F10

Dose-Proportionality and Steady-State Pharmacokinetics of KP415, an Investigational ADHD Product Containing Serdexmethylphenidate (SDX), a Novel Prodrug of d-Methylphenidate

Rene Braeckman[1], Sven Guenther[1], Travis C. Mickle[1], Andrew C. Barrett[1], Adam Smith[1], Wei Zang[2], Kathryn Roupe[2], Cynthia Zamora[2]

[1] KemPharm, Inc., Celebration, FL; [2] Worldwide Clinical Trials, San Antonio, TX

BACKGROUND

- KP415 is an investigational ADHD product containing serdexmethylphenidate (SDX), a novel prodrug of d-methylphenidate (d-MPH), co-formulated with d-MPH HCl in a fixed molar dose ratio of 70% SDX:30% d-MPH HCl
- KP415 has been designed as a once-daily, oral product that provides both early exposure to d-MPH and sustained exposure throughout the day
- Following a single KP415 dose, initial d-MPH exposure during the first few hours is governed by d-MPH HCl and subsequent sustained d-MPH exposure primarily by SDX conversion to d-MPH
- Determining the predictability of d-MPH exposure across the clinical dose range is critical for initial dose titration in patients with ADHD and for implementing dose adjustments over time

OBJECTIVE

- To assess the pharmacokinetics (PK) and dose-proportionality of single oral KP415 doses, as well as the steady-state PK after multiple-dose administration of the highest clinical daily dose

METHODS

Subjects and Study Design

- Eligible subjects were healthy males and non-pregnant, non-breastfeeding females 18-55 years of age
- This was a Phase 1, open-label, randomized, single-dose, 3-treatment, 3-period crossover study evaluating KP415 doses of:
 - 28 mg SDX/6 mg d-MPH HCl (“28/6 mg”, equimolar to 20 mg d-MPH HCl)
 - 42 mg SDX/9 mg d-MPH HCl (“42/9 mg”, equimolar to 30 mg d-MPH HCl),
 - 56 mg SDX/12 mg d-MPH HCl (“56/12 mg”, equimolar to 40 mg d-MPH HCl)
- All treatments (separated by a minimum 96-hour washout period) were administered orally under fasted conditions
- During the single-dose phase, pharmacokinetic sampling was conducted pre-dose and at 0.5, 1, 1.5, 2, 2.5, 3, 5, 7, 9, 12, 13, 24, 36, 48, 60 and 72 hours post-dose
- Following washout after the single-dose crossover phase, all subjects received 4 doses of 56/12 mg KP415 once daily for 4 days to evaluate the steady-state PK
- On Day 1 of the multiple-dose phase, PK sampling was conducted pre-dose and at 0.5, 1, 1.5, 2, 2.5, 3, 5, 7, 9, 12, 13, and 24 hours post-dose. The 24-hr postdose PK sample was taken before administration of the 2nd dose of study drug.
- On Days 2-3, PK sampling was conducted at 2, 8, and 24 hours postdose. The 24-hr post-dose PK samples were taken before administration of the 3rd and 4th doses of study drug
- On Day 4, PK sampling was conducted at 0.5, 1, 1.5, 2, 2.5, 3, 5, 7, 9, 12, 13, 24, 36, 48, 60 and 72 hours ±5 minutes post-dose
- Safety assessments were performed throughout the study

Statistical Analyses

- The following were among the pharmacokinetic parameters derived for d-MPH from the plasma concentration-time data under single- and multiple-dose conditions: C_{max}, C_{min} (at 24 hours post-dose), T_{max}, $AUC_{0\text{-}last}$, $AUC_{0\text{-}inf}$, $AUC_{0\text{-}24h}$, and $T_{1/2}$
- Evaluation of dose-proportionality and dose-linearity of C_{max}, $AUC_{0\text{-}last}$, $AUC_{0\text{-}24hr}$, and $AUC_{0\text{-}inf}$ for d-MPH was assessed using a power analysis with mixed effects (Smith 2000)
- A statistical comparison of the d-MPH PK parameters of exposure between the 1st and last (4th) dose was performed using an ANOVA model for the paired data after ln-transformation
- The least squares geometric means (LSGM) of the PK parameters for each dosing day (1st and 4th dose) were calculated, as well as point estimates and 90% Confidence Intervals (CI) for the Test to Reference ratios of geometric means

RESULTS

Subject Disposition

- A total of 24 subjects participated in the study and 23 completed both the single- and multiple-dose phases

Single-dose Phase

- After single-dose KP415 administration (**Figure 1**), d-MPH plasma concentrations increased rapidly for all dosage strengths, with peak concentrations (C_{max}) of 7.15, 9.88, and 13.85 ng/mL for the 28/6 mg, 42/9 mg, and 56/12 mg doses, respectively
- Median T_{max} occurred at 2.0 hours postdose for all 3 treatments
- Plasma d-MPH concentrations decreased gradually after C_{max}, with appreciable concentrations still apparent at 13 h post-dose.

