Assessment of the Intravenous Abuse Potential Of Serdexmethylphenidate (SDX), a Novel, Investigational Prodrug of D-Methylphenidate: Evidence from Nonclinical and Clinical Studies

157

ACNP Member Sponsor:
Edward M. Sellers

Megan J. Shram[1], Sven Guenther[2], Travis C. Mickle[2], Rene Braeckman[2], Adam Smith[2], Andrew C. Barrett[2]

[1] Altreos Research Partners, Inc., Toronto, Canada [2] KemPharm, Inc., Celebration, FL

BACKGROUND

- Serdexmethylphenidate (SDX) is a novel prodrug of d-methylphenidate (d-MPH) that has been incorporated into two investigational products (KP415 and KP484) for the treatment of Attention-Deficit Hyperactivity Disorder (ADHD)
- As a prodrug that requires enzymatic cleavage to become active, SDX was designed to provide an extended duration of d-MPH exposure when taken orally[1] but to release d-MPH less efficiently if manipulated for the purposes of abuse via non-oral routes (e.g., intranasal, intravenous [IV]) to achieve a rapid and intense "high" [2,3]
- A comprehensive evaluation of abuse potential is therefore critical for understanding the risk-benefit profile of novel stimulant-like drugs such as SDX

OBJECTIVE

- To evaluate the performance of SDX in a series of in vitro and in vivo studies that are relevant for understanding its IV abuse potential

METHODS

Primary and Secondary Pharmacology

- The primary pharmacology of SDX (0.03 to 10 μM) was evaluated in competition binding assays using human recombinant dopamine (DAT), norepinephrine (NET), and serotonin (SERT) transporters
- For experiments with human recombinant DAT and NET, aliquots were incubated with [^{125}I] RTI-55, respectively; for experiments with human recombinant SERT, aliquots were incubated with [^{3}H] paroxetine
- Potential binding of SDX (10 μM) to a panel of 68 molecular targets was also evaluated to detect potential adverse activity, unexpected activity, and relative selectivity and specificity
- A significant response was defined as ≥50% inhibition of radioligand binding

In vitro Metabolic Stability

- The metabolic stability of SDX (10 μM) was evaluated in human whole blood, plasma, and human liver S9 fractions
- Triplicate samples were collected at select time points after 5 to 90 minutes of incubation
- UPLC-MS/MS methods were used for analysis of the incubation samples
- The peak area ratio (PAR) of SDX to an internal standard (IS) was determined at each time point. The percent of SDX remaining was determined by comparing the PAR at each time point to the PAR at time zero, and the formation of d-MPH over time was also assessed by the PAR method

Intravenous Pharmacokinetics in Rats

- In male Sprague-Dawley rats, SDX (4.75 mg/kg) and d-MPH HCl (2.39 mg/kg) were administered via the tail vein at equimolar doses corresponding to 2.06 mg/kg d-MPH
- Plasma d-MPH concentrations were assessed via serial PK sampling up to 2 hours postdose
- PK parameters included: maximal d-MPH exposure (C_{max}), overall d-MPH exposure through 2 hours (AUC_{0-2h}), and time to C_{max} (T_{max})

Intravenous Human Abuse Potential

Study Design and Subjects

- This was a Phase 1, double-blind, placebo- and active-controlled, single-dose, randomized crossover study of IV administration of SDX API compared with d-MPH API in recreational stimulant users experienced with non-oral administration of stimulants, including cocaine
- Subjects who were able to discriminate a dose of IV d-MPH API from placebo were randomized to receive the following IV treatments (one per treatment period):
 - **Treatment A**: SDX API 30 mg (equimolar to d-MPH HCl, 15 mg)
 - **Treatment B**: d-MPH HCl API 15 mg
 - **Treatment C**: Placebo
- This single-center study was conducted at Vince & Associates/AltaSciences (Overland Park, KS)
- Written informed consent was obtained and the study protocol was approved by an Institutional Review Board

