53

Single- and Multiple-dose Pharmacokinetics of KP415, a Novel d-Methylphenidate Product Containing a Prodrug of d-Methylphenidate, in Healthy Volunteers

Rene Braeckman[1], Sven Guenther[1], Travis C. Mickle[1], Andrew C. Barrett[1], Adam Smith[1], Kathryn Anne Roupe[2], Michael Nutt[2], Cynthia Zamora[2]

[1]Kempharm, Inc., Coralville, IA; [2]Worldwide Clinical Trials, King of Prussia, PA

BACKGROUND

- KP415 is an investigational ADHD product containing a novel prodrug of d-methylphenidate (d-MPH) and immediate-release d-MPH ("d-MPH API")
- KP415 has been designed to provide both a rapid onset of d-MPH as well as sustained d-MPH concentrations throughout the day
- Preliminary pharmacokinetic data indicated that the prodrug component produces a gradual, extended-release of d-MPH throughout the day
- Following a single KP415 dose, initial d-MPH exposure during the first few hours is governed by d-MPH API and subsequent sustained d-MPH exposure primarily by prodrug conversion to d-MPH
- The optimal ratio of d-MPH API to prodrug most appropriate for a once-daily d-MPH product with a long total duration of efficacy has not yet been determined

OBJECTIVE

- To assess the single- and multiple-dose pharmacokinetics of d-MPH following KP415 doses containing different ratios of d-MPH API and prodrug

METHODS

Study design and subjects

- This was a Phase 1, open-label, multiple-dose, 4-treatment, randomized, parallel pharmacokinetic study evaluating oral solutions with different ratios of d-MPH API and prodrug compared with Concerta®, in healthy adult volunteers (N=48) under fasted conditions
- Eligible subjects were males or non-pregnant females between 18 and 55 years of age (inclusive), and weighing between 60 and 100 kg
- Enrolled subjects (12 per treatment group) were randomized to receive multiple doses of d-MPH API and prodrug (administered separately in 10-mL oral solutions), or one Concerta tablet each day for 7 days
- Oral KP415 solutions were administered containing d-MPH API:prodrug ratios (each component expressed in mol-% of total dose) of approximately 20%:80%, 30%:70%, and 40%:60%. The total d-MPH equivalent dose was 40 mg
 - **Treatment A:** 8/64 mg d-MPH API/prodrug (20%:80% ratio)
 - **Treatment B:** 12/56 mg d-MPH API/prodrug (30%:70% ratio)
 - **Treatment C:** 16/48 mg d-MPH API/prodrug (40%:60% ratio)
 - **Treatment D:** 54 mg Concerta tablet

Table 1. Treatments

Treatment	d-MPH API[1] (mg)	Prodrug API[2] (mg)	Total d-MPH dose[3] (mg)	d-MPH/ prodrug[3] (%)
A	8	64 (32)	40	20/80
B	12	56 (28)	40	30/70
C	16	48 (24)	40	40/60
D	Concerta® 54 mg	-	27[4]	-

1. The dose of d-MPH API is expressed in terms of d-methylphenidate hydrochloride.
2. The dose of KP415 is expressed in terms of KP415 chloride. The amount of d-methylphenidate hydrochloride equimolar to each KP415 dose is listed between parentheses.
3. Based on the equivalent amount of d-MPH in KP415.
4. In addition to d-MPH, a 54 mg Concerta® tablet includes 27 mg of l-MPH.

- Food was not allowed from at least 6 hours prior to dosing with study drug through approximately 4 hours following dosing with study drug

Pharmacokinetic Assessments and Analyses

- Blood samples for pharmacokinetic analysis were collected at the following times:
 - After the 1st dose of study drug (Day 1), blood samples were collected at pre-dose (0 hour; i.e., within 1 hour prior to dosing), and at 0.5, 1, 1.5, 2, 2.5, 3, 4.5, 6, 7, 7.5, 8, 8.5, 9, 10, 12, 13, and 24 hours ±5 minutes post-dose
 - After the 2nd, 3rd, 4th, 5th and 6th dose of study drug (Days 2-6), blood samples were collected at pre-dose (within 10 minutes prior to dosing), and at 1.5 and 8 hours ±5 minutes post-dose. The pre-dose sample on Day 2 was the same as the 24-hour post-dose sample after the 1st dose
 - After the last dose (7th dose) of study drug (Day 7), blood samples were collected at pre-dose (within 10 minutes prior to dosing), and at 0.5, 1, 1.5, 2, 2.5, 3, 4.5, 6, 7, 7.5, 8, 8.5, 9, 10, 12, 13, 24, 36, 48, 60 and 72 hours ±5 minutes post-dose
- Safety assessments were performed throughout the study

Statistical Analyses

- Pharmacokinetic (PK) parameters were calculated from plasma concentrations of d-MPH and KP415 using standard, non-compartmental methods using Phoenix™ WinNonlin® (Version 6.3 or higher, Pharsight Corporation)
- The following were among the pharmacokinetic parameters derived for d-MPH from the plasma concentration-time data for Day 1 and Day 7: C_{max}, C_{min} (at 24 hours post-dose), T_{max}, $AUC_{0\text{-last}}$, $AUC_{0\text{-inf}}$, $AUC_{0\text{-3h}}$, $AUC_{3\text{-7h}}$, $AUC_{7\text{-12h}}$, $AUC_{0\text{-24h}}$, $T_{1/2}$, and Accumulation Ratio (AR)

RESULTS

Subject Disposition and Demographics

- A total of 48 subjects were randomized (12 subjects per treatment group), all of whom completed the study
- Mean (SD) age was 33.6 (9.4) years, mean (SD) weight was 77.2 (9.3) kg, and 75% were males. Mean BMI (SD) at screening was 26.7 (2.7) kg/m^2
- No marked differences in demographic or baseline characteristics were apparent across treatment groups

Pharmacokinetic Findings for d-MPH

- **Figures 1-3** show single-dose (Day 1) and multiple-dose (Day 7) pharmacokinetic profiles of KP415-derived d-MPH after each treatment
- Following all treatments with d-MPH API/prodrug, d-MPH plasma concentrations increased rapidly and exhibited a single peak at approximately 1.5 to 2 hours after Dose 1 and Dose 7, followed by a gradual decline in d-MPH concentrations over the remainder of the dosing interval
- **Table 2** shows pharmacokinetic parameters describing the rate and extent of d-MPH exposure across all 4 treatments
- After both Dose 1 and Dose 7, initial onset of d-MPH plasma concentrations was faster and higher following KP415 administration through at least 4.5 hours post-dose when compared with Concerta
- Steady-state for d-MPH concentrations was achieved on Day 3 for all three KP415 treatments
- Peak plasma concentrations (C_{max}) on Day 7, primarily determined by d-MPH API in each KP415 dose, increased dose-dependently across doses of 8/64 mg (15.5 ng/mL), 12/56 mg (20.9 ng/mL), and 16/48 mg (23.8 ng/mL)
- Overall d-MPH exposure during the 24-hour dosing interval ($AUC_{0\text{-24h}}$) at steady-state was comparable across the three KP415 doses (187.0 – 207.7 h*ng/mL), and somewhat higher than observed for Concerta (176.7 h*ng/mL)
- When compared to Concerta, mean d-MPH plasma concentrations were higher following treatments with KP415 at 24 hours after Dose 1, as were all subsequent pre-dose d-MPH concentrations on Days 3 to 7
- Ritalinic acid (data not shown) was the primary metabolite for all 4 treatments