Figure 1. Plasma d-MPH concentration-time profiles following single-dose administrations of KP415

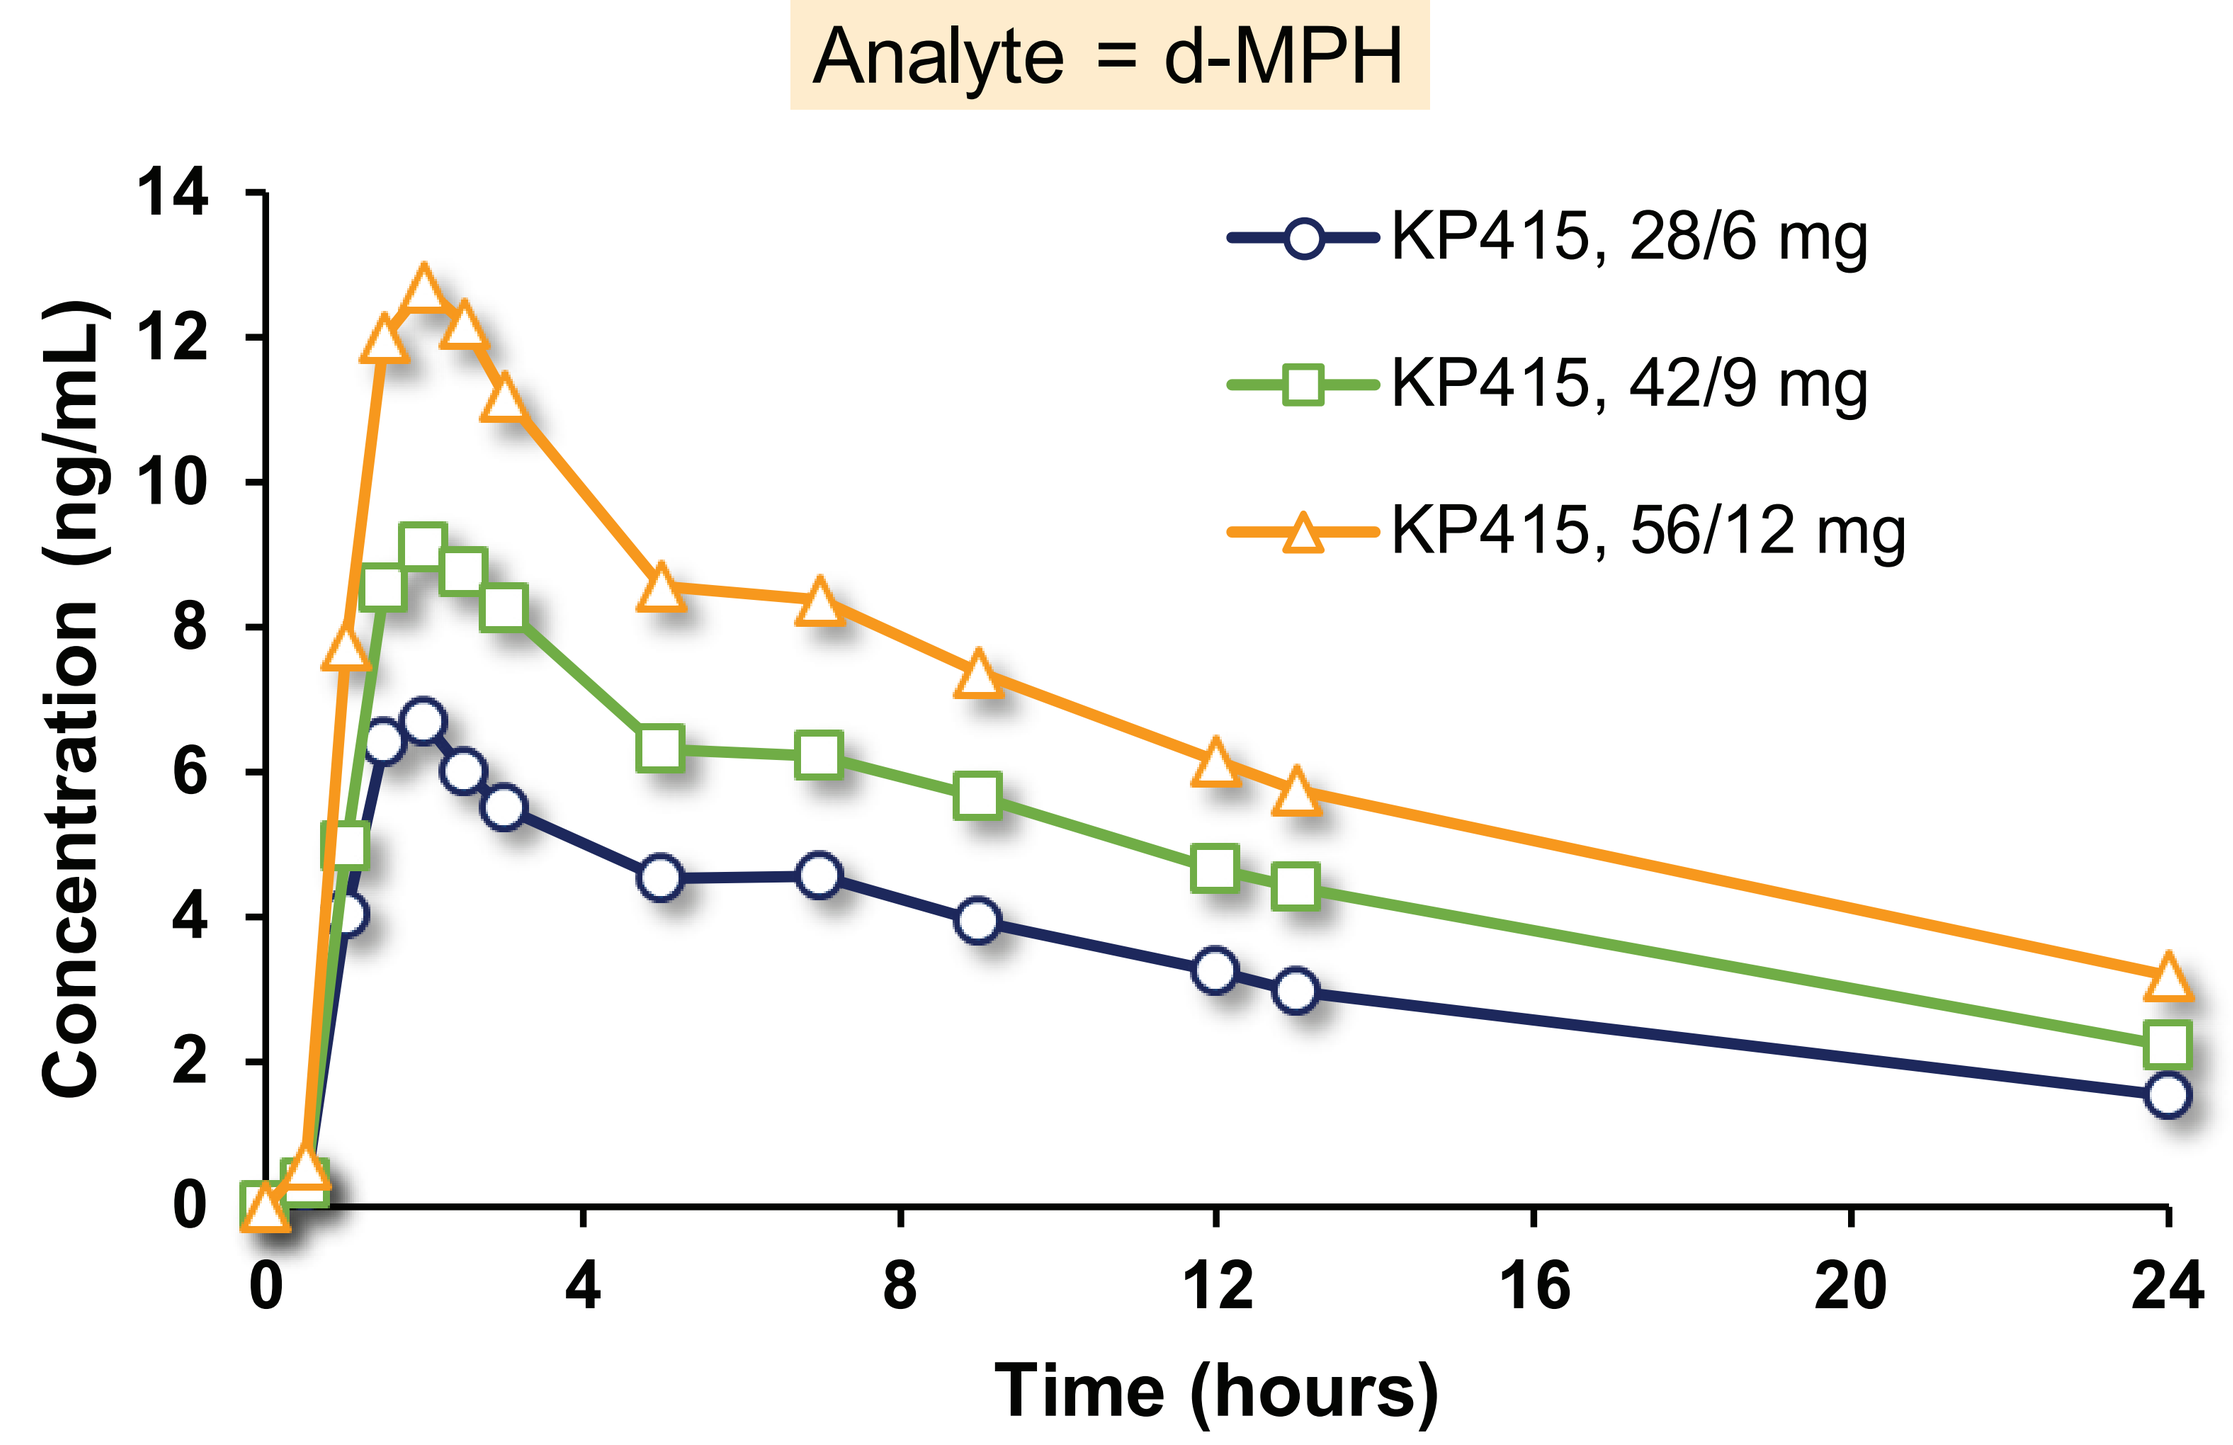
Analyte = d-MPH
KP415, 28/6 mg
KP415, 42/9 mg
KP415, 56/12 mg
Concentration (ng/mL)
Time (hours)