Pharmacokinetic Assessments and Analyses

- Blood samples were collected for the measurement of the plasma concentrations of SDX, d-MPH, l-methylphenidate (l-MPH), and ritalinic acid up to 36 hours post-dose
- Primary PK endpoints were C_{max}, T_{max}, AUC_{0-last}, and AUC_{0-inf} (d-MPH only)

Pharmacodynamic Assessments and Statistical Analyses

- Visual analog scale (VAS) assessments were conducted at various times post-dose, including:
 - Drug Liking, Feeling High, Good Effects, and Bad Effects, assessed at 2 (Drug Liking only) and 5 minutes post-dose, and at 0.25, 0.5, 1, 1.5, 2, 3, 4, 6 and 8 hours post-dose
 - Take Drug Again and Overall Drug Liking, assessed at 12 and 24 hours post-dose
- Pharmacodynamic analyses were performed using a mixed effects Analysis of Covariance (ANCOVA) model based on the Completers Population, with LS mean differences and associated confidence intervals (CIs) calculated for each pairwise comparison between treatments
- For endpoints that were not normally distributed, treatment differences were evaluated for symmetric distribution. If symmetric, these were analyzed using a paired t-test, and if non-symmetric, the Sign test was used
- The primary and key secondary endpoints, Drug Liking E_{max} and Take Drug Again E_{max}, respectively, were analyzed using one-sided, superiority-/noninferiorty-type hypothesis tests at a significance level of α=0.05 and reported with 95% confidence intervals (CIs), with margins (δ) defined as shown below

Comparison	Null hypothesis (H_0) (No difference)	Alternative hypothesis (H_a) (Significantly different)
d-MPH API (B) to Placebo (C)	$\mu_B - \mu_C \le 15$ (δ1)	$\mu_B - \mu_C > 15$ (δ1)
d-MPH API (B) to SDX (A)	$\mu_B - \mu_A \le 10$ (δ2)	$\mu_B - \mu_A > 10$ (δ2)
SDX (A) to Placebo (C)	$\mu_A - \mu_C \ge 11$ (δ3)	$\mu_A - \mu_C < 11$ (δ3)

- Other secondary endpoints were analyzed using two-sided, confirmatory hypothesis tests (e.g., H_0: $\mu_B - \mu_C = 0$; H_A: $\mu_B - \mu_C \neq 0$) with α=0.05 and reported with 95% CIs except for comparisons of A vs. C, where α=0.1

Safety

- Incidence of adverse events (AEs), physical examination findings, vital signs, electrocardiogram (ECG) parameters, and clinical laboratory tests

RESULTS

Primary and Secondary Pharmacology

- No significant binding was noted for SDX at DAT, NET, or SERT. All IC_{50} values were found to be >10 μM
- In the secondary screen of "off-site" pharmacology, SDX (10 μM) produced no significant binding at any of the 68 molecular targets assayed

In vitro Metabolic Stability

- Low levels of d-MPH were formed in all 3 matrices, human whole blood (PAR=0.44), plasma (PAR=0.42), and liver S9 fractions (PAR=5.16) at 90 minutes (data not shown)
 - Because there is no d-MPH at Time 0, its quantitative increase relative to the decrease in SDX is difficult to estimate using the PAR method
- Overall, conversion rates of SDX to d-MPH remained low and slow as indicated by virtually unchanged SDX concentrations (**Figure 1**)
- A modest SDX disappearance (73.8% parent remaining after 90 minutes) was only observed in human whole blood with a corresponding half-life of approximately 3.6 hours (216 minutes)

