F9

Single-dose Pharmacokinetics of KP415, an Investigational Product Containing the Prodrug Serdexmethylphenidate (SDX), in Children and Adolescents with ADHD

Rene Braeckman[1], Sven Guenther[1], Travis C. Mickle[1], Andrew C. Barrett[1], Adam Smith[1], Wei Zhang[2], Kathryn Roupe[2]

[1] KemPharm, Inc., Celebration, FL; [2] Worldwide Clinical Trials, San Antonio, TX

BACKGROUND

- KP415 is an investigational ADHD product designed to provide a rapid onset and extended duration of symptom improvement
- To achieve this profile, the drug product contains both d-MPH HCl, which provides rapid exposure to d-MPH following oral administration, and serdexmethylphenidate (SDX), a prodrug of d-methylphenidate that is gradually converted to d-MPH over many hours
- KP415 has been co-formulated with a fixed molar dose ratio of 70% SDX:30% d-MPH HCl
- In single- and multiple-dose PK studies with KP415 conducted in healthy adult volunteers, d-MPH exposure has been shown to be predictable across a relatively wide dose range (see Poster F10)

OBJECTIVE

- To examine the single-dose pharmacokinetics (PK) of KP415 in children and adolescents with ADHD, and secondarily, to determine the effect of body weight on the PK properties

METHODS

Subjects and Study Design

- Eligible subjects were males or females 6 to 17 years of age with a DSM-V primary diagnosis of ADHD (any subtype) per clinical evaluation
- Females of childbearing potential were required to remain abstinent or agree to use an effective and medically acceptable form of birth control
- This was a single-dose, single-period study of oral administration of KP415 capsules in children (6-12 years) and adolescents (13-17 years) with ADHD
- Following a standardized meal, eligible subjects (N=30) received treatments that were stratified into 3 age and 2 dose groups, whereby:
 - 6-8 year-olds (Cohort 1, n=10) received 28 mg SDX/6 mg d-MPH HCl ("28/6 mg")
 - 9-12 year-olds (Cohort 2, n=10) received 56 mg SDX/12 mg d-MPH HCl ("56/12 mg")
 - 13-17 year-olds (Cohort 3, total n=10) received either 28/6 mg (n=5) or 56/12 mg (n=5) assigned by a randomization schedule
 - The combined dose of SDX and d-MPH HCl in each capsule contained the same molar d-MPH as 20 and 40 mg d-MPH HCl, respectively
- Blood samples for PK were collected pre-dose and at 0.5, 1, 2, 4, 8, 10, 12, 13, 24, 36, and 48 hours post-dose
- Adverse events were continuously recorded, and safety assessments were conducted throughout the study.

Statistical Analyses

- The following plasma PK parameters for d-MPH were calculated: maximum concentration in plasma (C_{max}), time to reach maximum concentration (T_{max}), apparent elimination rate constant (λ_z), apparent terminal half-life ($T_{1/2}$), apparent clearance after oral administration (CL/F), apparent volume of distribution after oral administration (V_z/F), area under the concentration-time curve from time-zero to 24 hours postdose (AUC_{0-24}), AUC_{0-last}, AUC_{0-inf}, and partial AUCs
- Plasma concentrations and key PK parameters were normalized to the 56/12 mg dose
- For each subject, the dose-normalized plasma concentrations, key dose-normalized PK parameters (C_{max}, AUC_{0-last}, $AUC_{0-inf,}$ AUC_{0-24hr}), and CL/F and V_z/F were divided by body weight (in kg)
- The relationship between CL/F and V_z/F for d-MPH versus body weight was evaluated using a nonlinear regression analysis according to the following equation:

$$P = P_{70} * (BW/70\ Kg)^{exp}$$

 - Where P is the individual PK parameter (either CL/F and V_z/F), BW is the individual body weight in kg, the coefficient P70 is the PK parameter standardized for a 70-kg body weight, and exp is the allometric scaling exponent
- Geometric means for d-MPH CL/F (L/h/kg) and V_z/F (L/kg) were calculated with the 95% CI around the geometric means. If the 95% CIs fell within 60% and 140% of the geometric means, it was concluded that the study was sufficiently powered

RESULTS

Subject Disposition and Demographics

- All 31 subjects who enrolled completed the study, and 30 subjects were included in the PK Population (1 subject was excluded due to out-of-window PK sample collections caused by blood draw difficulties)
- **Table 1** provides demographic characteristics for each cohort

Table 1. **Demographic Characteristics – PK Population (N=30)**

Parameter	Cohort 1 6-8 years 28/6 mg (N=10)	Cohort 2 9-12 years 56/12 mg (N=10)	Cohort 3 13-17 years 28/6 mg (N=5)	Cohort 3 13-17 years 56/12 mg (N=5)
Age (years) at Dosing Day, Mean (SEM)	7.0 (0.30)	10.1 (0.38)	14.0 (0.77)	14.0 (0.32)
Gender				
Male, N (%)	7 (70.0)	9 (90.0)	2 (40.0)	2 (40.0)
Female, N (%)	3 (30.0)	1 (10.0)	3 (60.0)	3 (60.0)
Weight (kg) at Screening, Mean (SEM)	29.33 (1.51)	39.75 (2.55)	65.68 (5.11)	65.02 (2.92)
BMI (kg/m²) at Screening, Mean (SEM)	17.83 (0.50)	18.66 (0.95)	24.64 (1.37)	25.70 (0.79)