Figure 1. Plasma d-MPH concentrations following single doses of KP415 containing 8/64 mg, 12/56 mg, and 16/48 mg d-MPH API/KP415 prodrug, and 54 mg Concerta tablet

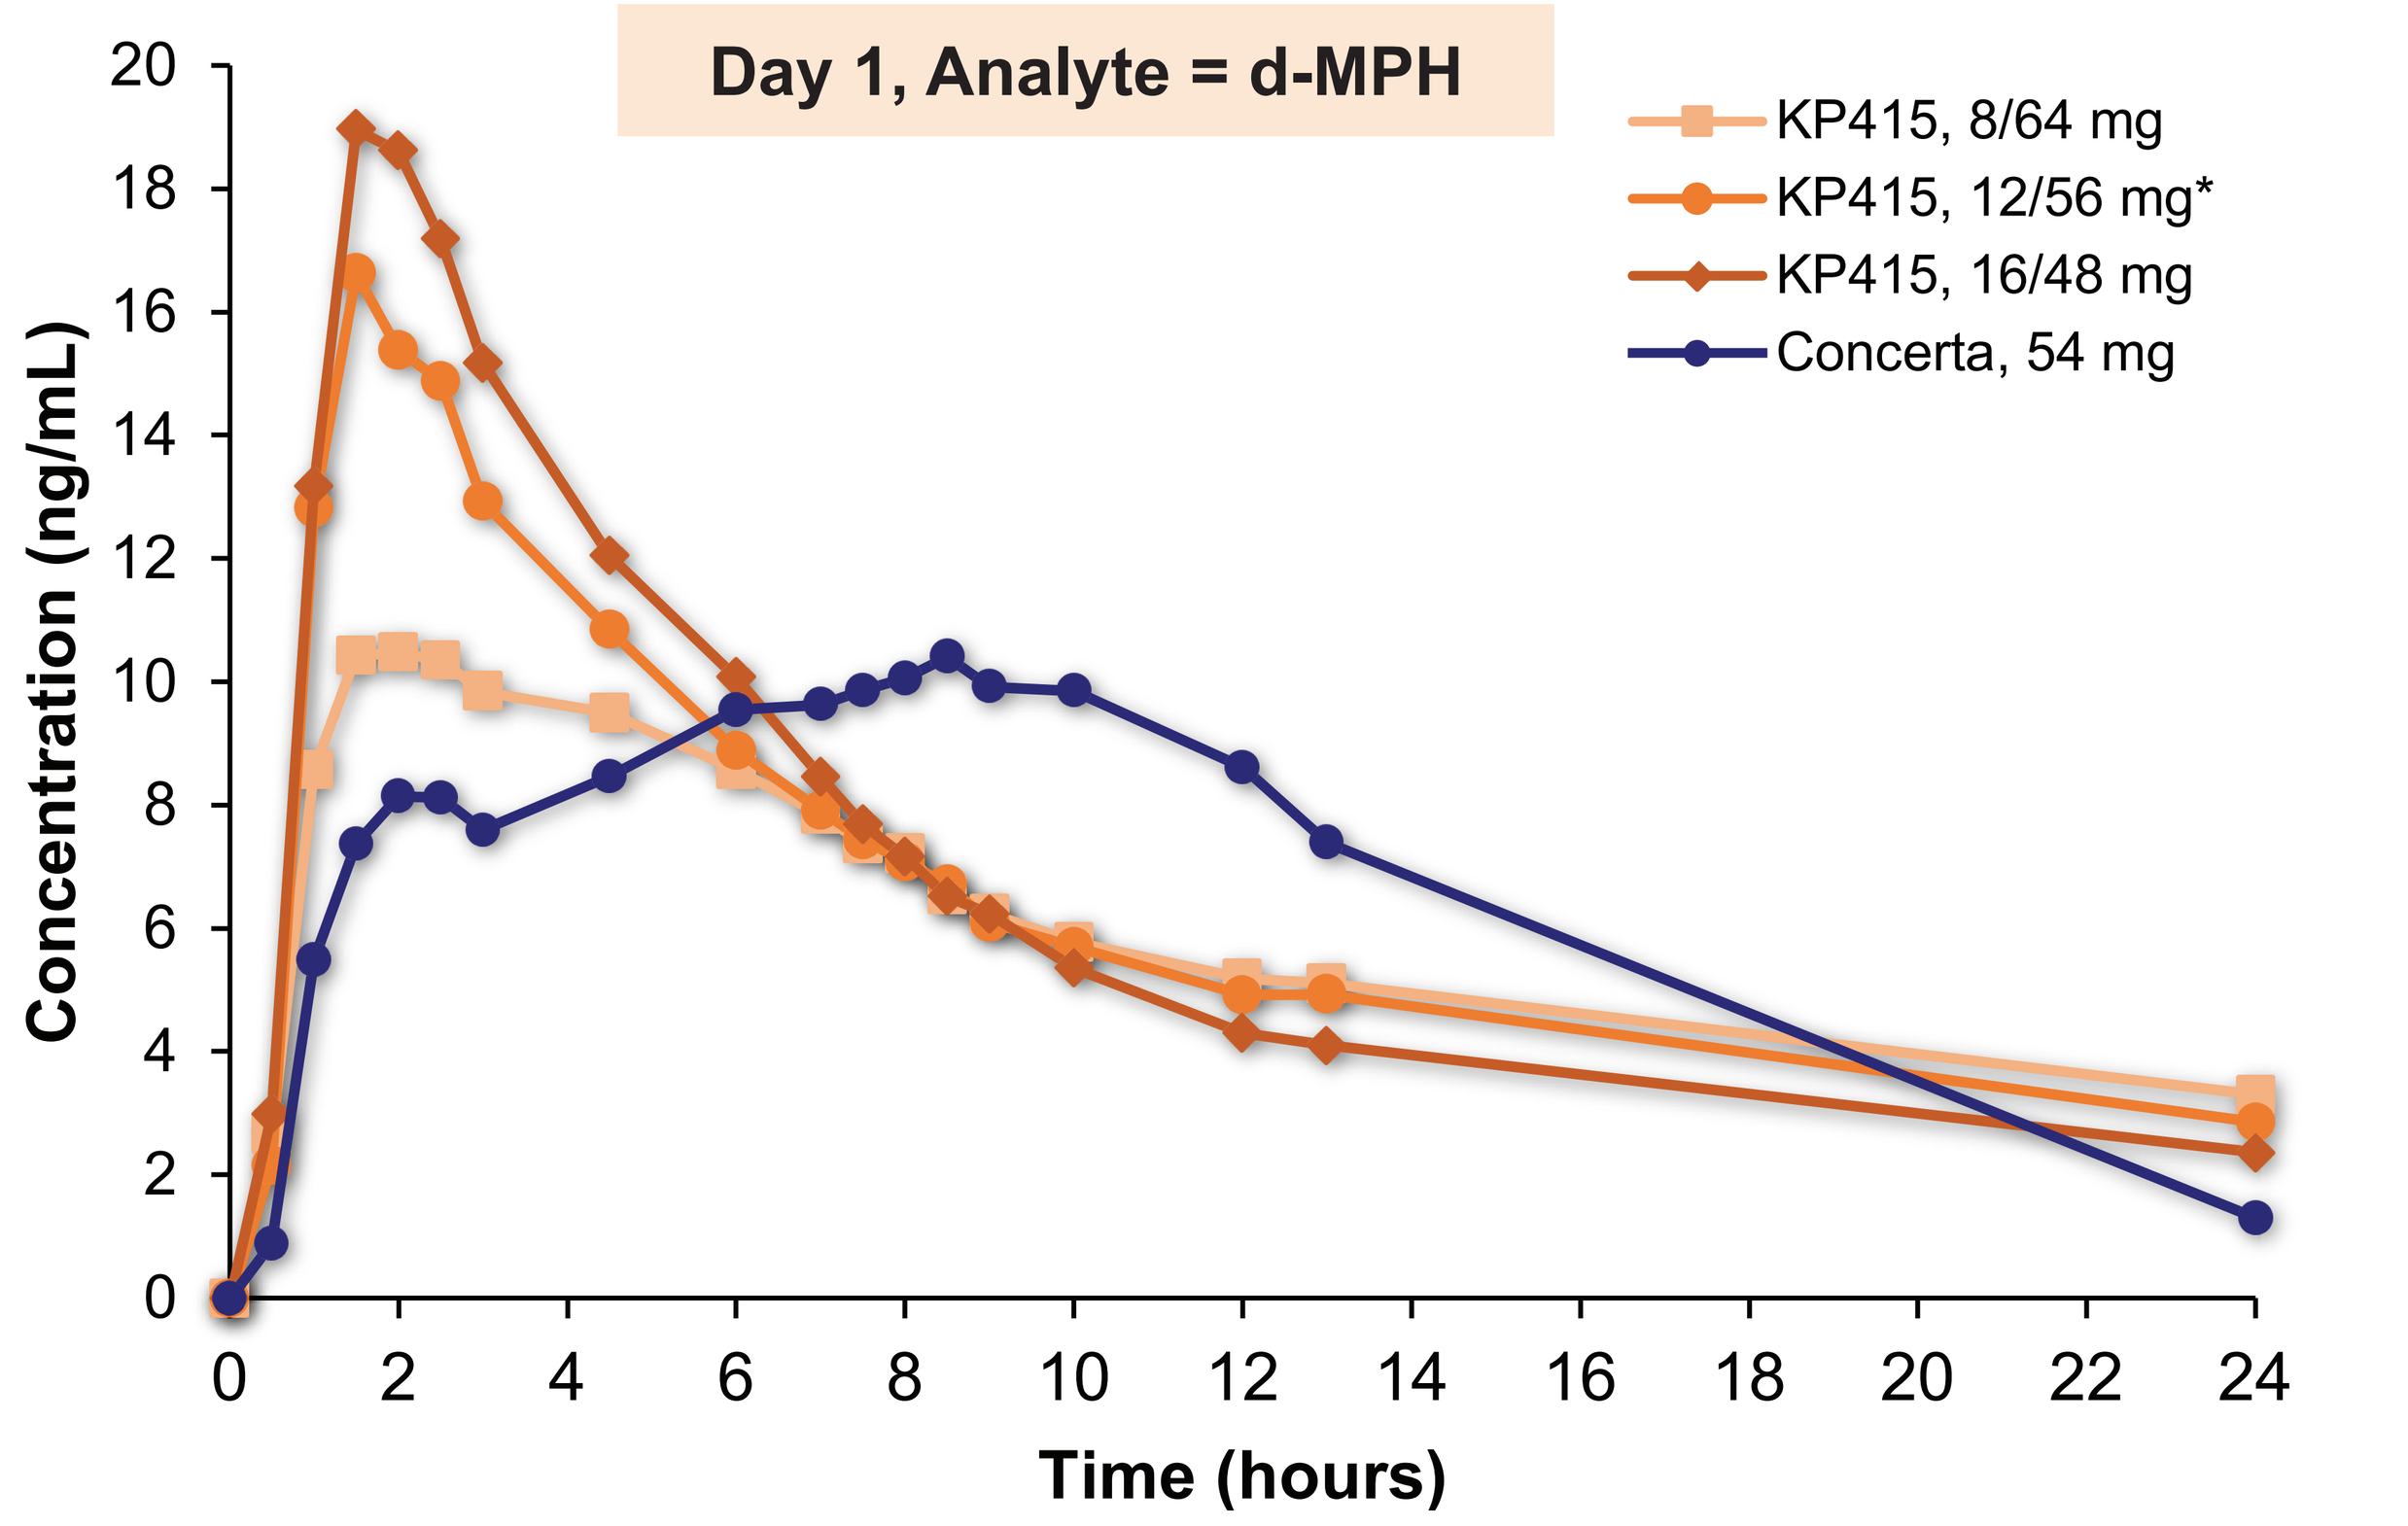
Day 1, Analyte = d-MPH
KP415, 8/64 mg
KP415, 12/56 mg*
KP415, 16/48 mg
Concerta, 54 mg
Concentration (ng/mL)
0
2
4
6
8
10
12
14
16
18
20
0 2 4 6 8 10 12 14 16 18 20 22 24
Time (hours)

