Poster S37

Mass Balance and Metabolic Pathway Following Oral Administration of [14C]-Serdexmethylphenidate, a Novel Prodrug of d-Methylphenidate

Rene Braeckman,[1] Andrew C. Barrett,[1] Sven Guenther,[1] Travis C. Mickle,[1] Meg Corliss[2]

[1]KemPharm, Inc., Celebration, FL, USA; [2]Corium, Inc., Grand Rapids, MI, USA

INTRODUCTION

- Serdexmethylphenidate (SDX) 70%/d-methylphenidate (d-MPH) 30% (AZSTARYS™) is available as a once-daily, orally administered capsule for the treatment of attention-deficit hyperactivity disorder (ADHD).
- Early exposure to the medication is governed by d-MPH, and mid- to late-day exposure is governed primarily by its prodrug, SDX, which is gradually converted to d-MPH throughout the day.
- d-MPH is a well-known compound, but SDX is a new molecular entity; therefore, a study was conducted to understand the mass balance and metabolite profile of SDX.

OBJECTIVE

- To investigate the absorption, metabolism, and excretion of single-entity SDX following oral administration of radiolabeled 60 mg [^{14}C]-SDX chloride (molar equivalent to 30 mg d-MPH hydrochloride [HCl]).

METHODS

Study Design, Subjects, and Test Compound

- This was an open-label, single radiolabeled dose, nonrandomized study.
- The study was conducted in 8 healthy adult male subjects under fasted conditions.
- Subjects were administered a single dose of 20 mL mixture of nonradiolabeled SDX chloride and ^{14}C-isotopically modified [^{14}C]-SDX chloride in deionized water. The dose was 60 mg [^{14}C]-SDX chloride per subject, which is the molar equivalent of 30 mg (112 µmol) d-MPH HCl.

Assessments

- Blood samples (for whole blood and plasma analysis), urine samples, and fecal samples were collected for the measurement of SDX prodrug and metabolites, radioanalysis, mass balance, and/or metabolite identification.
 - Whole blood and plasma was collected at predose (0 hour) and at 0.5, 1, 1.5, 2, 2.5, 3, 4, 5, 6, 7, 8, 10, 12, 14, 16, 24, 36, 48, 60, 72, 96, 120, 144, and 168 hours postdose.
 - Urine samples were collected at predose (0 hour) and at 0-4, 4-8, 8-12, 12-24, 24-36, 36-48, and then in 24-hour intervals postdose until discharge criteria were met.
 - Fecal samples were collected at predose (0 hour) and at 0-24 hours, then at 24-hour intervals postdose until discharge criteria were met.
- Mass balance was assessed by calculating the amount of drug-related material (based on total radioactivity) collected in urine and feces relative to the amount of drug administered (radioactive dose).
- SDX and its metabolites were analyzed by liquid chromatography coupled with radioactivity detection (LC-RAD).

RESULTS

Subject Demographics

- Subjects' age ranged from 21 to 49 years.
- Body mass index of the subjects ranged from 23.5 to 30.2 kg/m^2.

Recovery of SDX

- Nearly all of the radiolabeled dose was recovered in feces and urine.
 - The mean total radioactivity recovered from the administration of [^{14}C]-SDX chloride in urine and feces was 62.1% and 36.8%, respectively. The mean combined total recovery of radioactivity over 168 hours was 98.9% (**Figure 1**).

Figure 1. **Mean cumulative amount of total radioactivity recovered in urine, feces, and total recovery (percentage of administered dose) after single oral administration of 60 mg [^{14}C]- SDX chloride.**