Pharmacokinetic Data

- **Figure 1** shows mean concentration-time profiles for d-MPH across cohorts, and **Figure 2** shows the same data dose-normalized and scaled by body weight
- While the overall shape of the PK curves was similar across cohorts, d-MPH concentrations over the dosing interval were generally higher in younger children (**Figure 1**)
- Median T_{max} occurred at 4 hours in all cohorts
- When the plasma concentrations were dose-normalized and scaled by body weight, the 4 mean PK curves were nearly superimposable (**Figure 2**)

Figure 1. **Mean plasma d-MPH concentration-time profiles following KP415 administration**

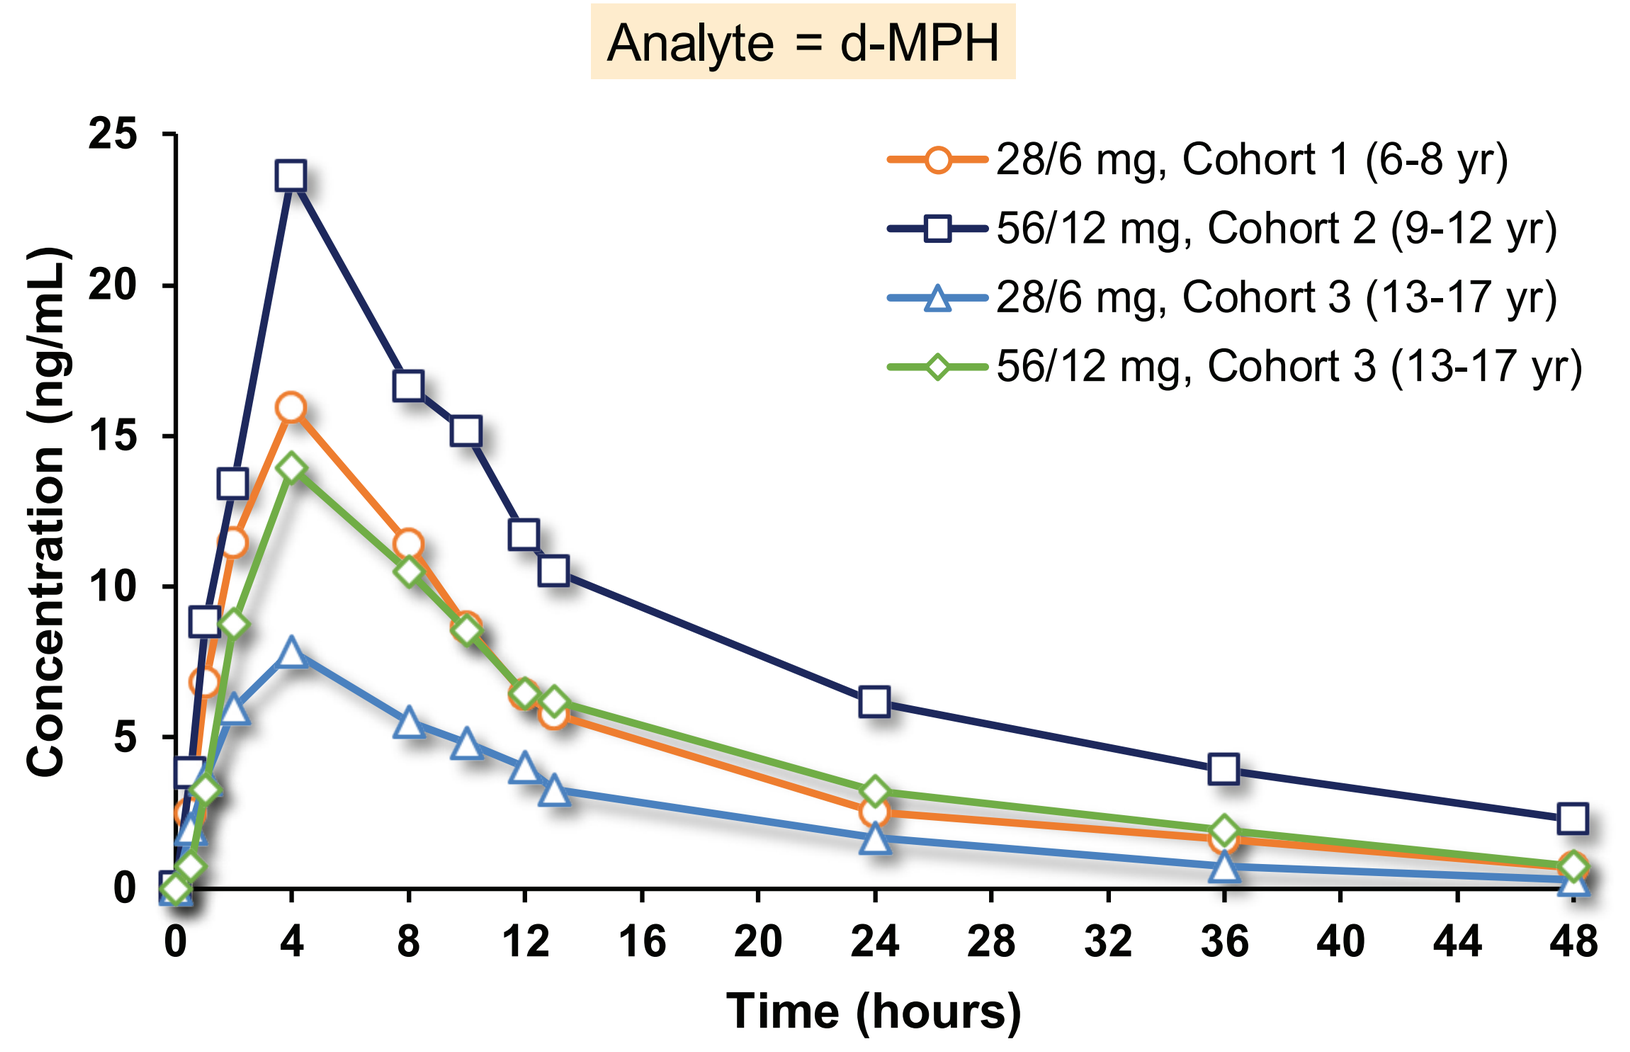
Analyte = d-MPH
28/6 mg, Cohort 1 (6-8 yr)
56/12 mg, Cohort 2 (9-12 yr)
28/6 mg, Cohort 3 (13-17 yr)
56/12 mg, Cohort 3 (13-17 yr)
Concentration (ng/mL)
Time (hours)

