	5 (33.3)	1 (7.1)
Serious adverse events	0	0	0	0
D/C study drug due to TEAE	0	0	1 (6.7)	0
Treatment emergent cardiovascular adverse events				
Palpitations	1 (7.1)	1 (7.1)	0	1 (7.1)
Tachycardia	0	0	1 (6.7)	0
Chest discomfort	0	0	1 (6.7)	0
ECG T wave abnormal	0	0	0	2 (14.3)
Hypertension	0	0	1 (6.7)	0
Most frequently reported adverse events (≥2 subjects overall)				
Energy increased	0	0	2 (13.3)	1 (7.1)
Fatigue	0	1 (7.1)	2 (13.3)	0
ECG T wave abnormal	0	0	0	2 (14.3)
Dizziness	0	2 (14.3)	3 (20.0)	2 (14.3)
Headache	1 (7.1)	1 (7.1)	2 (13.3)	1 (7.1)
Somnolence	2 (14.3)	1 (7.1)	0	0
Anxiety	0	1 (7.1)	2 (13.3)	2 (14.3)
Change in sustained attention	0	1 (7.1)	2 (13.3)	1 (7.1)
Depression	1 (7.1)	1 (7.1)	0	0
Euphoric mood	2 (14.3)	0	3 (20.0)	2 (14.3)
Hypervigilance	0	2 (14.3)	1 (6.7)	0
Mood swings	1 (7.1)	1 (7.1)	0	0

Abbreviations: D/C = discontinue; SDX = Serdexmethylphenidate; d-MPH = d-methylphenidate; ECG = Electrocardiogram; IR = immediate release; LA = long acting.

Psych Congress • September 6-10, 2023

Disclosures: The study was funded by Zevra Therapeutics. RB, SG,TCM and AS are employees and shareholders of Zevra Therapeutics.