- As shown in **Figure 2**, after single dose administrations of 28/6 mg, 42/9 mg, and 56/12 mg KP415, d-MPH C_{max} and $AUC_{0\text{-}inf}$ values appeared to increase proportionally with an increase in KP415 dose

Figure 2. Regression analysis of plasma d-MPH C_{max} (top panel) and $AUC_{0\text{-}inf}$ (bottom panel) versus dose

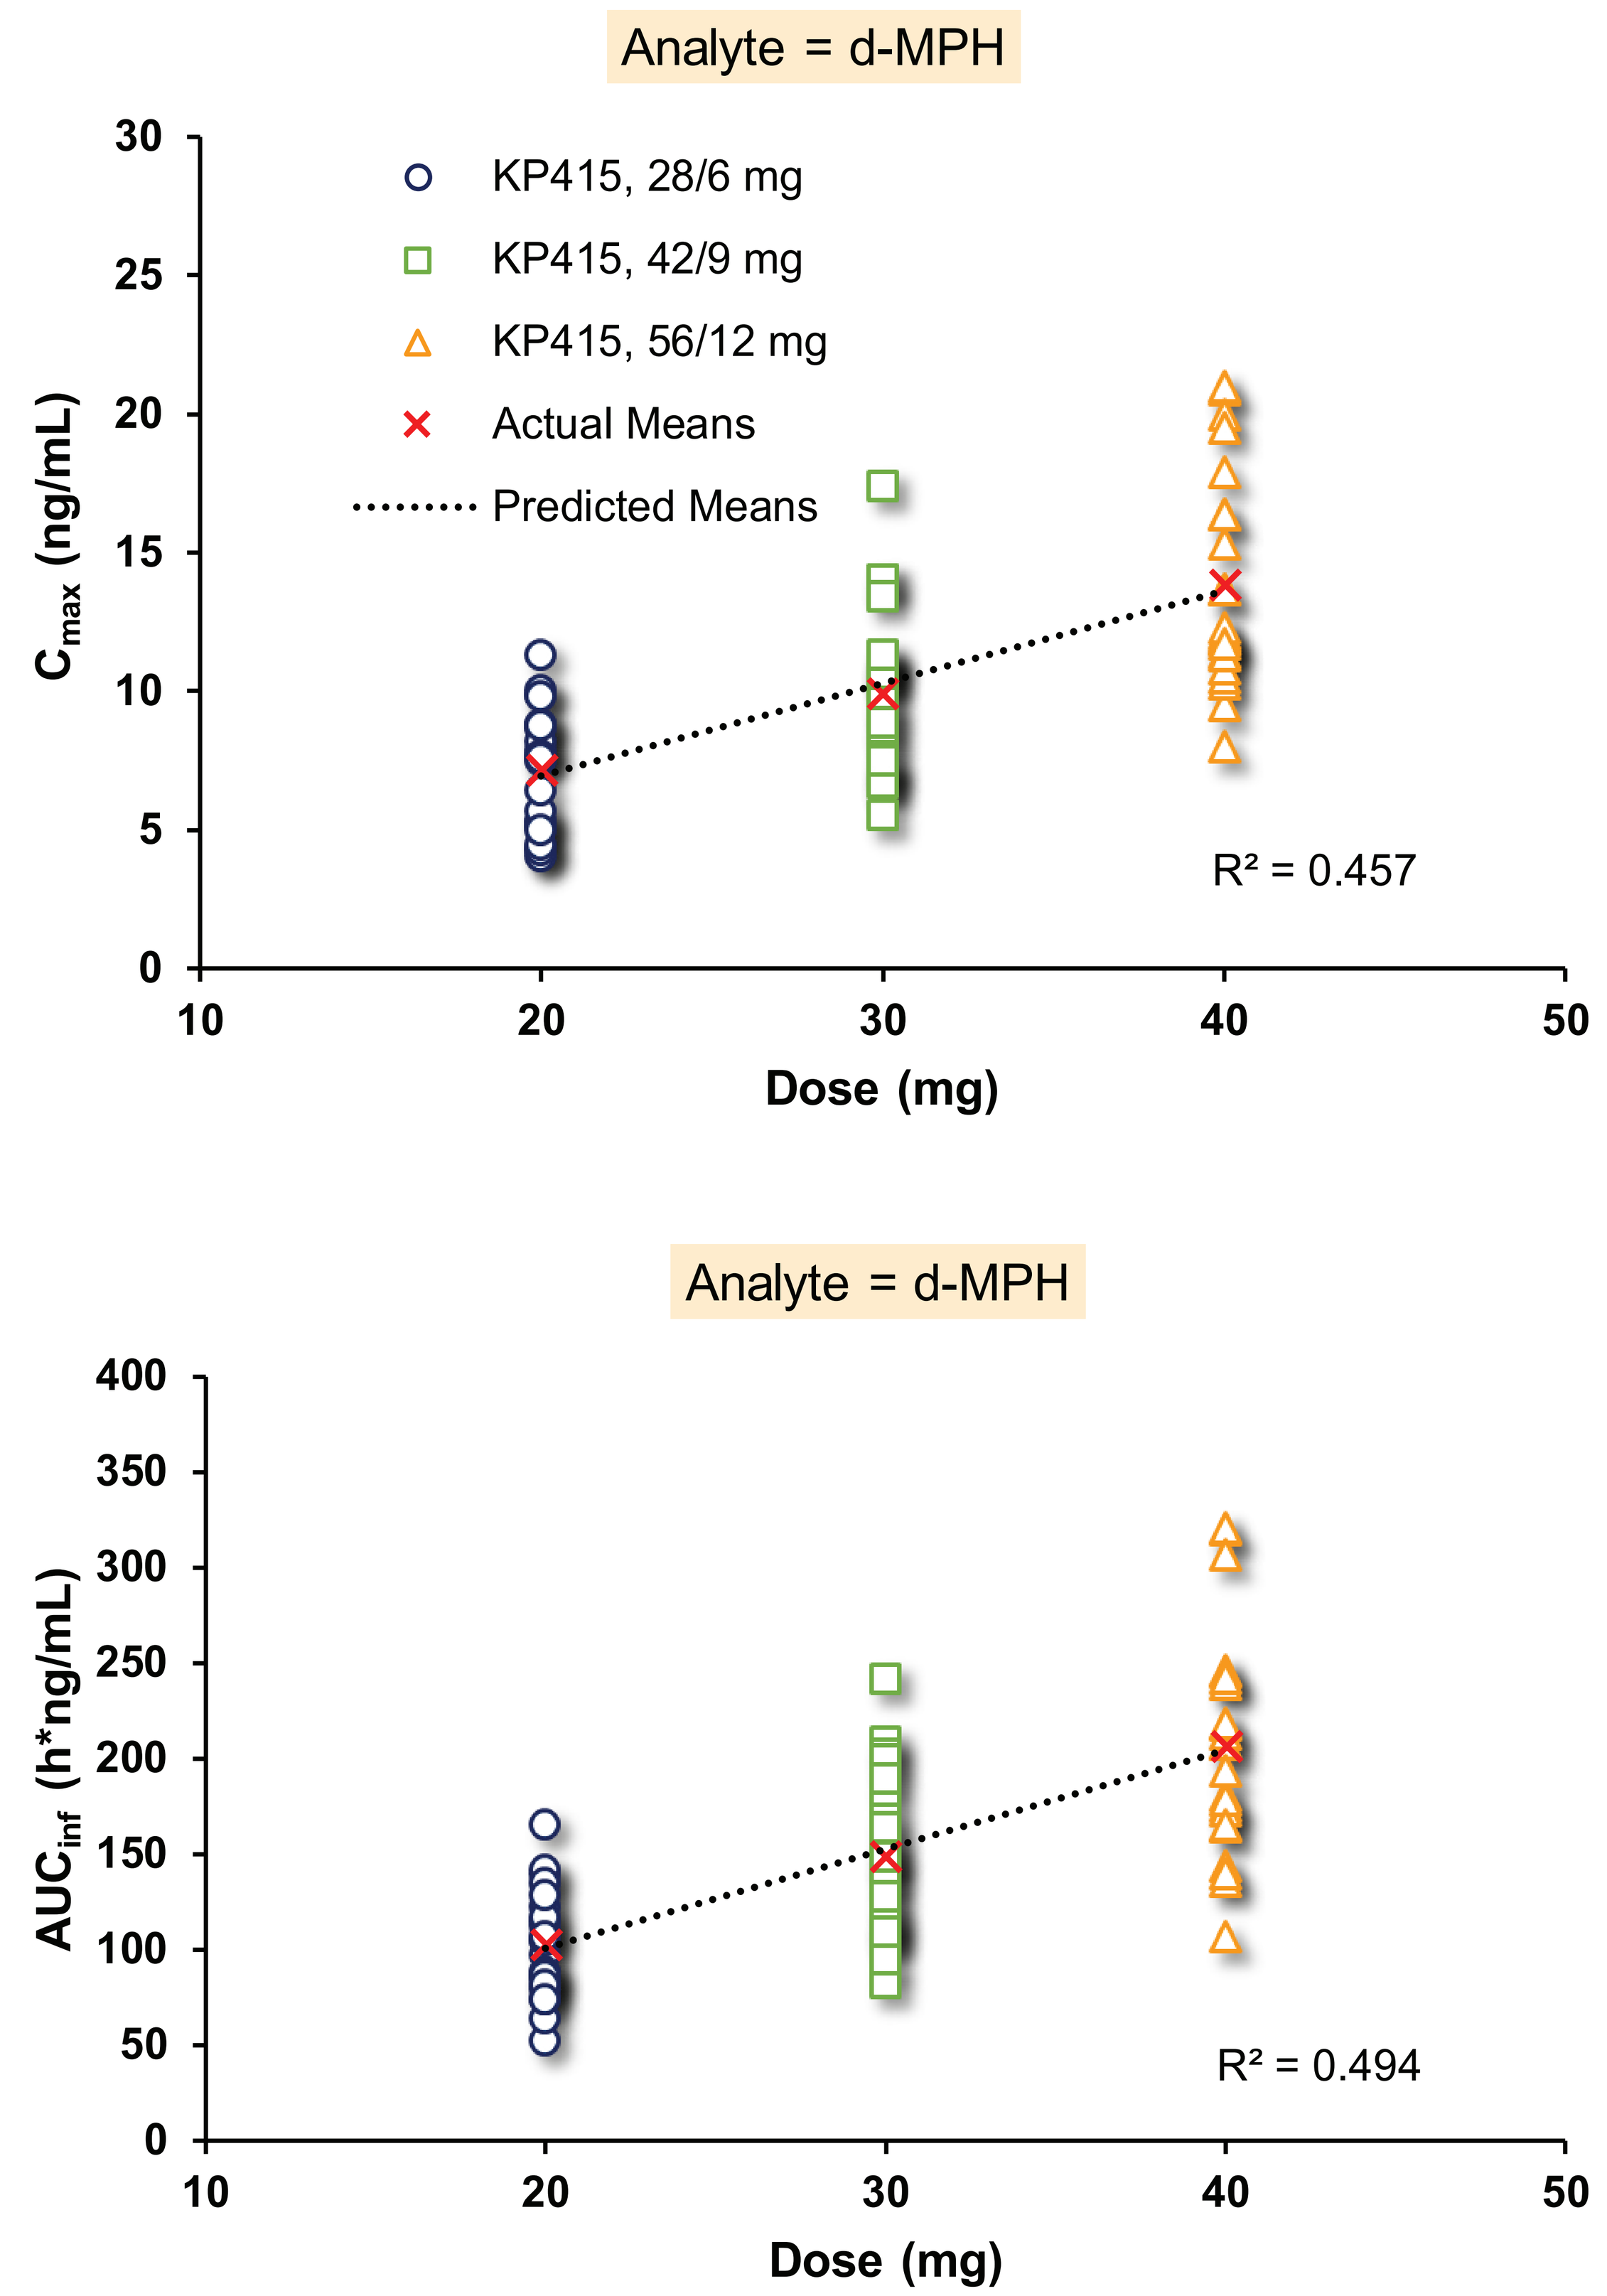
Analyte = d-MPH
KP415, 28/6 mg
KP415, 42/9 mg
KP415, 56/12 mg
Actual Means
Predicted Means
Cmax (ng/mL)
R² = 0.457
Dose (mg)
Analyte = d-MPH
AUCinf (h*ng/mL)
R² = 0.494
Dose (mg)