Figure 2. **Mean dose-normalized/body weight-scaled plasma d-MPH concentration-time profiles following KP415 administration**

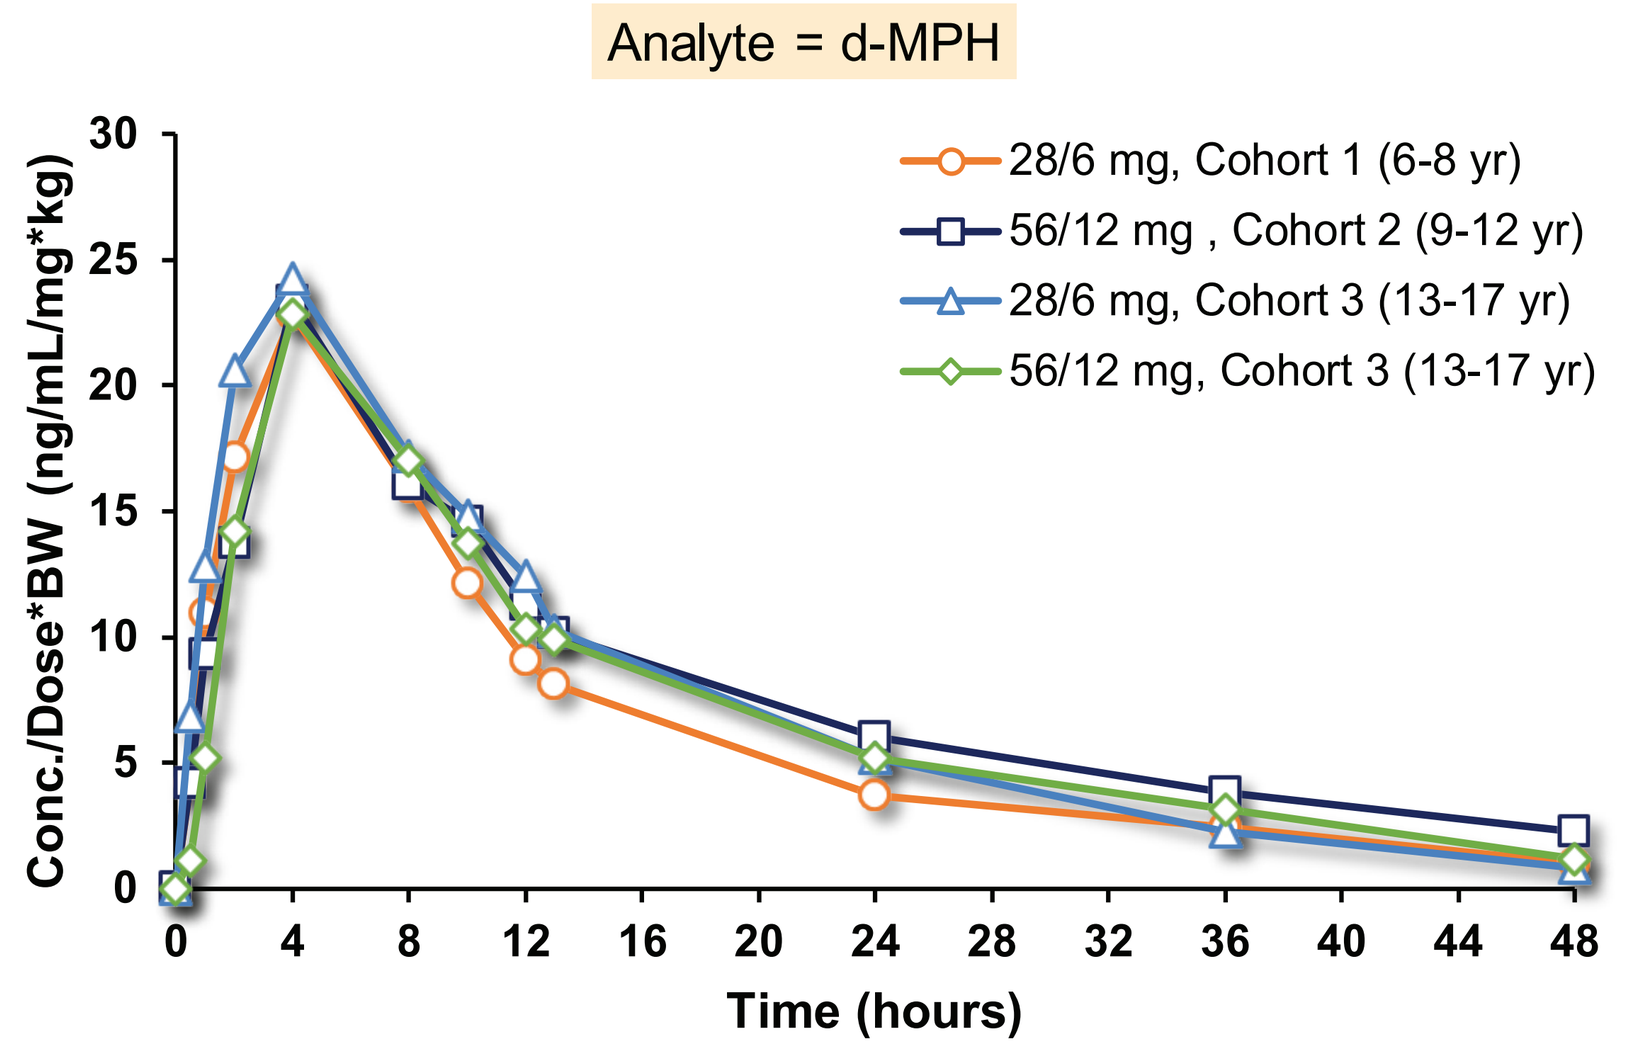
Analyte = d-MPH
28/6 mg, Cohort 1 (6-8 yr)
56/12 mg , Cohort 2 (9-12 yr)
28/6 mg, Cohort 3 (13-17 yr)
56/12 mg, Cohort 3 (13-17 yr)
Conc./Dose*BW (ng/mL/mg*kg)
Time (hours)