* Two subjects treated with 12/56 mg d-MPH API/KP415 prodrug who appeared to be slow metabolizers of d-MPH were excluded from the analysis

Figure 2. Steady-state plasma d-MPH concentrations following 7 doses of KP415 containing 8/64 mg, 12/56 mg, and 16/48 mg d-MPH API/KP415 prodrug, and 54 mg Concerta tablet

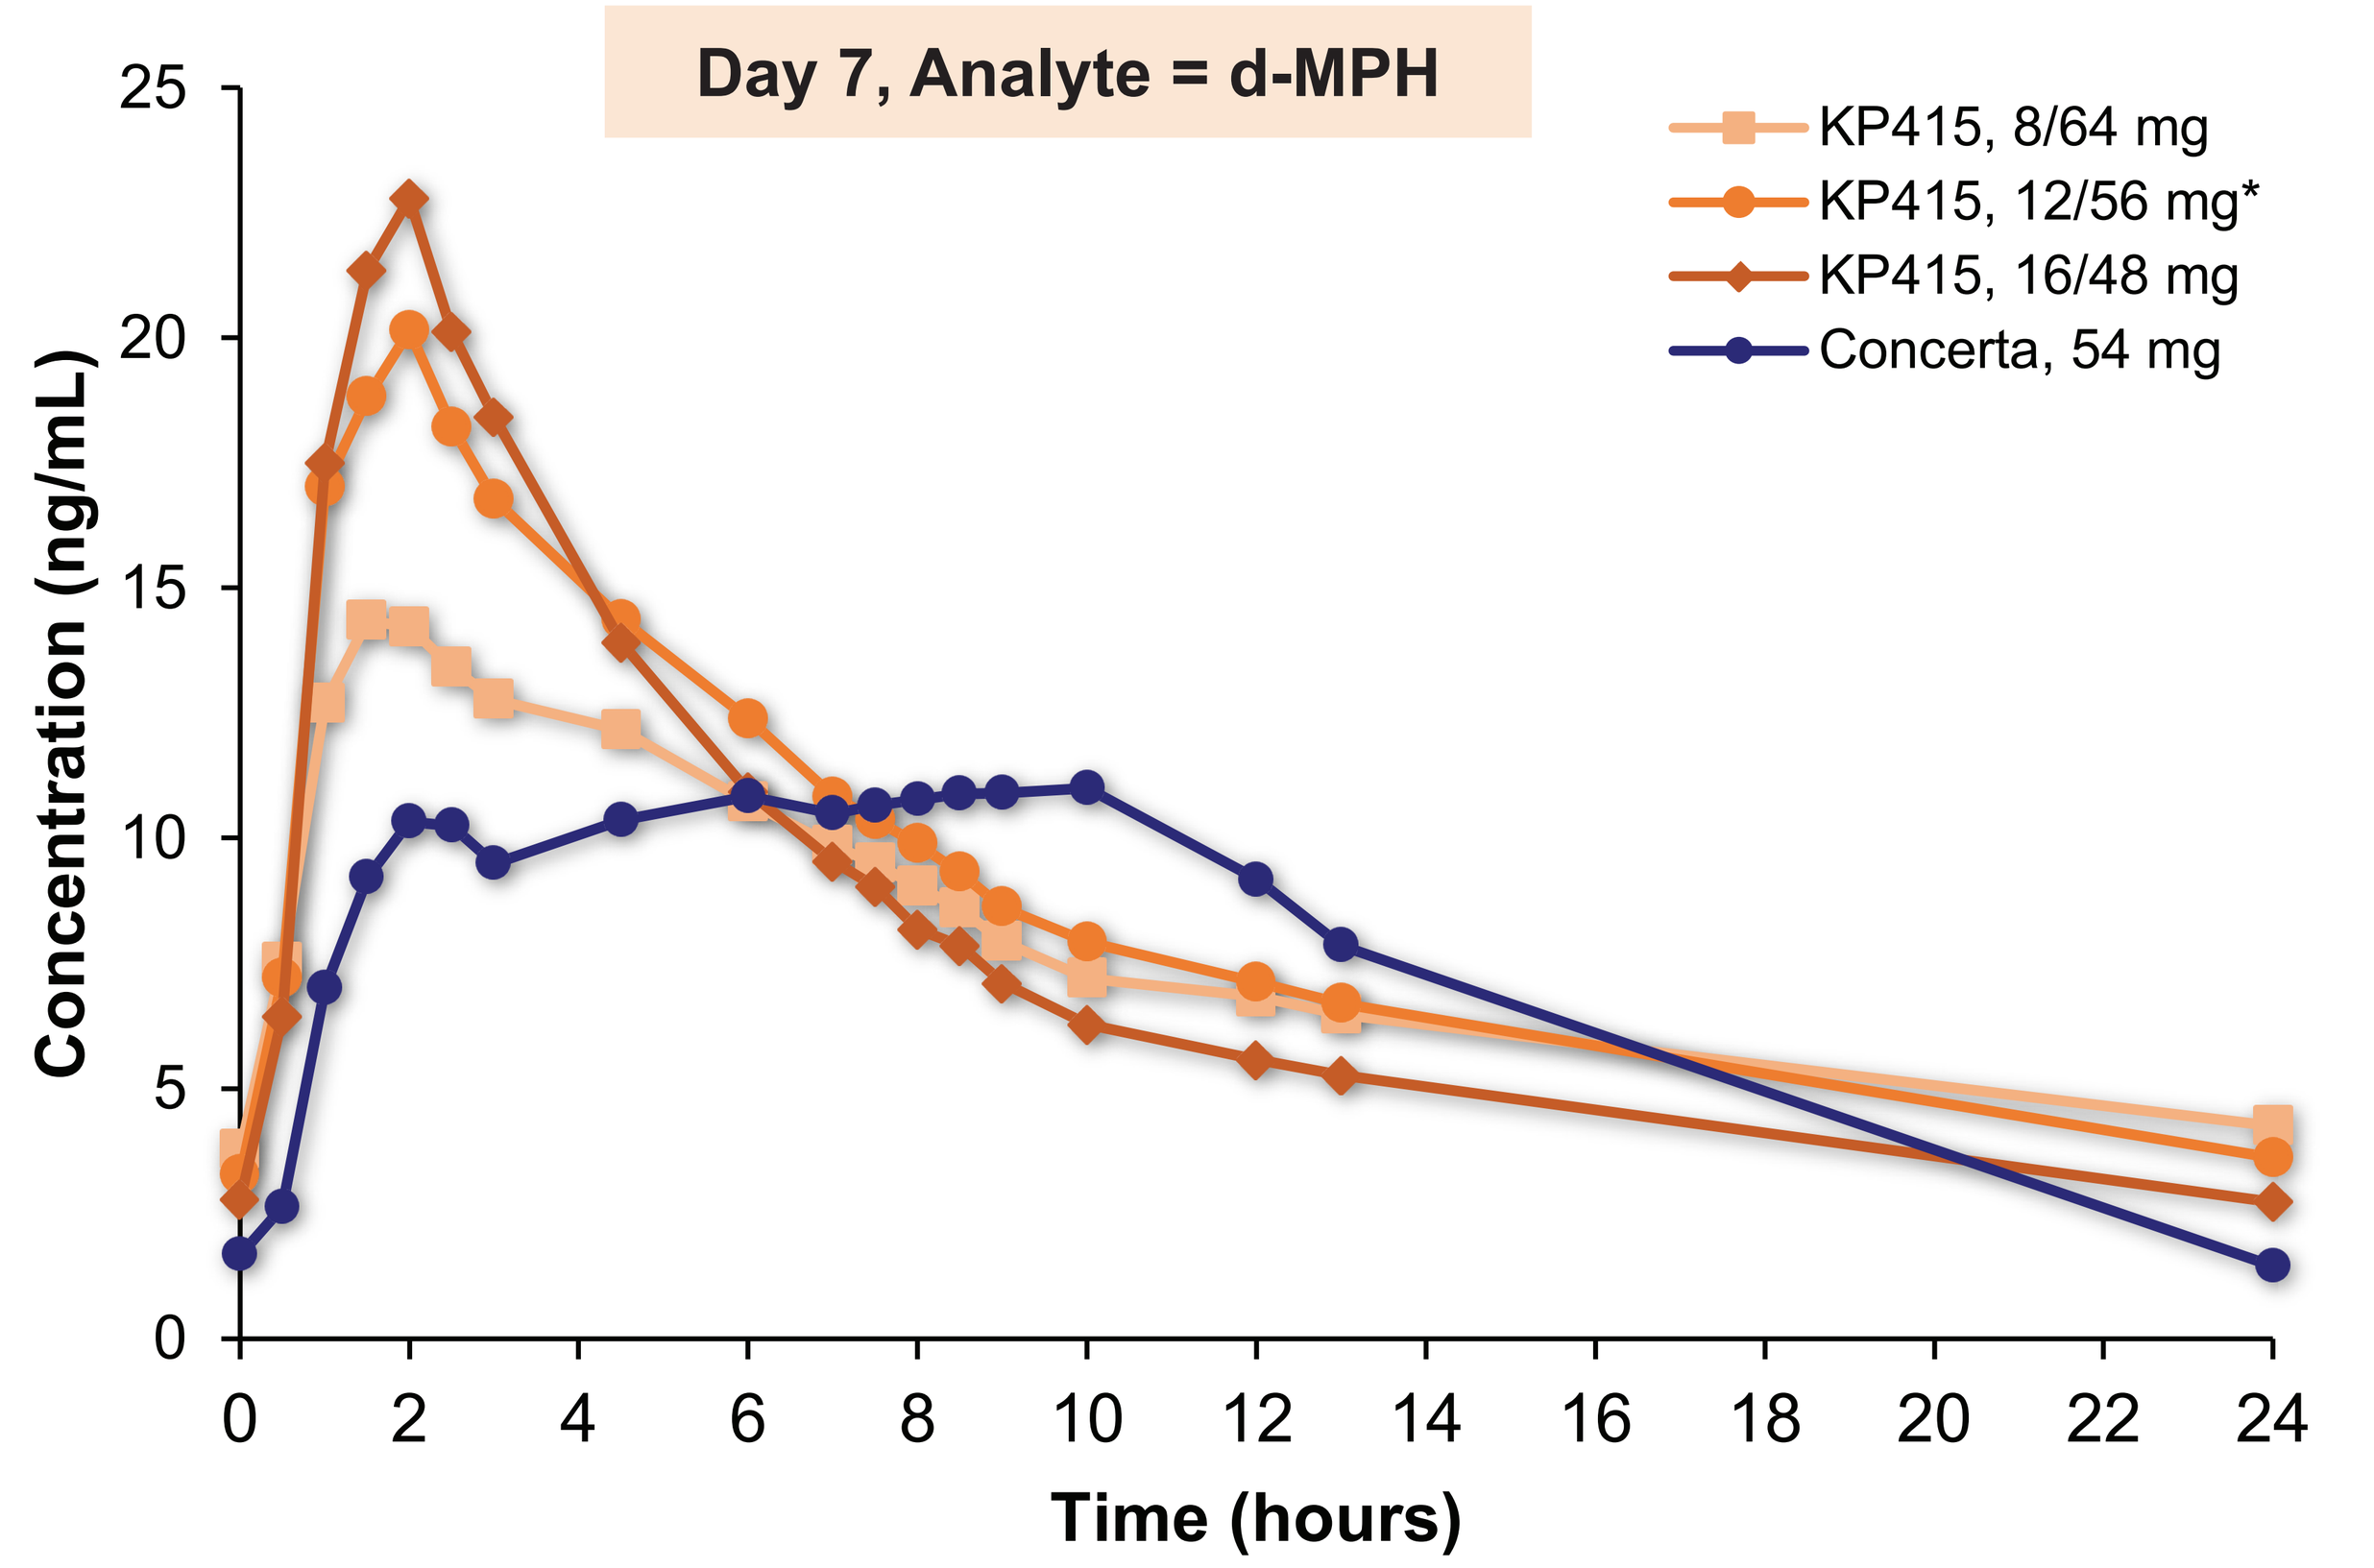
Day 7, Analyte = d-MPH
KP415, 8/64 mg
KP415, 12/56 mg*
KP415, 16/48 mg
Concerta, 54 mg
Concentration (ng/mL)
0
5
10
15
20
25
0 2 4 6 8 10 12 14 16 18 20 22 24
Time (hours)