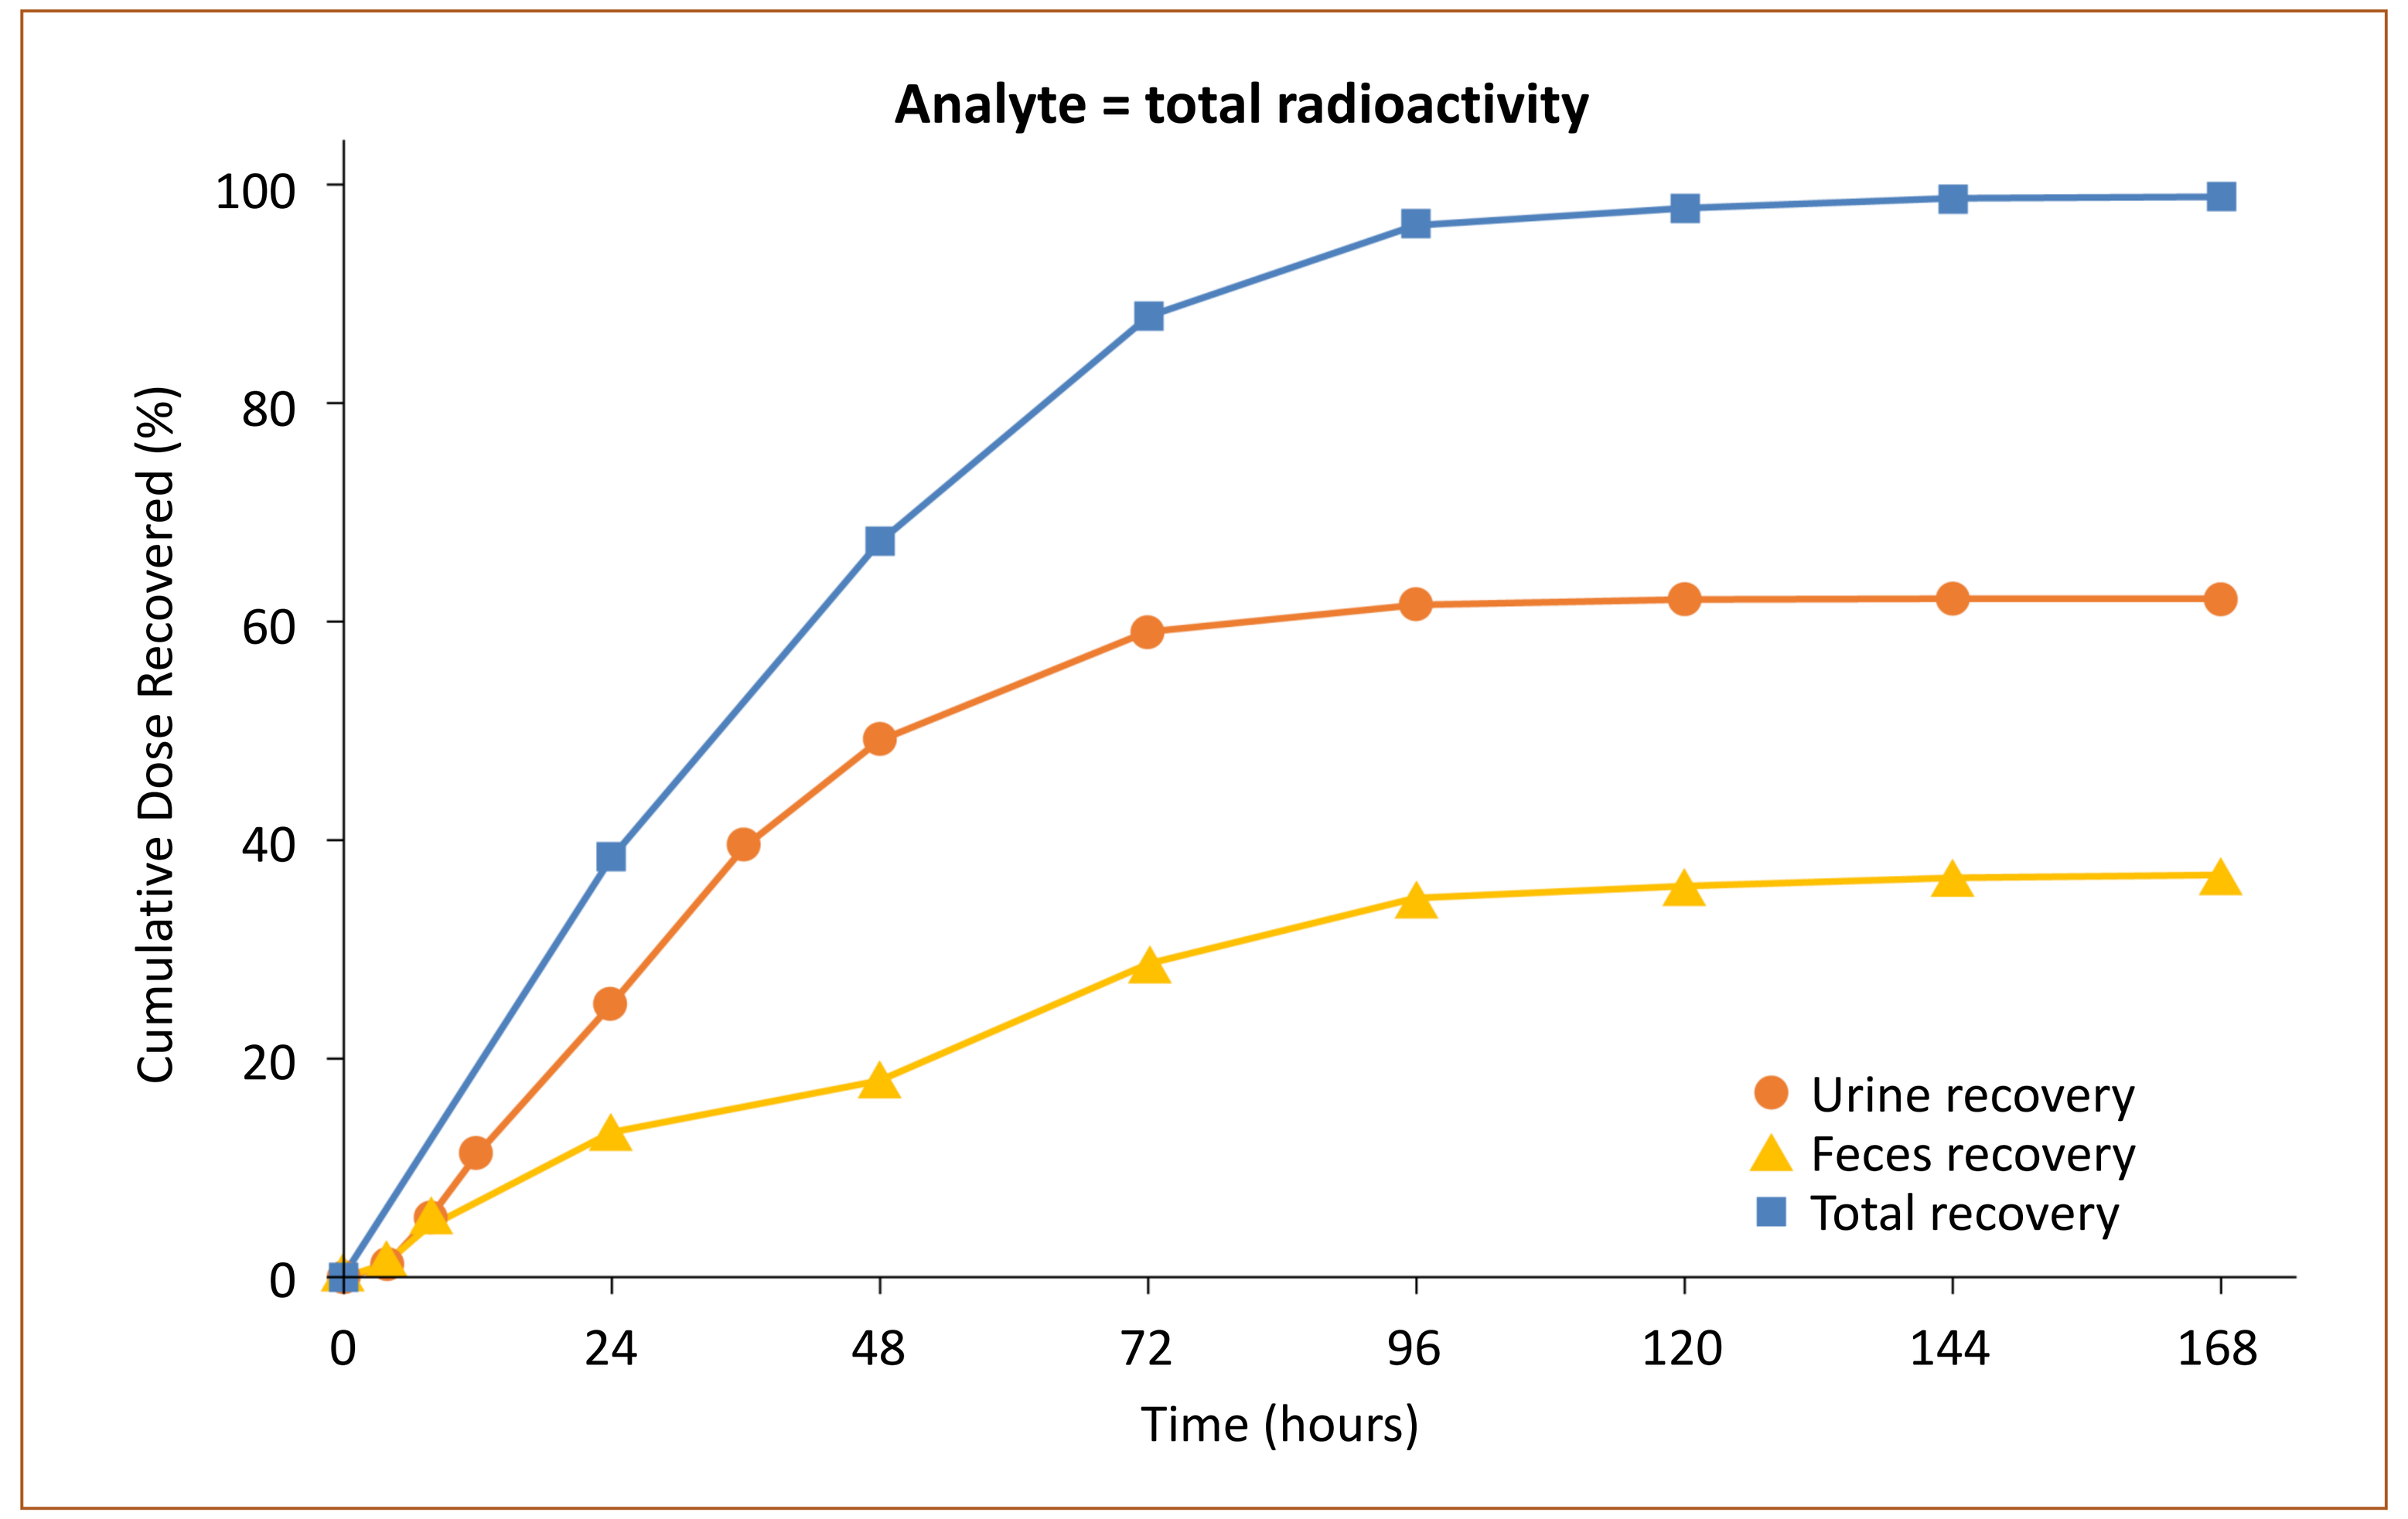
Analyte = total radioactivity
Cumulative Dose Recovered (%)
0
20
40
60
80
100
0
24
48
72
96
120
144
168
Time (hours)
Urine recovery
Feces recovery
Total recovery