- **Table 2** shows derived PK parameters for non-normalized, original data, dose-normalized data, and dose-normalized/body weight-scaled data

- Dose-normalized (to the 56/12 mg dose) peak and overall exposure to d-MPH was highest in Cohort 1 (C_{max} = 34.4 ng/mL, AUC_{0-24} = 362.0 h*ng/mL), followed by Cohort 2 (C_{max} = 25.9 ng/mL, AUC_{0-24} = 294.1 h*ng/mL), and lowest in Cohort 3 (C_{max} = 17.8 ng/mL and 14.0 ng/mL, for the low and high doses, respectively; AUC_{0-24} = 195.0 ng/mL and 171.1 h*ng/mL, respectively)
- When scaled by body weight, mean dose-normalized C_{max} and AUC_{0-24} values were similar across cohorts (C_{max} range across the 3 cohorts: 25.0 – 25.3 ng/mL/(mg/kg); $AUC_{0-24\ hr}$ range across cohorts: 259.4 – 291.8 (h*ng/mL/(mg/kg))
- Clearance (CL/F) values were lower in Cohorts 1 and 2 (96.85 and 97.44 L/h, respectively) than Cohort 3 (170.3 L/h for low dose and 172.3 L/h for high dose), although when adjusted for weight differences, clearance values were similar

Table 2. **Non-normalized, dose-normalized, and dose-normalized/body weight-scaled plasma pharmacokinetic parameters of d-MPH across cohorts**

Parameter, Mean (SD)	Cohort 1 6-8 years 28/6 mg (N=10)	Cohort 2 9-12 years 56/12 mg (N=10)	Cohort 3 13-17 years 28/6 mg (N=5)	Cohort 3 13-17 years 56/12 mg (N=5)
Non-Normalized				
C_{max} (ng/mL)	17.2 (5.0)	25.9 (9.7)	8.9 (3.2)	14.0 (1.7)
$AUC_{0-24\ hr}$ (h*ng/mL)	181.0 (61.6)	294.1 (98.2)	97.5 (29.7)	171.1 (19.4)
AUC_{0-last} (h*ng/mL)	219.8 (72.3)	391.6 (129.9)	116.5 (39.2)	217.0 (24.4)
AUC_{0-inf} (h*ng/mL)	228.2 (79.4)	459.7 (145.4)	125.3 (41.0)	234.6 (25.6)
CL/F (L/h)	96.9 (33.1)	97.4 (38.1)	170.3 (41.5)	172.3 (20.5)
V_z/F (L)	1680 (463)	2659 (1433)	2487 (771)	2718 (864)
Dose-Normalized				
DN-C_{max} (ng/mL)	34.4 (10.0)	25.9 (9.7)	17.8 (6.4)	14.0 (1.7)
DN-$AUC_{0-24\ hr}$ (h*ng/mL)	362.0 (123.2)	294.1 (98.2)	195.0 (59.4)	171.1 (19.4)
DN-AUC_{0-last} (h*ng/mL)	439.7 (144.7)	391.6 (129.9)	233.1 (78.3)	217.0 (24.4)
DN-AUC_{0-inf} (h*ng/mL)	456.4 (158.7)	459.7 (145.4)	250.7 (81.9)	234.6 (25.6)
Dose-Normalized/Scaled by Body Weight				
Dose/Weight-C_{max} (ng/mL/(mg/kg))	25.0 (6.9)	25.3 (9.8)	27.8 (5.0)	22.8 (4.4)
Dose/Weight-$AUC_{0-24\ hr}$ (h*ng/mL/(mg/kg))	259.4 (66.9)	282.6 (83.0)	306.9 (27.3)	276.8 (26.9)
Dose/Weight-AUC_{0-last} (h*ng/mL/(mg/kg))	316.2 (82.8)	375.5 (106.1)	364.9 (36.2)	350.9 (37.2)
Dose/Weight-AUC_{0-inf} (h*ng/mL/(mg/kg))	328.3 (90.2)	443.5 (131.8)	393.0 (39.4)	379.6 (41.7)
Dose/Weight-CL/F (L/h/kg)	3.4 (1.4)	2.5 (0.7)	2.6 (0.3)	2.7 (0.3)
Dose/Weight-V_z/F (L/kg)	57.5 (14.5)	66.0 (32.0)	37.6 (8.4)	41.8 (13.6)