* Two subjects treated with 12/56 mg d-MPH API/KP415 prodrug who appeared to be slow metabolizers of d-MPH were excluded from the analysis

Figure 3. Plasma d-MPH concentrations across each daily dose of KP415 containing 12/56 mg d-MPH API/KP415 prodrug, and 54 mg Concerta tablet. For simplicity, only one KP415 treatment (B) is plotted

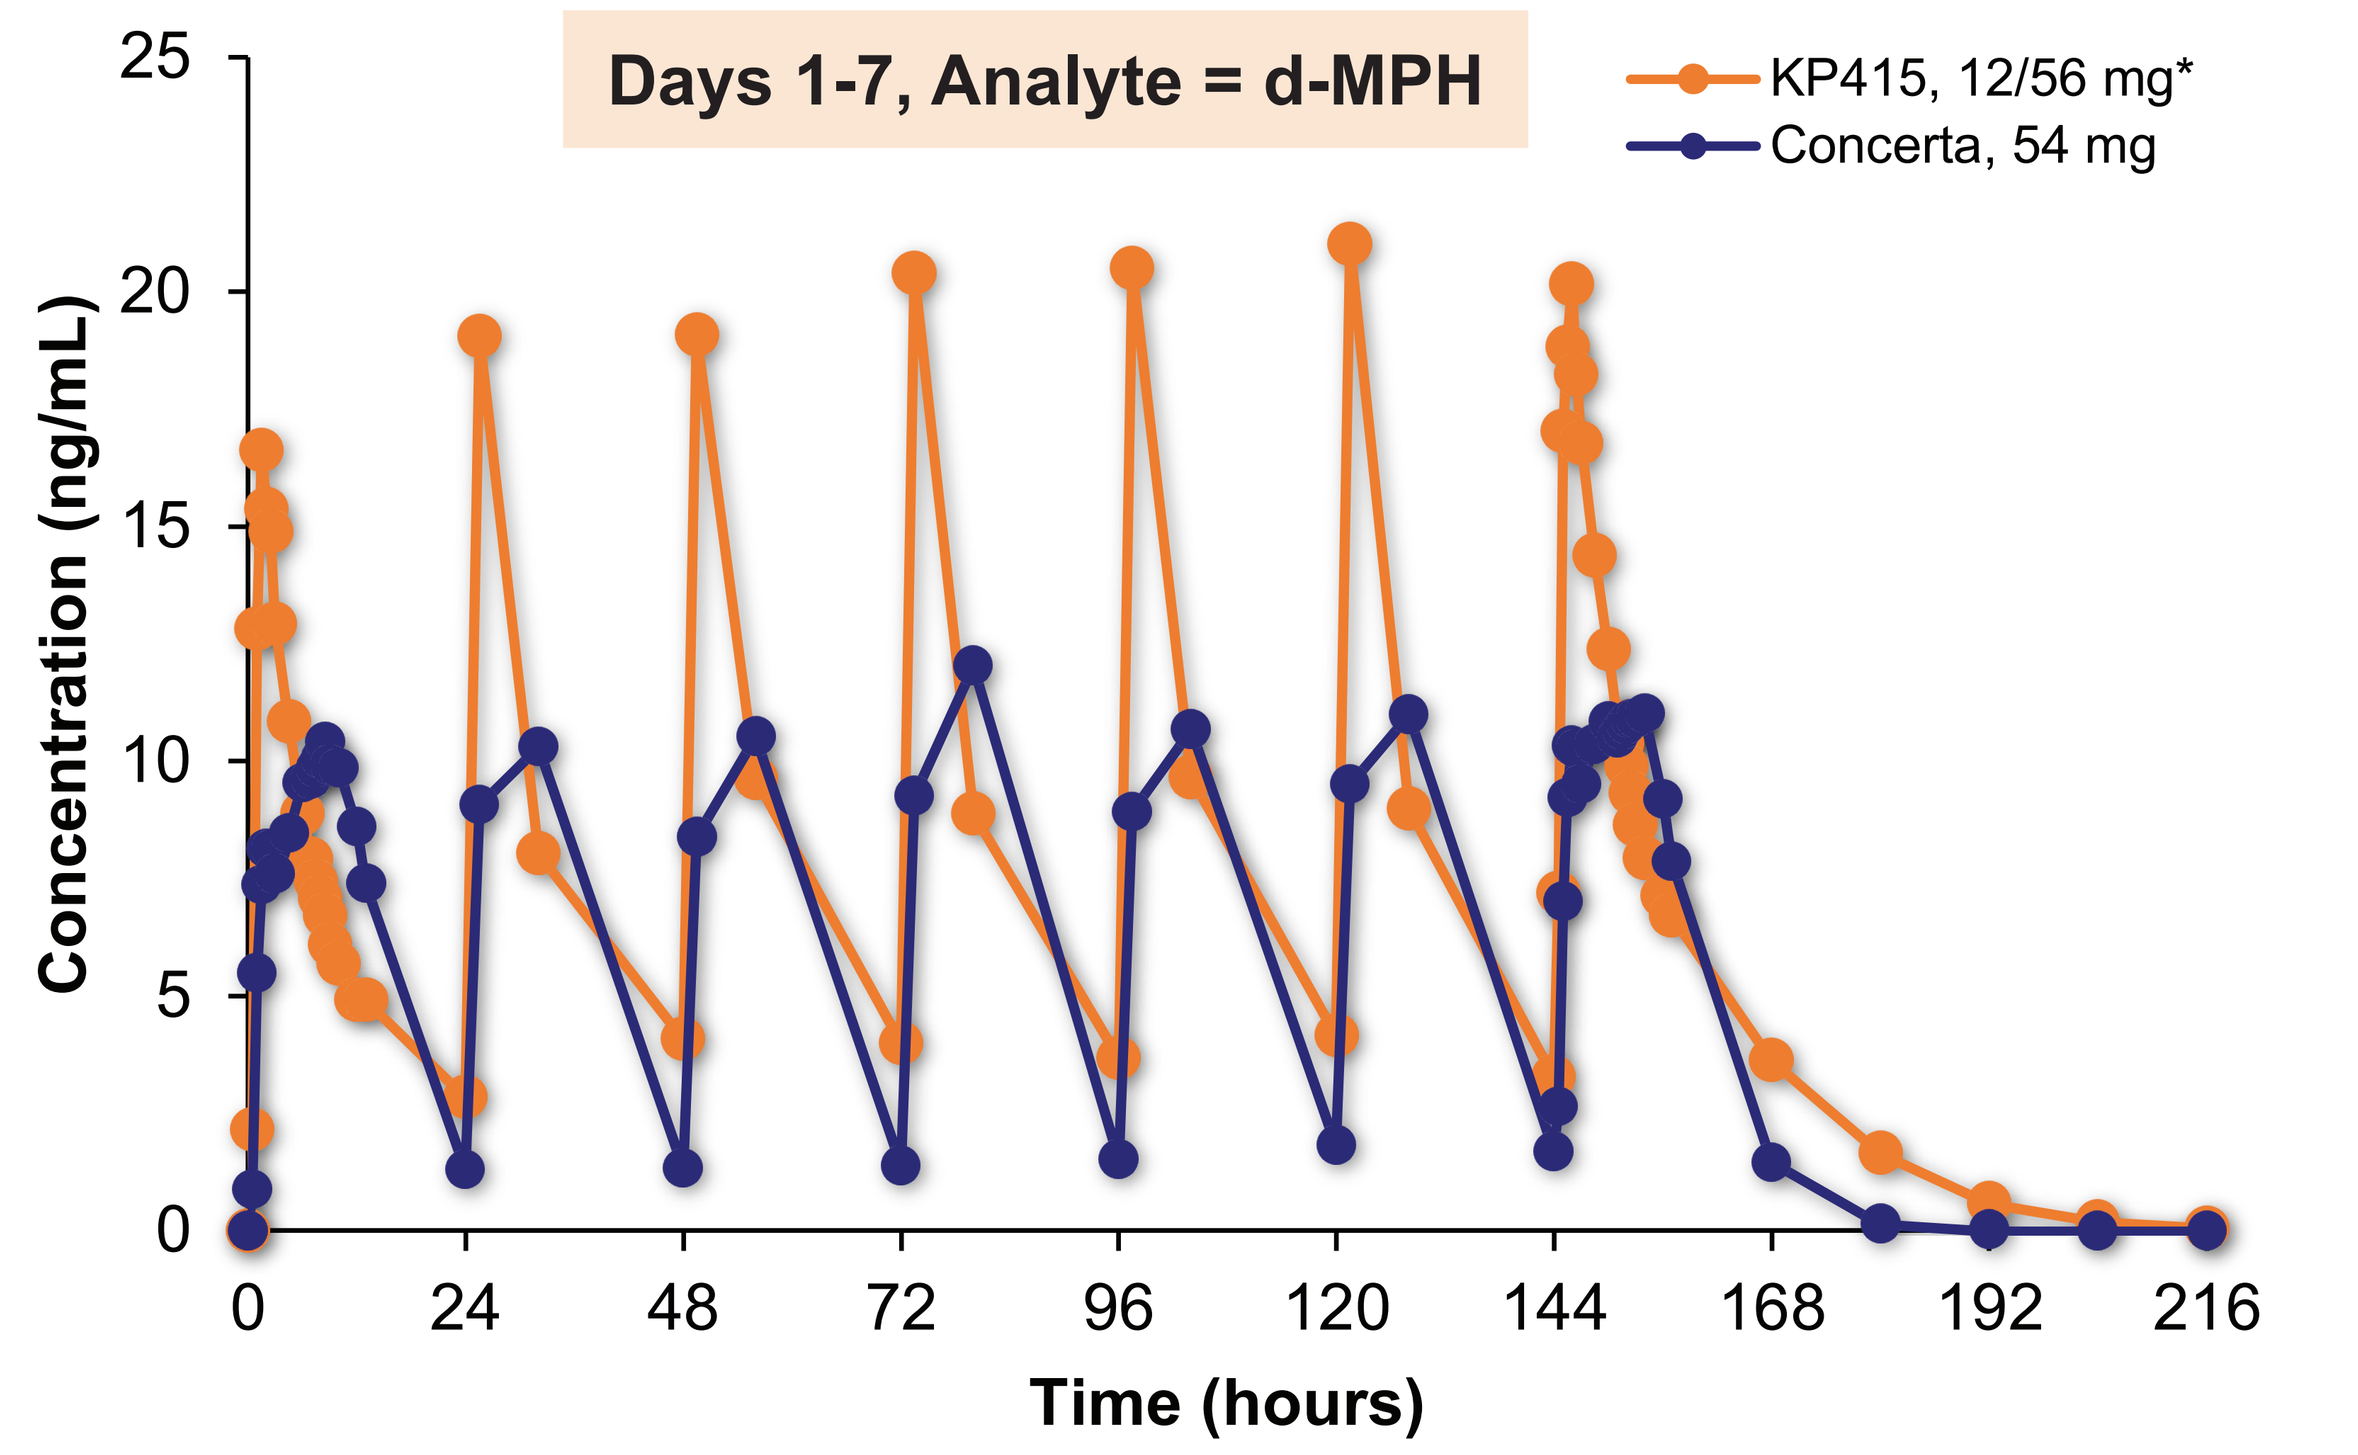
Days 1-7, Analyte = d-MPH
KP415, 12/56 mg*
Concerta, 54 mg
Concentration (ng/mL)
0
5
10
15
20
25
0 24 48 72 96 120 144 168 192 216
Time (hours)

* Two subjects treated with 12/56 mg d-MPH API/KP415 prodrug who appeared to be slow metabolizers of d-MPH were excluded from the analysis

Table 2. Steady-state (Day 7) pharmacokinetic parameters for d-MPH following oral administration of KP415 doses containing 8/64 mg, 12/56 mg, and 16/48 mg d-MPH API/KP415 prodrug, and 54 mg Concerta