Figure 1. In vitro human metabolic stability of SDX

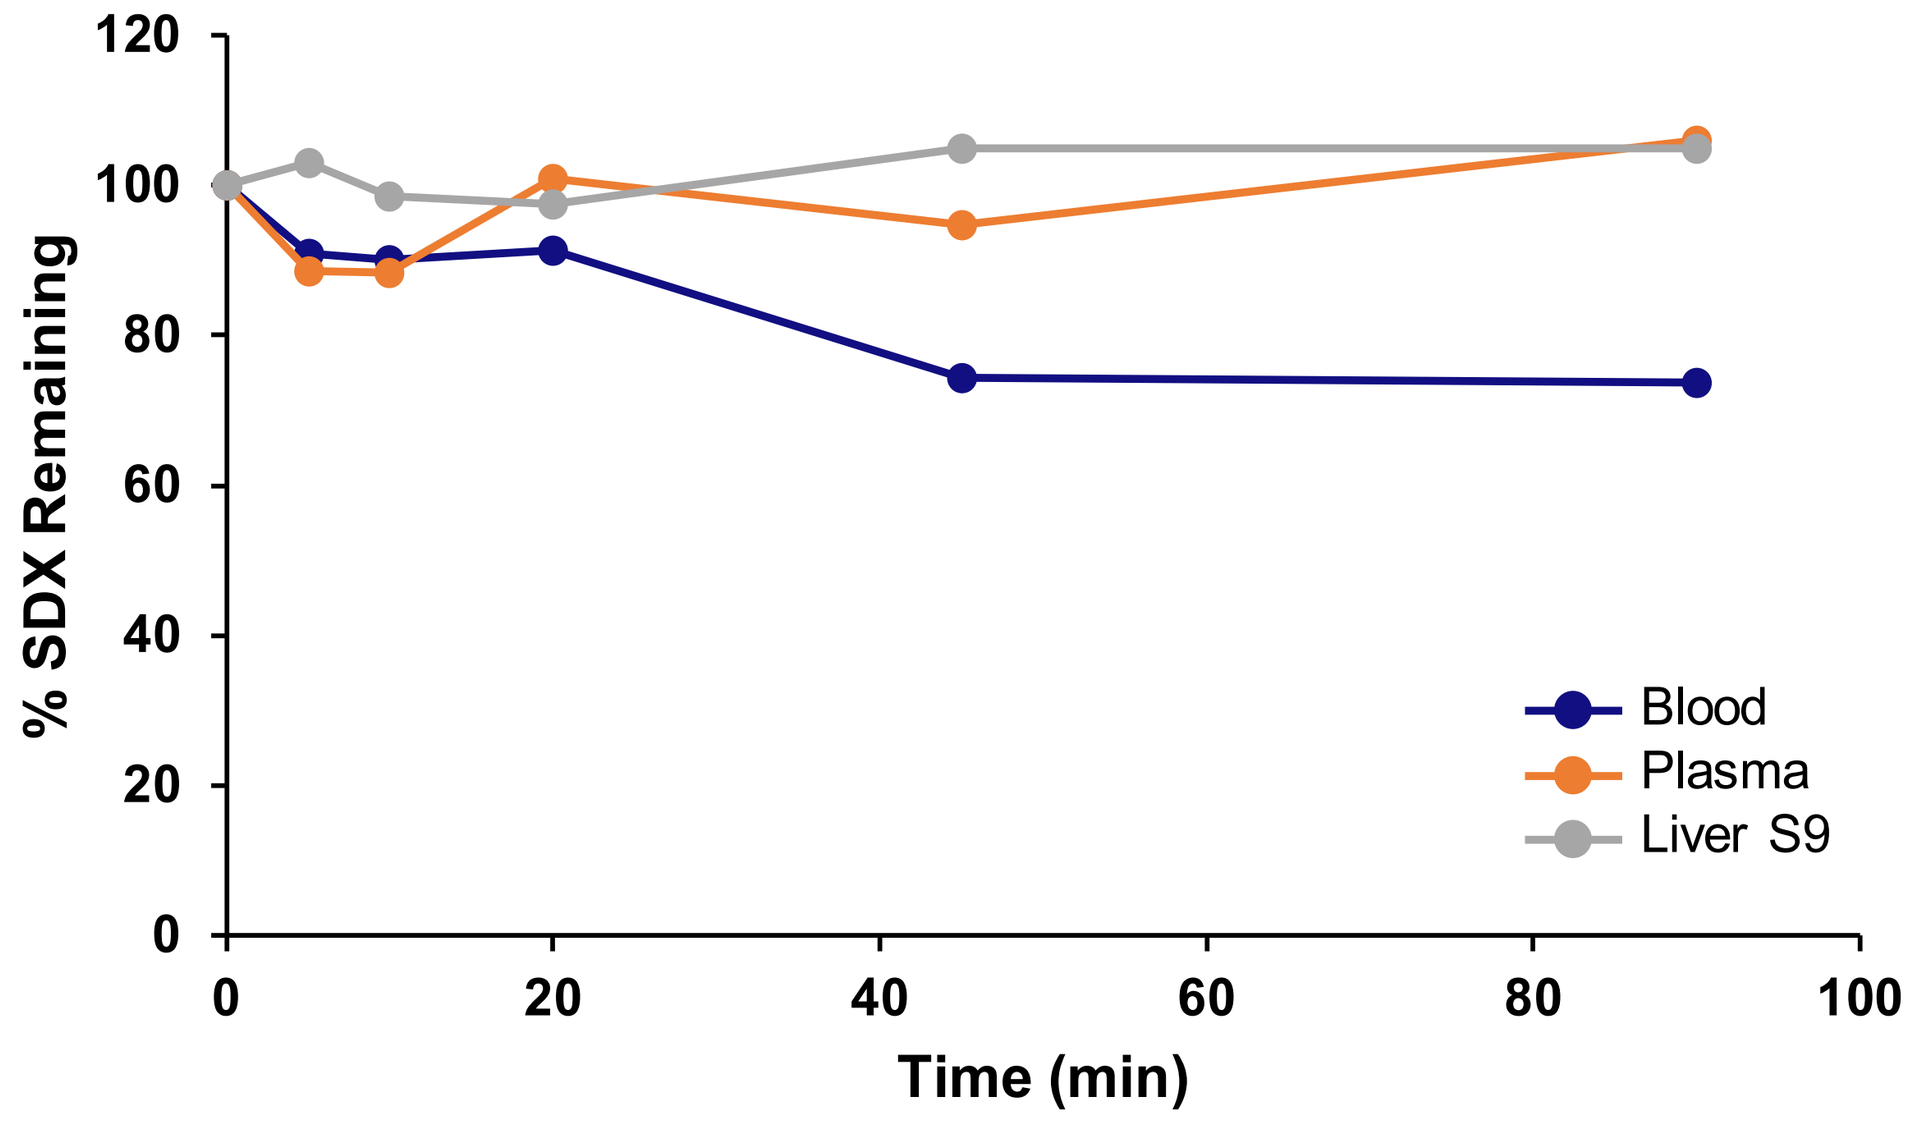

% SDX Remaining
120
100
80
60
40
20
0
0
20
40
60
80
100
Time (min)
Blood
Plasma
Liver S9

Intravenous Pharmacokinetics in Rats

- **Figure 2** demonstrates significantly lower plasma concentrations of d-MPH following IV SDX compared to IV d-MPH HCl at all timepoints
- Peak (C_{max}) and overall (AUC_{0-2h}) d-MPH exposure for SDX-derived d-MPH were 11.6% and 17.5% of the respective values observed with d-MPH HCl