- A nonlinear regression model evaluating allometric scaling indicated a moderate correlation (R^2=0.628) between d-MPH clearance (CL/F) and body weight and a weak correlation (R^2=0.200) between d-MPH volume of distribution (V_z/F) and body weight (**Figure 3**)

Figure 3. **Nonlinear regression model evaluating allometric scaling**

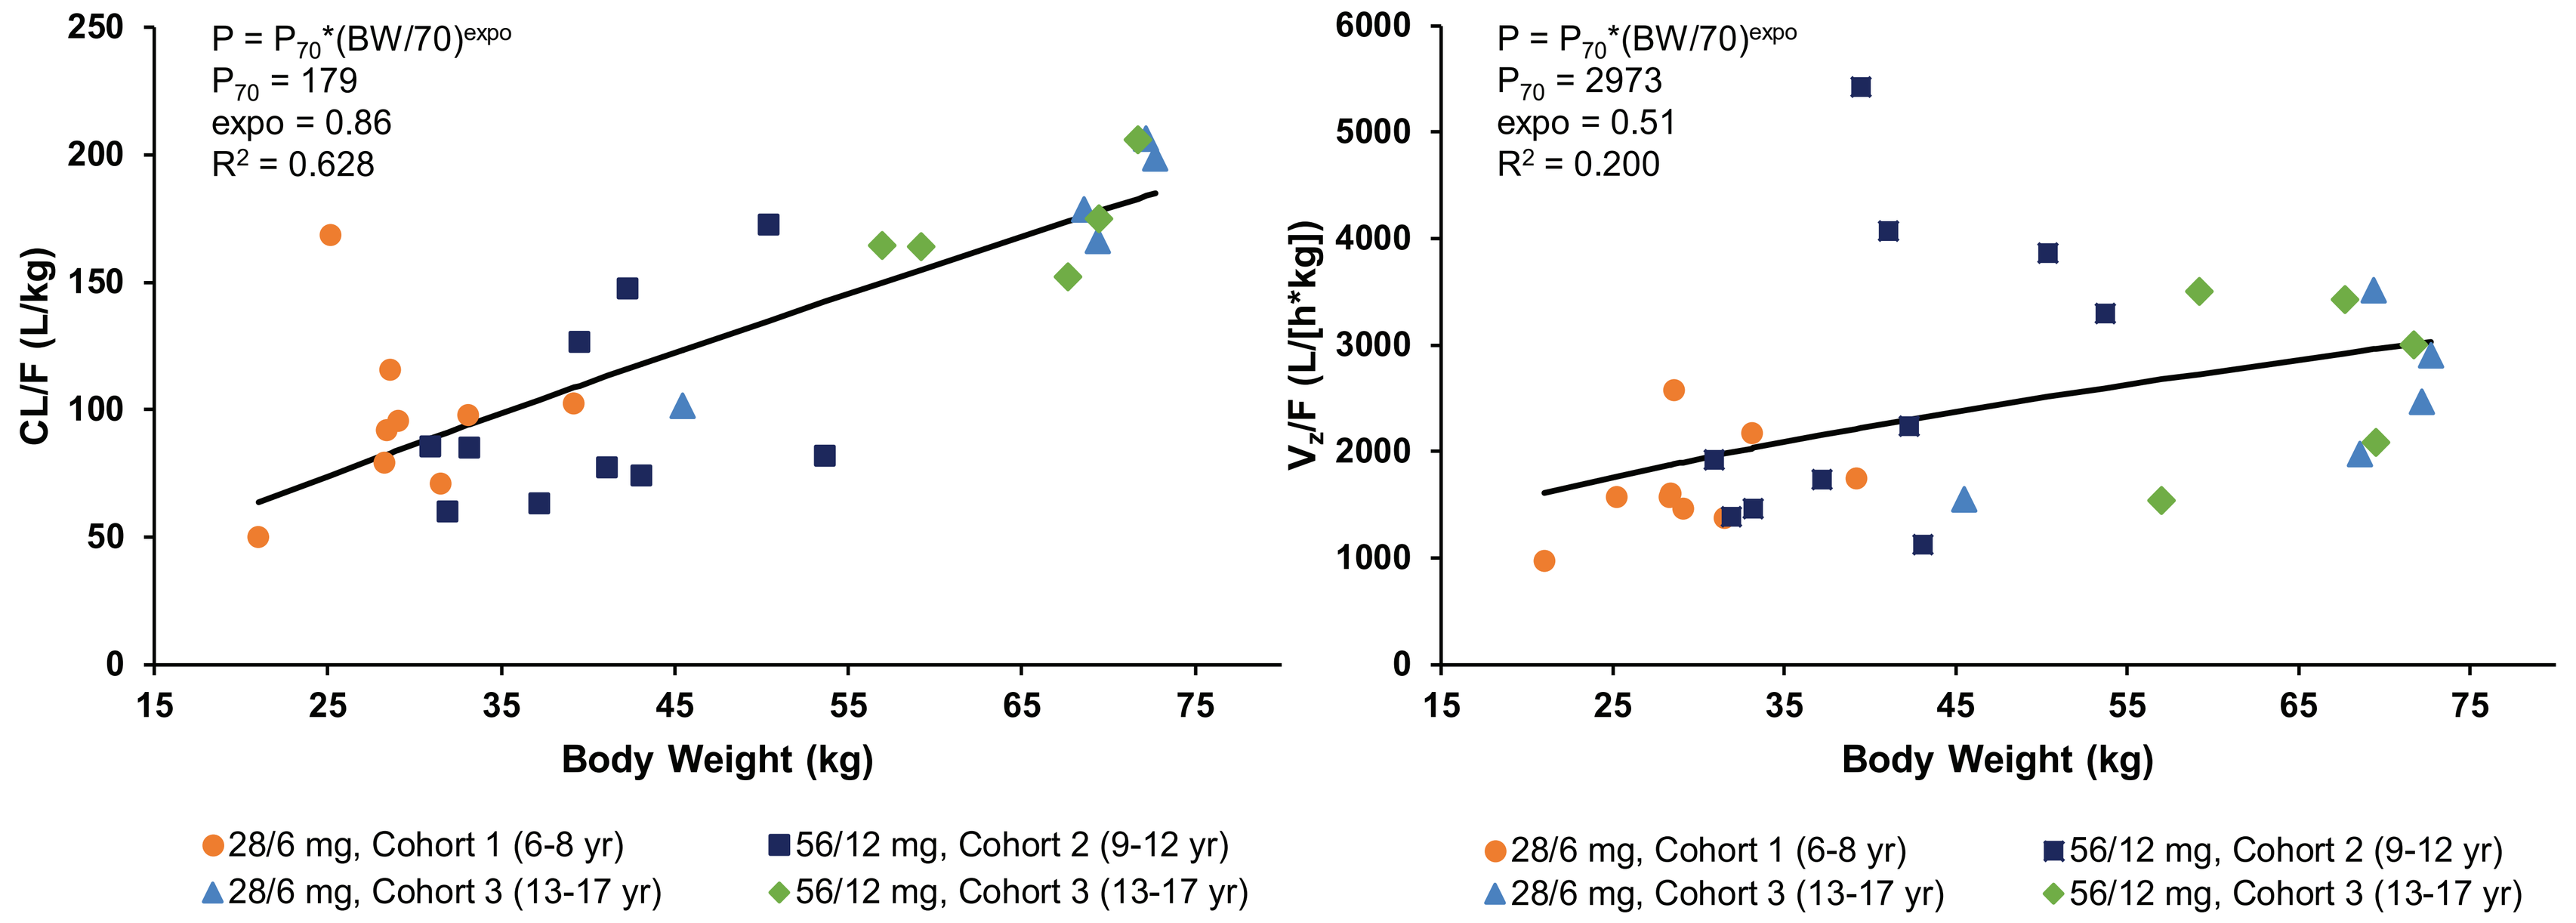
P = P70*(BW/70)expo
P70 = 179
expo = 0.86
R² = 0.628
CL/F (L/kg)
Body Weight (kg)
P = P70*(BW/70)expo
P70 = 2973
expo = 0.51
R² = 0.200
Vz/F (L/[h*kg])
Body Weight (kg)
28/6 mg, Cohort 1 (6-8 yr)
56/12 mg, Cohort 2 (9-12 yr)
28/6 mg, Cohort 3 (13-17 yr)
56/12 mg, Cohort 3 (13-17 yr)