PK Parameter	8/64 mg d-MPH API/KP415 prodrug Mean (SD)	12/56 mg d-MPH API/KP415 prodrug Mean (SD)	16/48 mg d-MPH API/KP415 prodrug Mean (SD)	Concerta®, 54 mg Mean (SD)
C_{max} (ng/mL)	15.5 (3.7)	20.9 (3.1)	23.8 (5.7)	12.6 (4.3)
$AUC_{0\text{-24h}}$ (h*ng/mL)	187.0 (41.0)	207.6 (54.4)	187.2 (39.8)	176.7 (66.7)
$AUC_{0\text{-3h}}$ (h*ng/mL)	32.3 (9.5)	45.8 (8.0)	49.5 (10.0)	22.5 (6.8)
$AUC_{3\text{-7h}}$ (h*ng/mL)	46.2 (9.9)	55.1 (13.5)	53.2 (11.6)	41.5 (13.9)
$AUC_{7\text{-12h}}$ (h*ng/mL)	39.7 (8.9)	43.0 (14.2)	35.2 (8.9)	52.7 (21.6)
$T_{1/2}$ (h)	8.5 (2.3)	8.9 (2.2)	9.2 (3.5)	4.5 (0.9)
T_{max} (h)	1.6 (0.5)	1.8 (0.6)	1.7 (0.3)	5.0 (3.0)

- Across the three KP415 doses, accumulation of d-MPH at steady-state (Dose 7 vs. Dose 1) ranged from 20-33% for C_{max}, 18-31% for C_{min}, and 25-34% for $AUC_{0\text{-24h}}$
- Following Concerta, accumulation of d-MPH at steady-state was 13% for C_{max}, 38% at C_{min}, and 12% for $AUC_{0\text{-24h}}$

Pharmacokinetic Findings for Intact Prodrug

- **Figure 4** shows that exposure to intact prodrug peaked at approximately 2 hours post-dose and was largely eliminated by 24 hours
- Accumulation of intact prodrug at steady-state (Dose 7 vs. Dose 1) was 24% for C_{max}, 13% for C_{min}, and 11% for $AUC_{0\text{-24h}}$
- Similar findings were observed for the other KP415 ratios

Adverse Events

- Adverse events after all treatments were typical of stimulants
- The most commonly reported AEs were euphoria (2 subjects, 16.7%) and headache (2 subjects, 16.7%) for 8/64 mg d-MPH/prodrug, headache (2 subjects, 16.7%) for 12/56 mg, anxiety (3 subjects, 25.0%) for 16/48 mg, and decreased appetite (2 subjects, 16.7%) and dry mouth (2 subjects, 16.7%) for Concerta
- All AEs were rated as mild by Investigators
- No serious or severe AEs occurred

Figure 4. Plasma concentrations of intact prodrug following Dose 1 and Dose 7 of KP415 containing 12/56 mg d-MPH API/KP415. For simplicity, only one KP415 treatment (B) is plotted.