- **Table 1** shows results from the power analysis assessing dose-proportionality across the 3 doses administered
- The slope (β_1) for all parameters was close to 1 and the 90% confidence intervals estimated for the slope were contained within the lower and upper critical regions of 0.6781 and 1.3219, respectively, indicative of dose-proportionality
- Rho1 predicts a large theoretical range of dose proportionality, (approximately 6.5-fold [C_{max}] to 82.7-fold [$AUC_{0\text{-}inf}$]), suggesting that d-MPH increases proportionally with an increase in dose over the entire dose range administered for this study and beyond

Table 1. Assessment of dose-proportionality across KP415 doses of 28/6 mg, 42/9 mg, and 56/12 mg

Dependent Variable	Model Variable	Estimate (β_1)	90% CI Lower	90% CI Upper	Rho1[a]
$ln(C_{max})$	ln(Dose)	0.9625	0.8807	1.0444	6.4861
$ln(AUC_{0\text{-}24hr})$	ln(Dose)	0.9421	0.8921	0.9921	7.9077
$ln(AUC_{0\text{-}last})$	ln(Dose)	1.0403	0.9878	1.0928	11.0605
$ln(AUC_{0\text{-}inf})$	ln(Dose)	1.0021	0.9537	1.0505	82.7245

Power Model: $ln(PK)=ln(\beta_0)+\beta_1 * ln(Dose)+\varepsilon$, where PK is the pharmacokinetic parameter tested, $ln(\beta_0)$ is the y intercept, β_1 is the slope, and ε is an error term

[a] Maximal dose ratio for definitive proportionality. Rho1 was calculated as: Rho1=θ_H^(1/max(1-lower, upper-1)), in which θ_H=1.25.

Multiple-dose Phase

- **Figure 3** shows plasma d-MPH concentrations following single (Day 1) and multiple (Day 4) doses of KP415, 56/12 mg
- Mean maximum (C_{max}), minimum (C_{min}), and overall ($AUC_{0\text{-}24h}$) d-MPH exposure were approximately 35%, 12%, and 36% higher, respectively, after multiple doses (Dose 4) of KP415, 56/12 mg, relative to a single dose (Dose 1), as expressed by the least square geometric mean ratios of the respective parameters values (**Table 2**)
- Steady-state plasma concentrations, as assessed by trough concentrations of d-MPH, were achieved prior to the third dose (i.e., between the second and the third day of multiple dosing)
- There was no accumulation of the intact prodrug, SDX, during once-daily dosing

Figure 3. Mean plasma d-MPH concentration-time profiles after single- (Day 1) and multiple-dose (Day 4) administrations of KP415, 56/12 mg