- The total (all radioactive chemical species) radioactivity-time curves for whole blood and plasma were very similar, with peak levels observed at 20.3 and 21.3 hours postdose, respectively (**Figure 2**).

Figure 2. **Mean concentration-time profiles of total radioactivity in whole blood and plasma after 60 mg [^{14}C]-SDX chloride.**

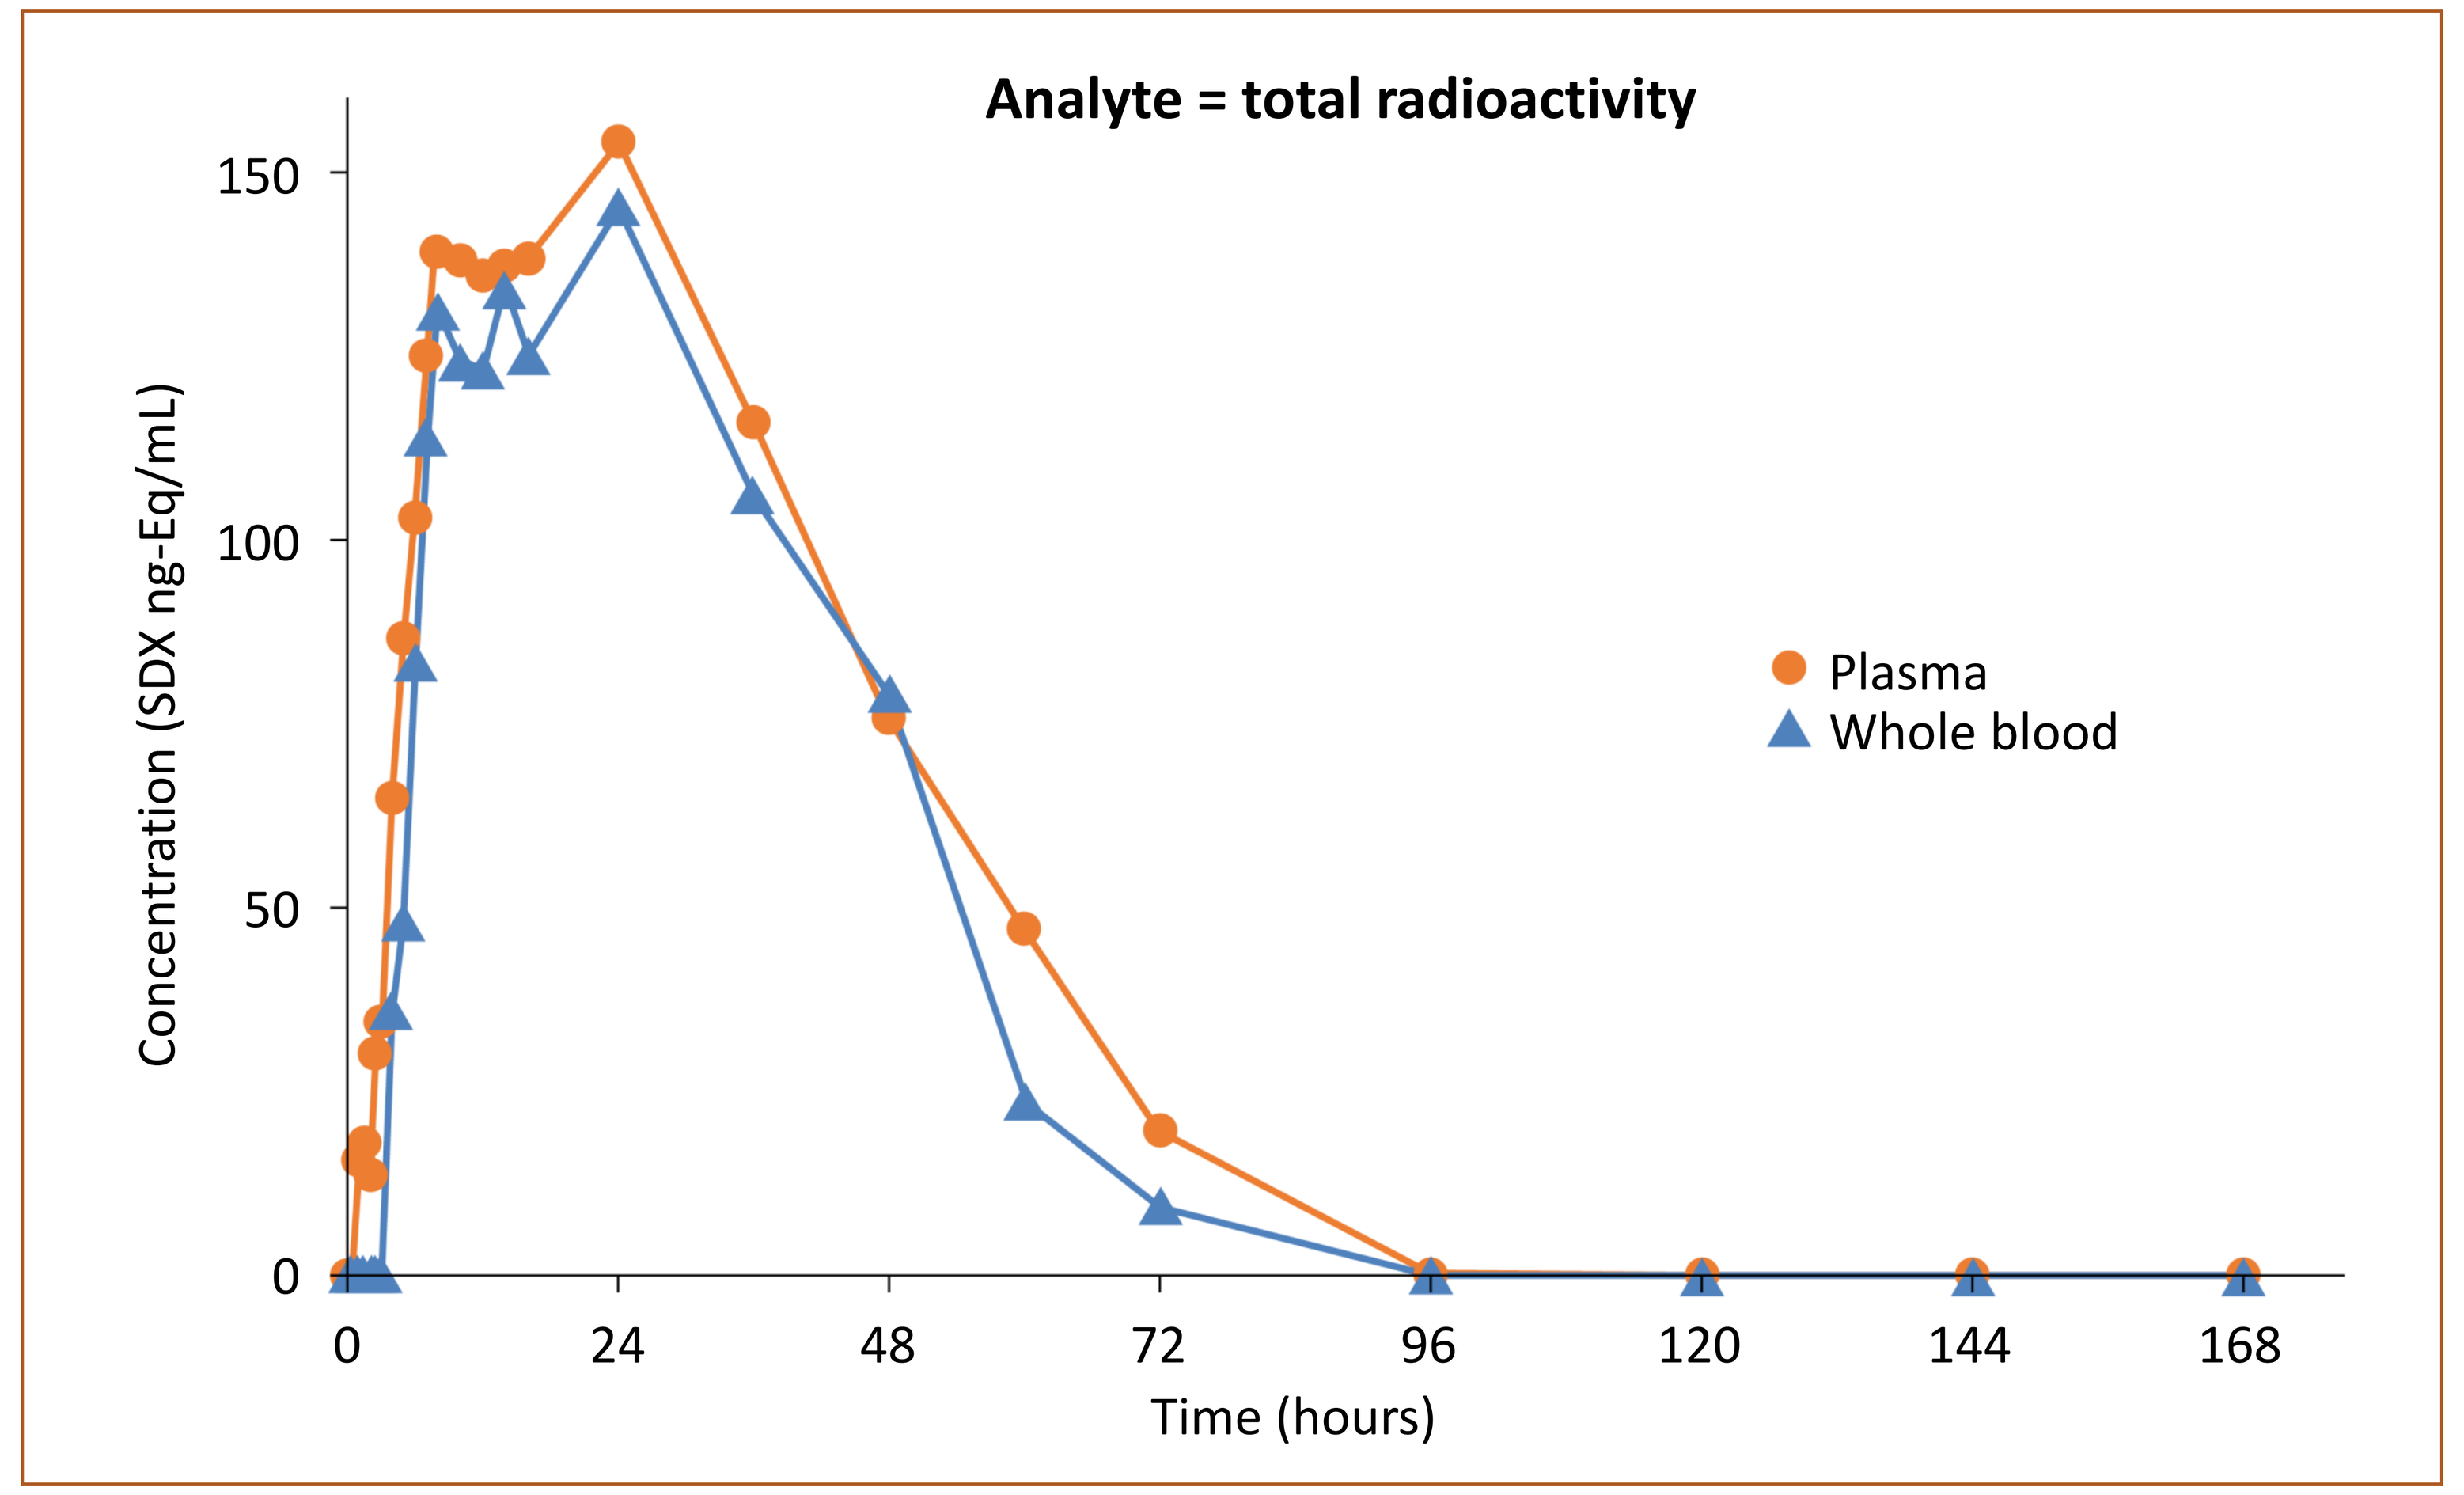
Analyte = total radioactivity
Concentration (SDX ng-Eq/mL)
0
50
100
150
0
24
48
72
96
120
144
168
Time (hours)
Plasma
Whole blood

The mean concentration-time profiles are based on the median values of each individual subject.

Analysis of SDX and Its Metabolites in Plasma, Urine, and Feces

- The following species were identified in plasma: ritalinic acid, SDX, d-MPH, and 6-oxo- ritalinic acid.
- Ritalinic acid was the most abundant metabolite in plasma (**Figure 3**) as determined by LC-RAD.
- Ritalinic acid recovery was 45.2% and 18.1% of the total dose in urine and feces, respectively.