- **Figure 4** shows that the 95% CIs of the geometric means for d-MPH CL/F and V_z/F were within the target range of 60% to 140% of each respective geometric mean for all cohorts

Figure 4. **95% confidence intervals of weight-normalized clearance (CL/F/BW) and volume of distribution (V_z/F/BW)**

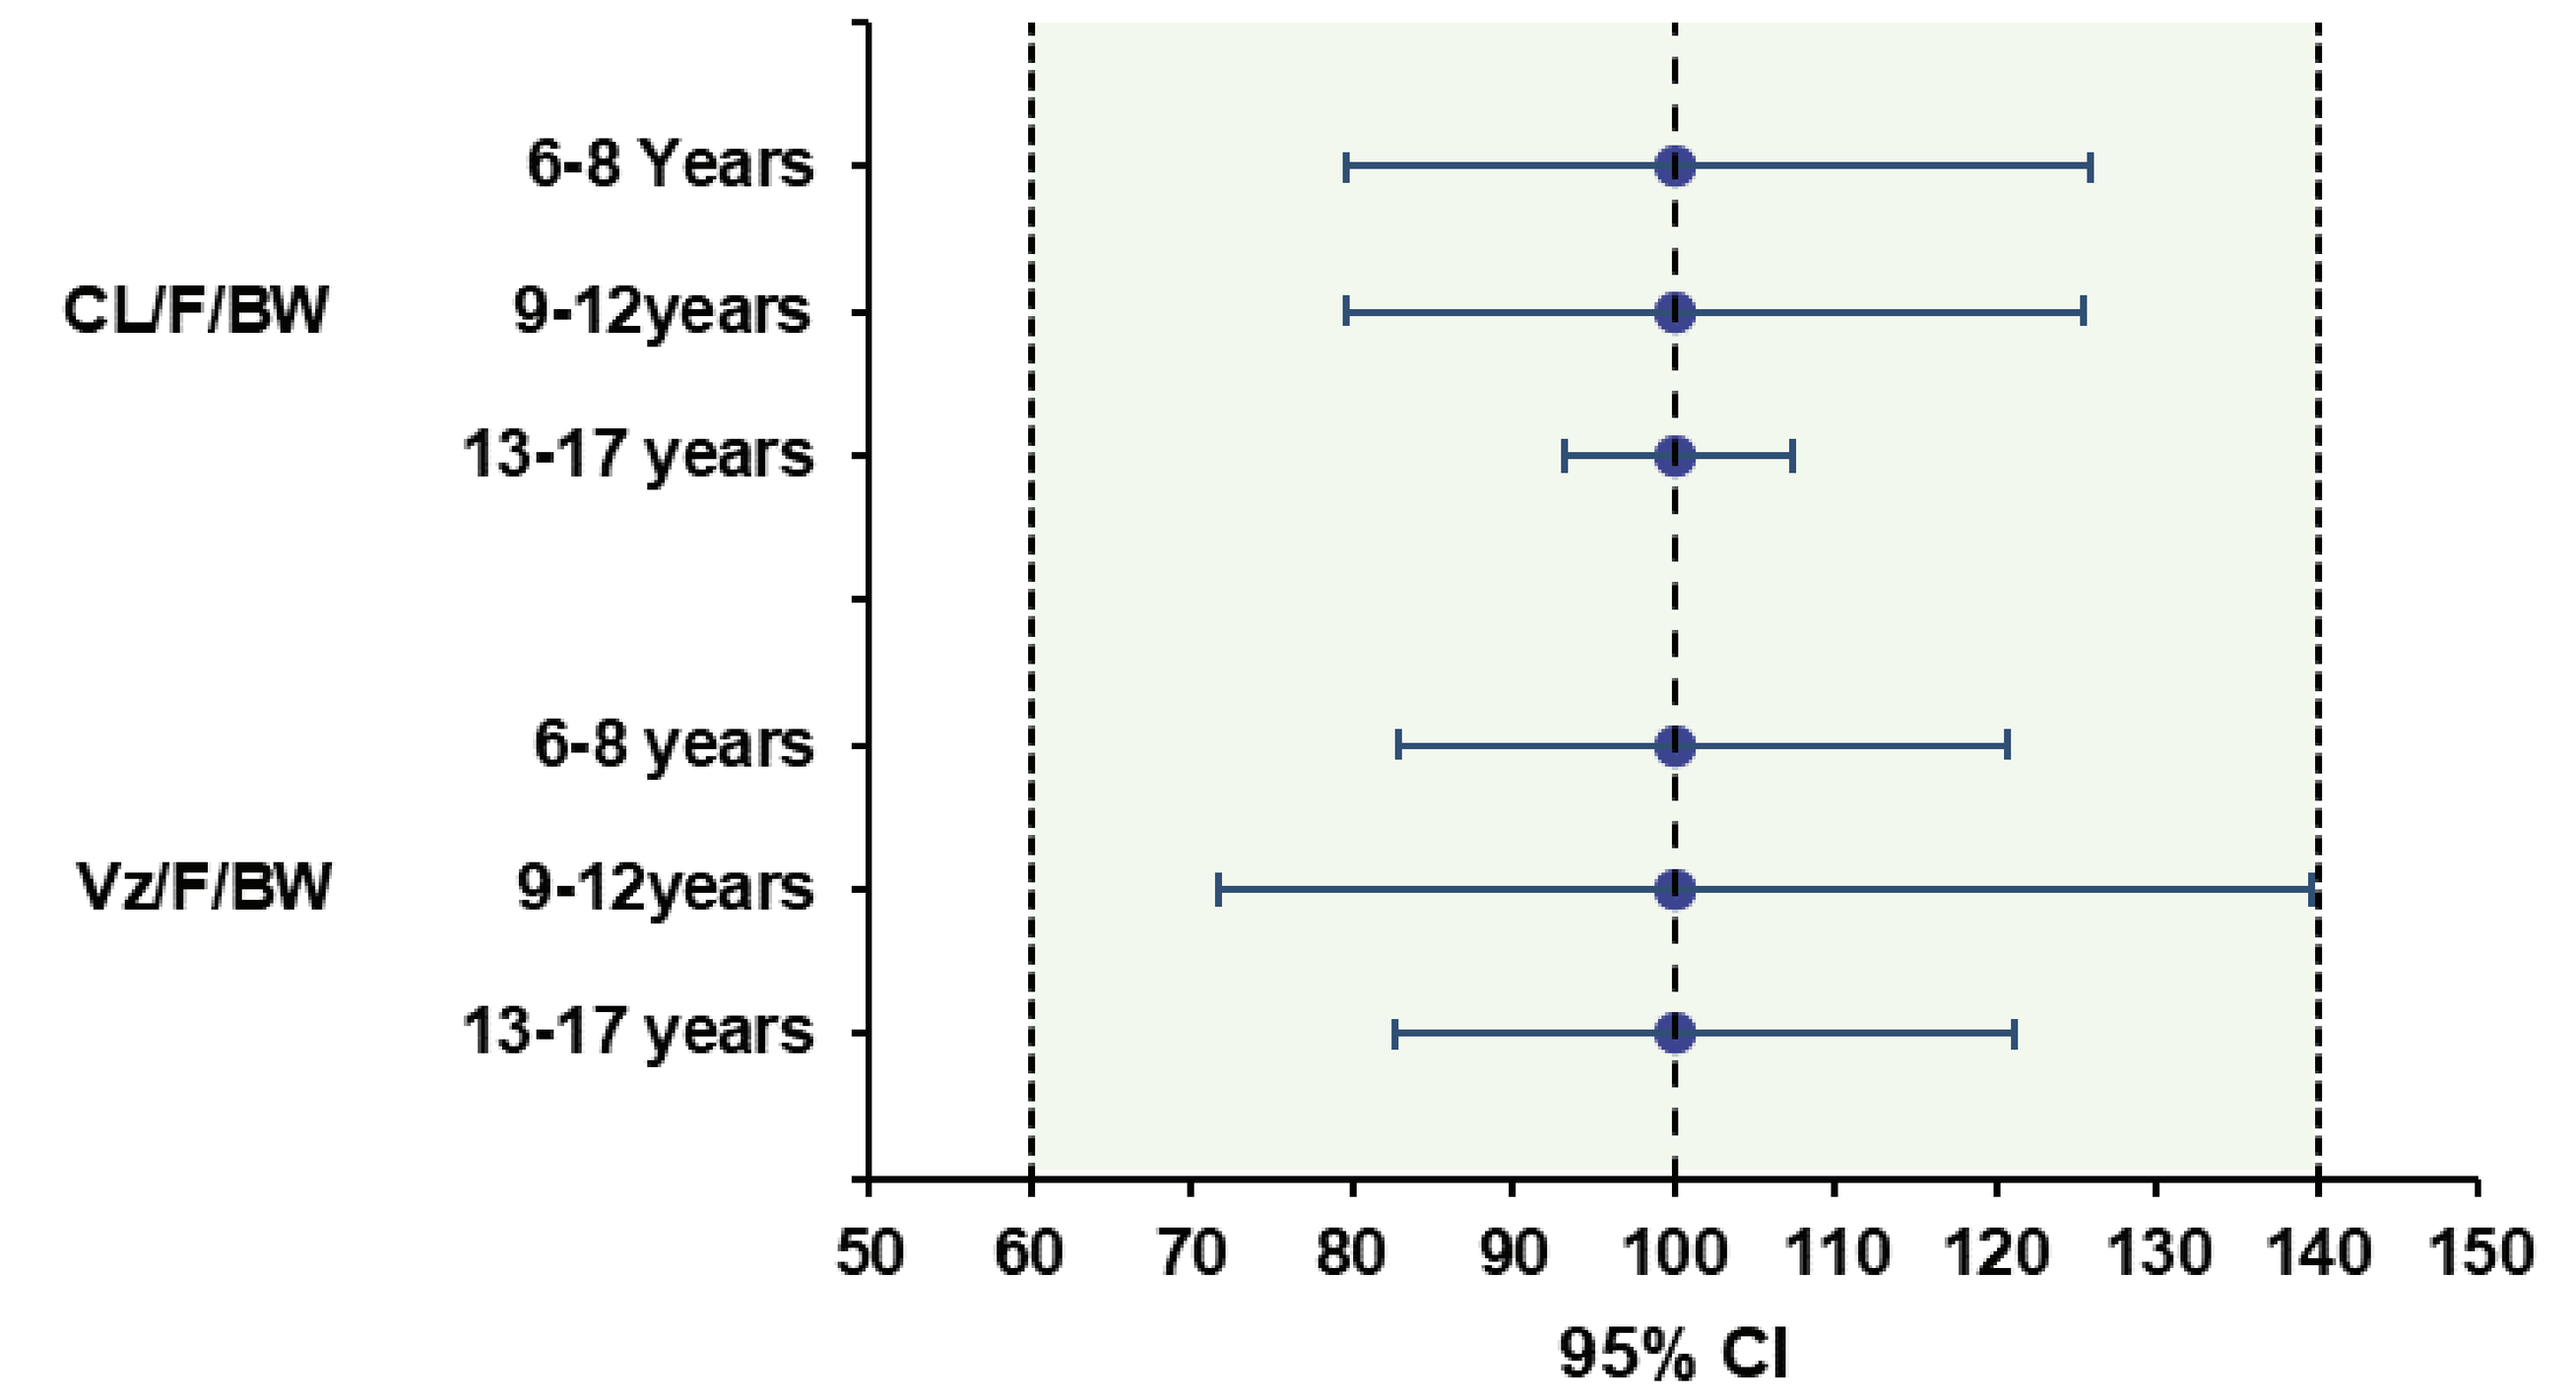
CL/F/BW
6-8 Years
9-12years
13-17 years
Vz/F/BW
6-8 years
9-12years
13-17 years
95% CI