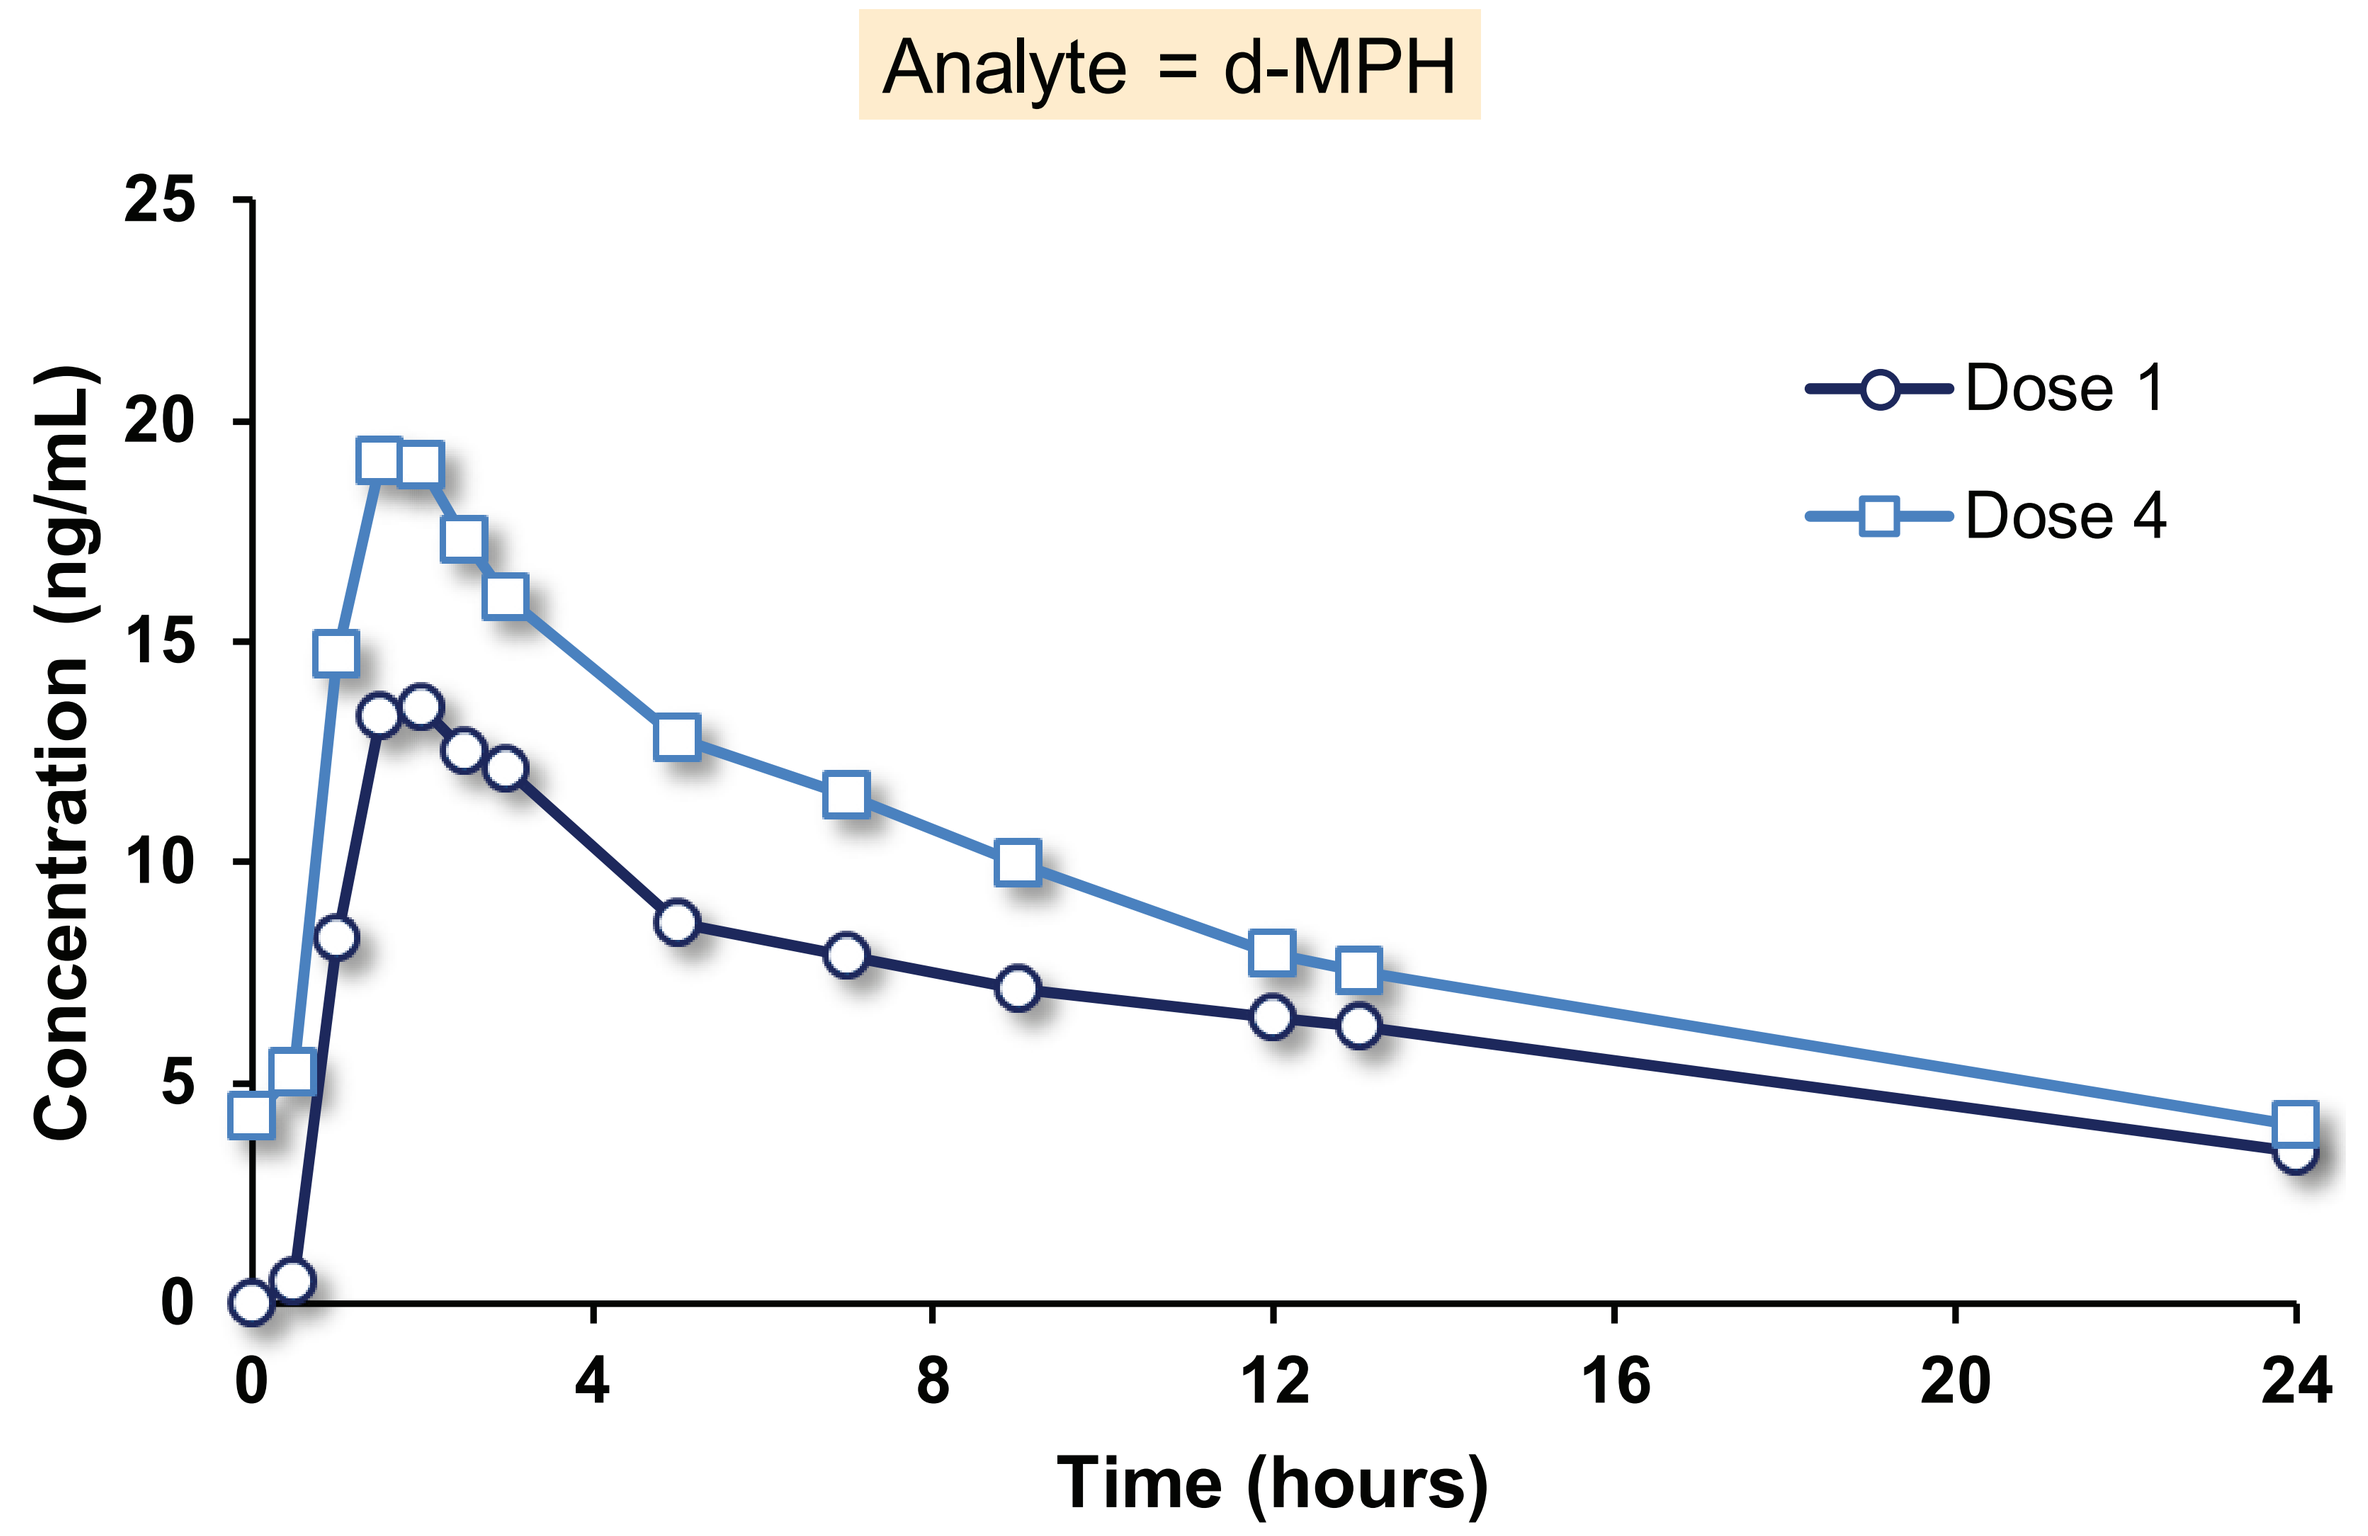
Analyte = d-MPH
Dose 1
Dose 4
Concentration (ng/mL)
Time (hours)