Figure 3. **Mean molar plasma concentrations of nonradioactive SDX, d-MPH, and ritalinic acid, and the sum of SDX, d-MPH, and ritalinic acid in plasma vs time profiles after single oral administration of 60 mg [^{14}C]-SDX chloride.**

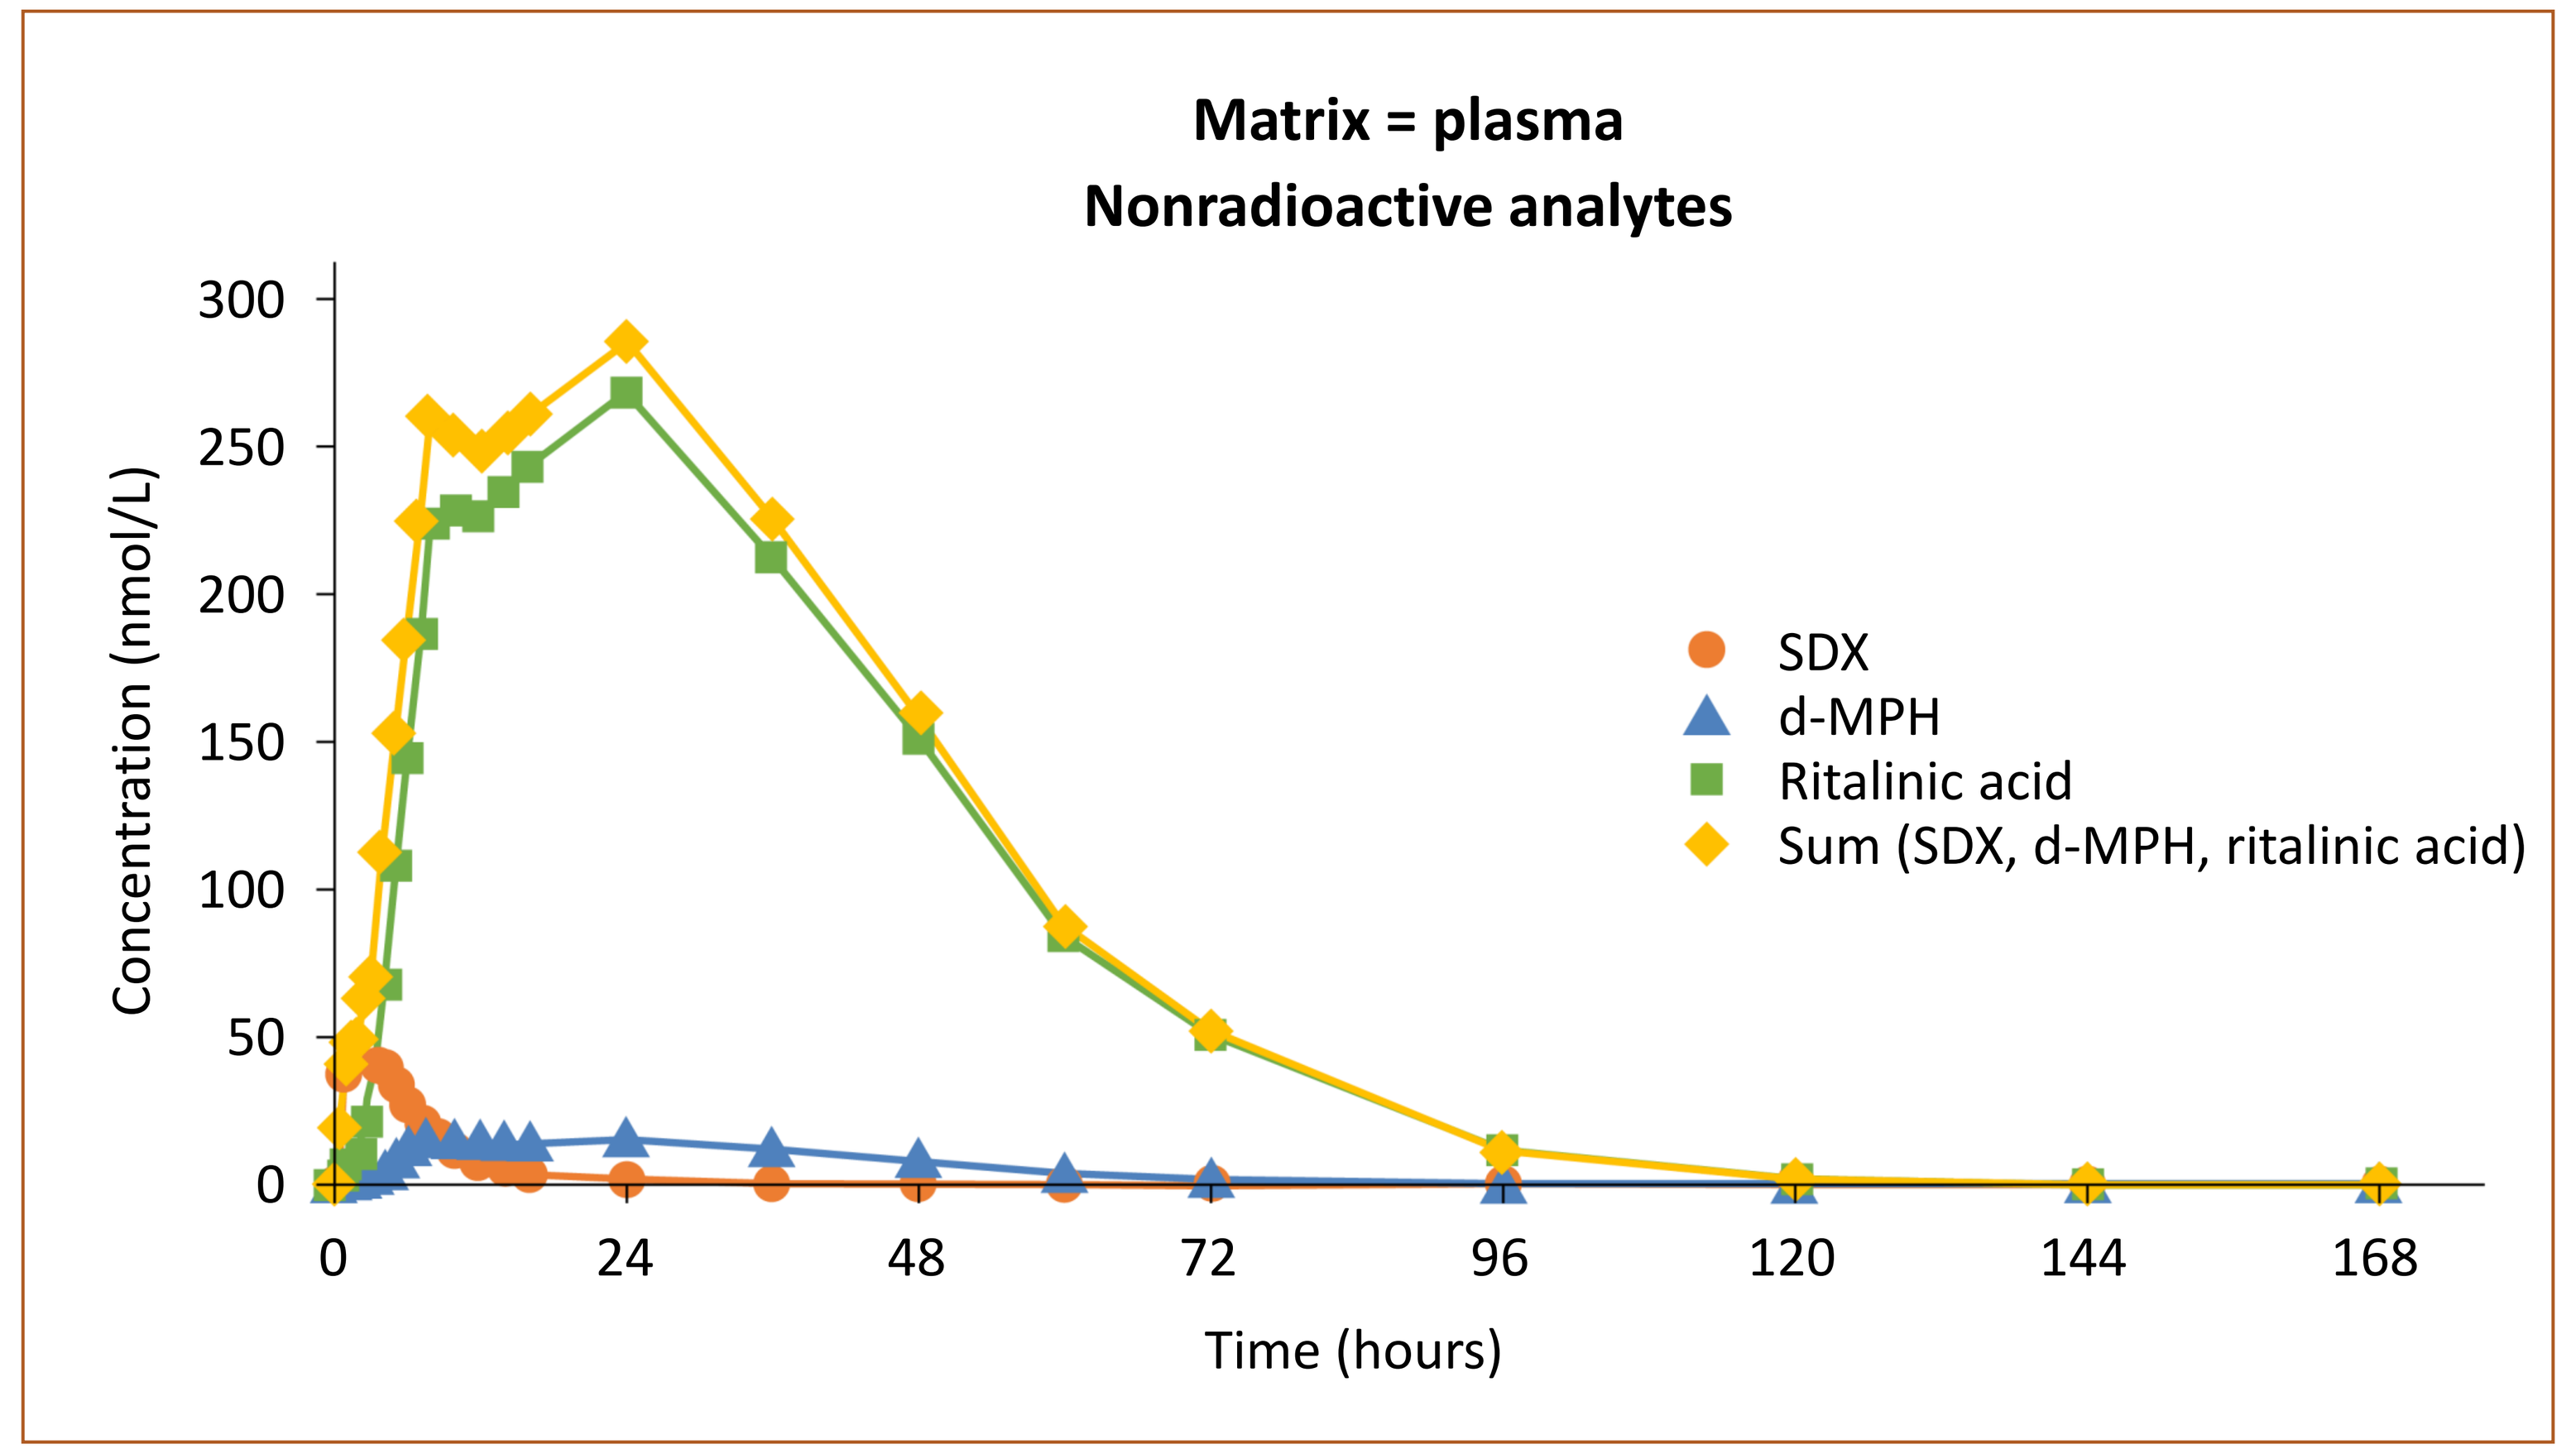
Matrix = plasma
Nonradioactive analytes
Concentration (nmol/L)
0
50
100
150
200
250
300
0
24
48
72
96
120
144
168
Time (hours)
SDX
d-MPH
Ritalinic acid
Sum (SDX, d-MPH, ritalinic acid)