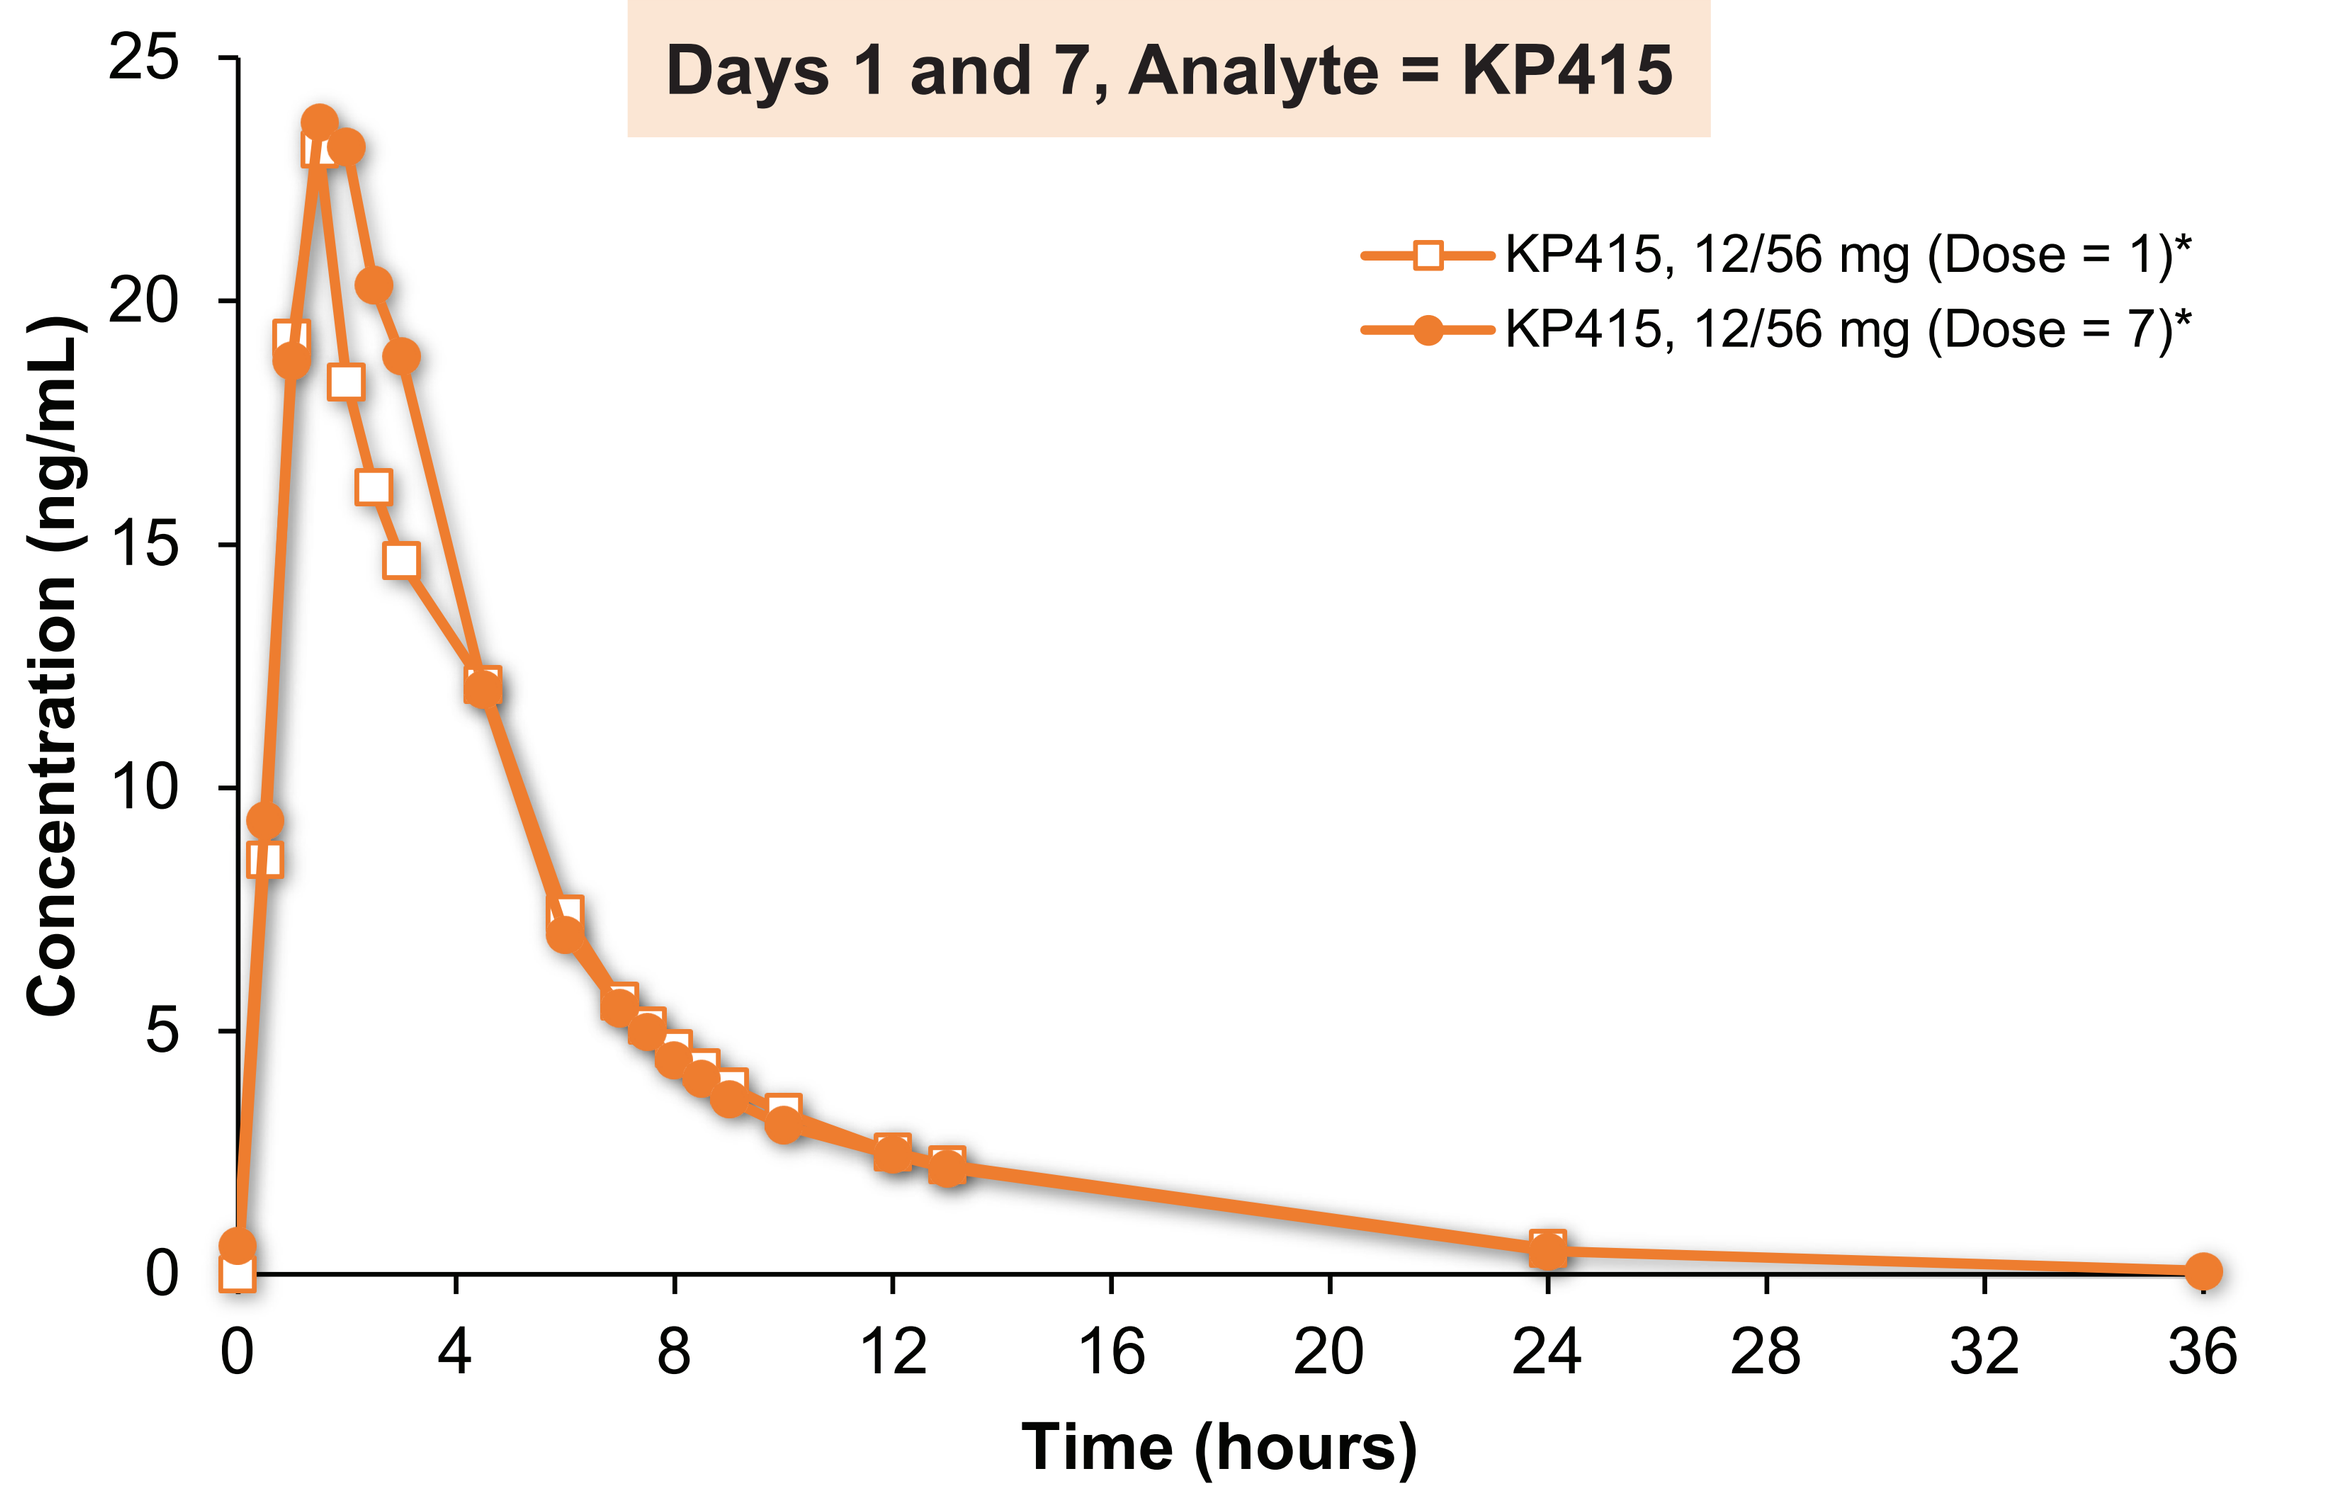
Days 1 and 7, Analyte = KP415
KP415, 12/56 mg (Dose = 1)*
KP415, 12/56 mg (Dose = 7)*
Concentration (ng/mL)
0
5
10
15
20
25
0 4 8 12 16 20 24 28 32 36
Time (hours)

* Two subjects treated with 12/56 mg d-MPH API/KP415 prodrug who appeared to be slow metabolizers of d-MPH were excluded from the analysis

CONCLUSIONS

- Based on a desired product profile that demonstrates rapid onset and sustained delivery of d-MPH at steady-state, with low levels of accumulation of the intact prodrug, a molar dose ratio of 30% d-MPH API to 70% prodrug (corresponding to KP415 dose of 12/56 mg d-MPH API/prodrug) was selected for further clinical development of KP415
- KP415-associated adverse events were mild and typical of stimulant therapy

Disclosures

Rene Braeckman, Sven Guenther, Travis C. Mickle, Andrew C. Barrett, and Adam Smith are employees and shareholders of KemPharm, Inc. Kathryn Anne Roupe, Michael Nutt, and Cynthia Zamora have no conflicts of interest to declare.

Support

This study was funded by KemPharm, Inc., Coralville, IA, USA. Design support was provided by Research Triangle Graphics, LLC

American Professional Society for ADHD and Related Disorders • January 12-14, 2018 • Washington, D.C.