Table 2. Statistical analysis of natural log transformed systemic exposure of d-MPH comparing Dose 4 of KP415, 56/12 mg (Test) vs. Dose 1 of KP415, 56/12 mg (Reference)

Dependent Variable	GeoMean[a] Test	GeoMean[a] Ref	Ratio (%)[b] (Test/Ref)	90% CI Lower	90% CI Upper
$ln(C_{max})$	19.49	14.43	**134.99**	127.52	142.90
$ln(C_{min})$	3.61	3.22	**111.94**	102.23	122.58
$ln(AUC_{0\text{-}24hr})$	209.83	154.62	**135.71**	130.25	141.40

[a] Geometric Mean based on Least Squares Mean of log transformed parameter values
[b] Ratio (%) = Geometric Mean (Test)/Geometric Mean (Ref)
CI – Confidence Interval

Safety and Tolerability

- The most frequently reported adverse events (AEs) were dizziness, tachycardia, and feeling jittery, each reported by 3 (12.5%) subjects
- There were no serious AEs

CONCLUSIONS

- **KP415 demonstrated proportionality across the tested dose range with respect to rate and extent of d-MPH exposure. A power model predicted significant additional dose proportionality beyond the test range**
- **Following once-daily KP415 dosing, steady-state plasma concentrations of d-MPH were achieved prior to the third dose**
- **KP415 has the potential to provide a rapid onset and extended duration of therapeutic benefit, with predictable d-MPH exposure during titration and maintenance**

Disclosures

RB, SG, TCM, ACB, and AS are employees and shareholders of KemPharm, Inc. This study was funded by KemPharm, Inc., Celebration, FL. WZ, KAR, and CZ have no conflicts of interest to declare. Design support was provided by Research Triangle Graphics, LLC.

References

Smith BP, Vandenhende FR, DeSante KA, et al. Confidence interval criteria for assessment of dose proportionality. *Pharm Res*. 2000; 17: 1278–1283.

American Professional Society for ADHD and Related Disorders • January 18-20, 2019 • Washington, D.C.