- The SDX level was very low in urine (0.43%), but SDX was the second most abundant species in feces (10.8%) (**Table 1**).
- Recovery of d-MPH was low in urine and feces (both 2.74%). Other metabolites—all with low abundance—included 6-oxo-ritalinic acid, SDX without the serine moiety attached (SDX-des-Ser), and one unidentifiable species.

Table 1. **Percent of dose recovered of SDX and metabolites identified by LC-RAD in urine and feces per collection interval after administration of 60 mg [^{14}C]-SDX chloride.**

	% of dose excreted						
	SDX	d-MPH	RA	6-oxo-RA	Unknown	SDX-des-Ser	Sum[a]
Urine (total)	**0.426**	***2.742***	45.171	4.079	0.671	ND	53.089
Feces (total)	**10.804**	***2.741***	18.060	ND	ND	1.334	32.938

[a]Sum is sum of % dose of SDX, d-MPH, RA, and SDX-des-Ser excreted in urine or feces per time interval.
d-MPH, d-methylphenidate; LC-RAD, liquid chromatography coupled with radioactivity detection; ND, not detected; RA, ritalinic acid; SDX, serdexmethylphenidate; SDX-des-Ser, SDX without serine.

- The proposed overall metabolic pathway of SDX after oral administration is shown in **Figure 4**.