- A total of 5 subjects reported AEs (pruritus, upper respiratory tract infection, pyrexia, upper abdominal pain, and headache), none of which were serious or led to discontinuation

CONCLUSIONS

- **Systemic dose-normalized exposure to d-MPH following oral administration of KP415 was generally higher in younger children who typically have lower body weights**
- **Age-dependent differences in d-MPH exposure appear to be due to lower clearance in younger children which is, in turn, primarily related to intrinsic body weight differences across the age spectrum examined in this study**
- **These findings are consistent with prior studies of methylphenidate products[2,3] and indicate that KP415 produces predictable, body weight-dependent exposure to d-MPH in pediatric subjects**

Disclosures

RB, SG, TCM, and ACB are employees and shareholders of KemPharm, Inc.

This study was funded by KemPharm, Inc., Celebration, FL. WZ and KAR have no conflicts of interest to declare.

Design support was provided by Research Triangle Graphics, LLC.

References

1. Mahmood I. *Ther Drug Monit*. 2007; 29(3):271-8.
2. Wigal SB et al. *J Child Adolesc Psychopharmacol.* 2007; 17:153–164.
3. Childress A, et al. *J Child Adolesc Psychopharmacol.* 2016 Aug; 26(6):505-12.

American Professional Society of ADHD and Related Disorders (APSARD) • January 18-20, 2019 • Washington, DC