Figure 4. **Prominent metabolic pathways of stand-alone SDX.**

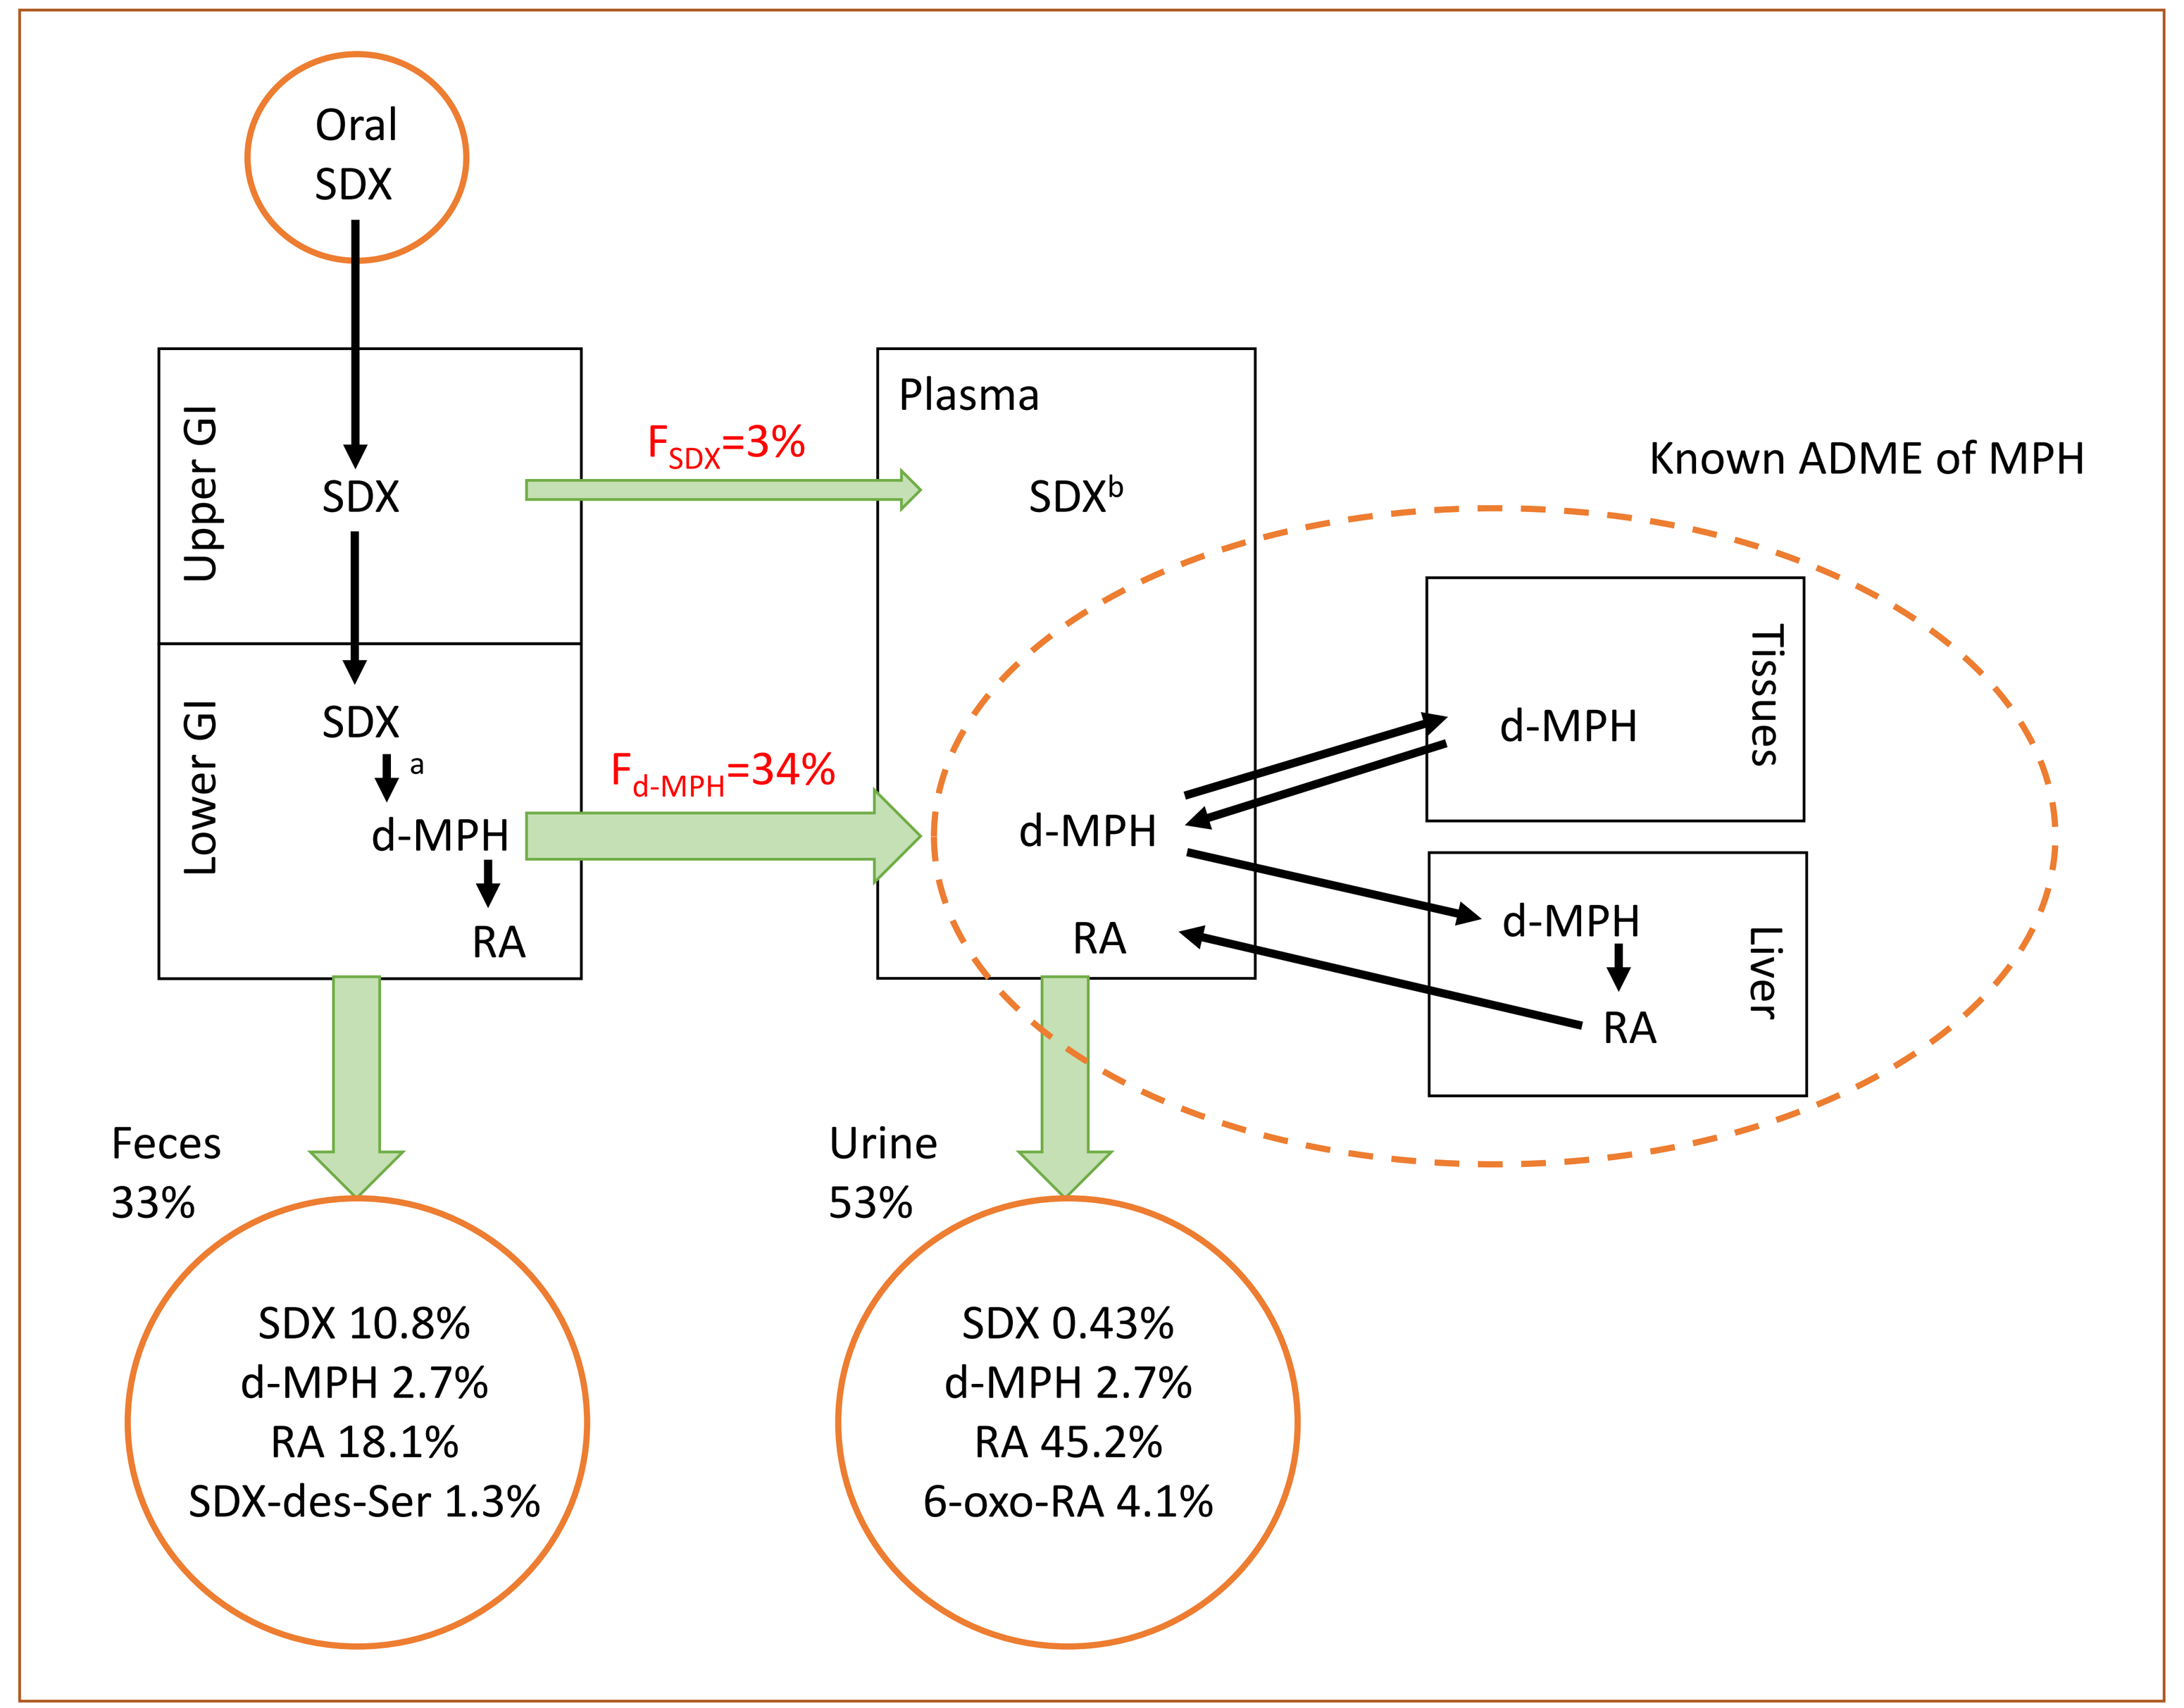
Oral
SDX
Upper GI
SDX
Lower GI
SDX
a
d-MPH
RA
F_{SDX}=3%
F_{d-MPH}=34%
Plasma
SDX[b]
d-MPH
RA
Known ADME of MPH
d-MPH
Tissues
d-MPH
RA
Liver
Feces
33%
SDX 10.8%
d-MPH 2.7%
RA 18.1%
SDX-des-Ser 1.3%
Urine
53%
SDX 0.43%
d-MPH 2.7%
RA 45.2%
6-oxo-RA 4.1%

Percentages are % of the administered SXD dose, on a molar basis.
[a]Mechanism unknown; location (lower GI) consistent with plasma T_{max} of SDX-derived d-MPH of 8 hours.
[b]Based on intravenous data with SDX chloride and in vitro data (not shown in this study), only a small amount of SDX in the systemic circulation and tissues is converted to d-MPH and RA (not shown); based on SDX volume of distribution, distribution of SDX into tissues is minimal.
ADME, absorption, distribution, metabolism, and excretion; d-MPH, methylphenidate; $F_{d\text{-}MPH}$ and F_{SDX}, absolute oral bioavailability of d-MPH and SDX, respectively (based on intravenous data and oral data from different studies); GI, gastrointestinal tract; RA, ritalinic acid; SDX, serdexmethylphenidate; SDX-des-Ser, SDX without serine.

Adverse Events

- Only 1 adverse event, mild headache, was reported by 1 subject.
- There were no serious adverse events.

CONCLUSIONS

- Overall, the mass balance data for oral administration of SDX chloride indicate no meaningful long-term retention of SDX or its metabolites in any tissues.
- The data show no indication of any major metabolic pathways of SDX other than conversion to d-MPH or evidence of new metabolic processes that affect the downstream metabolism of d-MPH. As for d-MPH, ritalinic acid was the major metabolite.

DISCLOSURES: RB, **ACB**, **SG**, and **TCM** are employees and shareholders of KemPharm, Inc. **MC** is an employee and shareholder of Corium, Inc.

This study was funded by KemPharm, Inc., Celebration, FL, USA.

Medical writing support for the development of this poster, under the direction of the authors, was provided by Gautam Bijur, PhD, and editing support by Anne Cooper, MA, both of Ashfield MedComms, an Ashfield Health company, and funded by Corium, Inc. (Grand Rapids, MI, USA).

The American Professional Society of ADHD and Related Disorders (APSARD) 2022 Annual Conference | January 14-16, 2022 | Tucson, AZ