Figure 2. Plasma d-MPH concentrations following IV SDX and d-MPH HCl in rats

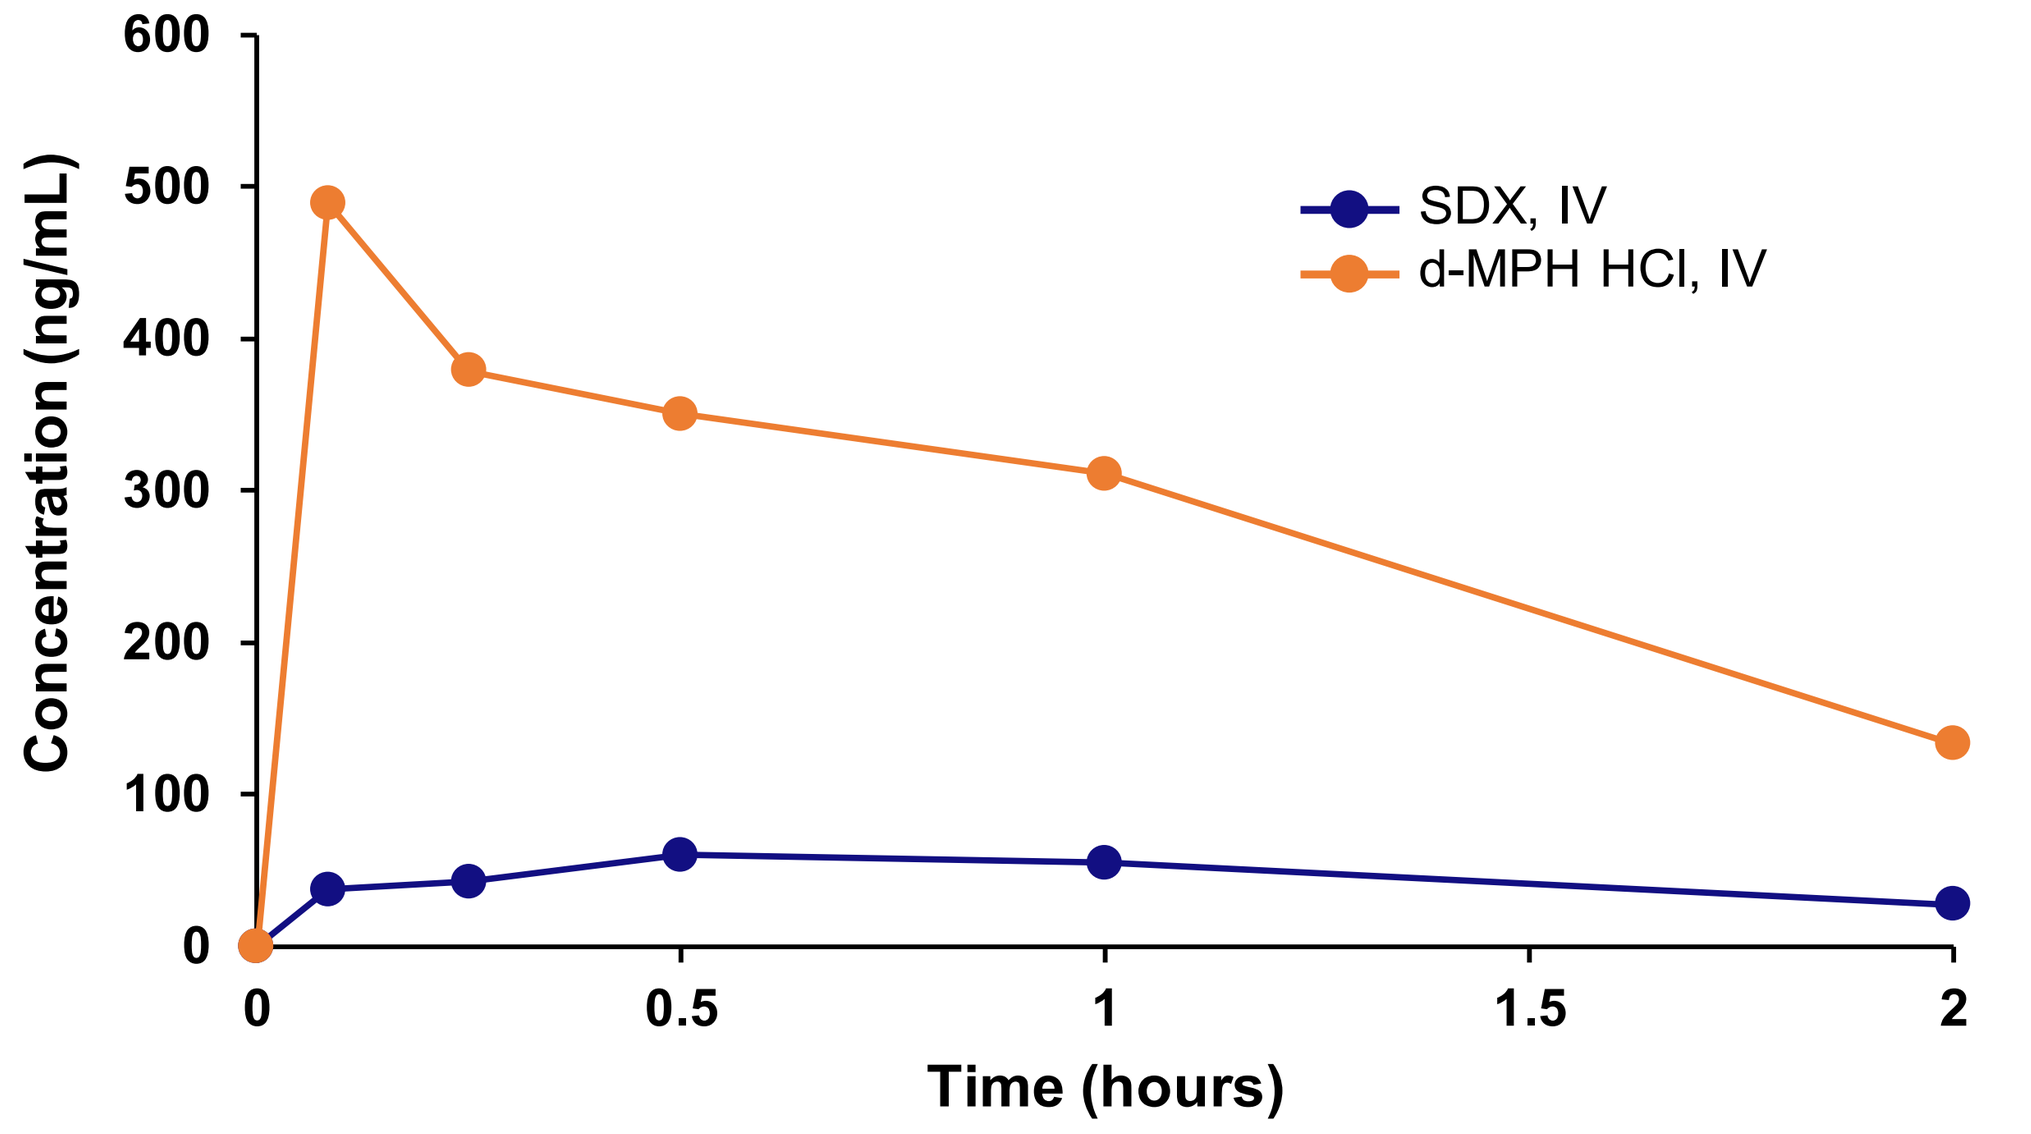

Concentration (ng/mL)
600
500
400
300
200
100
0
0
0.5
1
1.5
2
Time (hours)
SDX, IV
d-MPH HCl, IV

Human Abuse Potential

Subject Disposition and Demographics

- A total of 30 subjects (mean age = 32, 80% male) completed all 3 treatment periods and had at least one response on Drug Liking VAS within 2 hours of T_{max} for each treatment and thus comprised the Completer Population

Pharmacokinetics

- **Figure 3** shows that following IV SDX administration, peak (C_{max}) and overall (AUC_{inf}) d-MPH exposure were approximately 21.5% and 12.7%, respectively, of the exposure observed with IV d-MPH HCl

Figure 3. d-MPH concentrations following IV SDX and d-MPH HCl

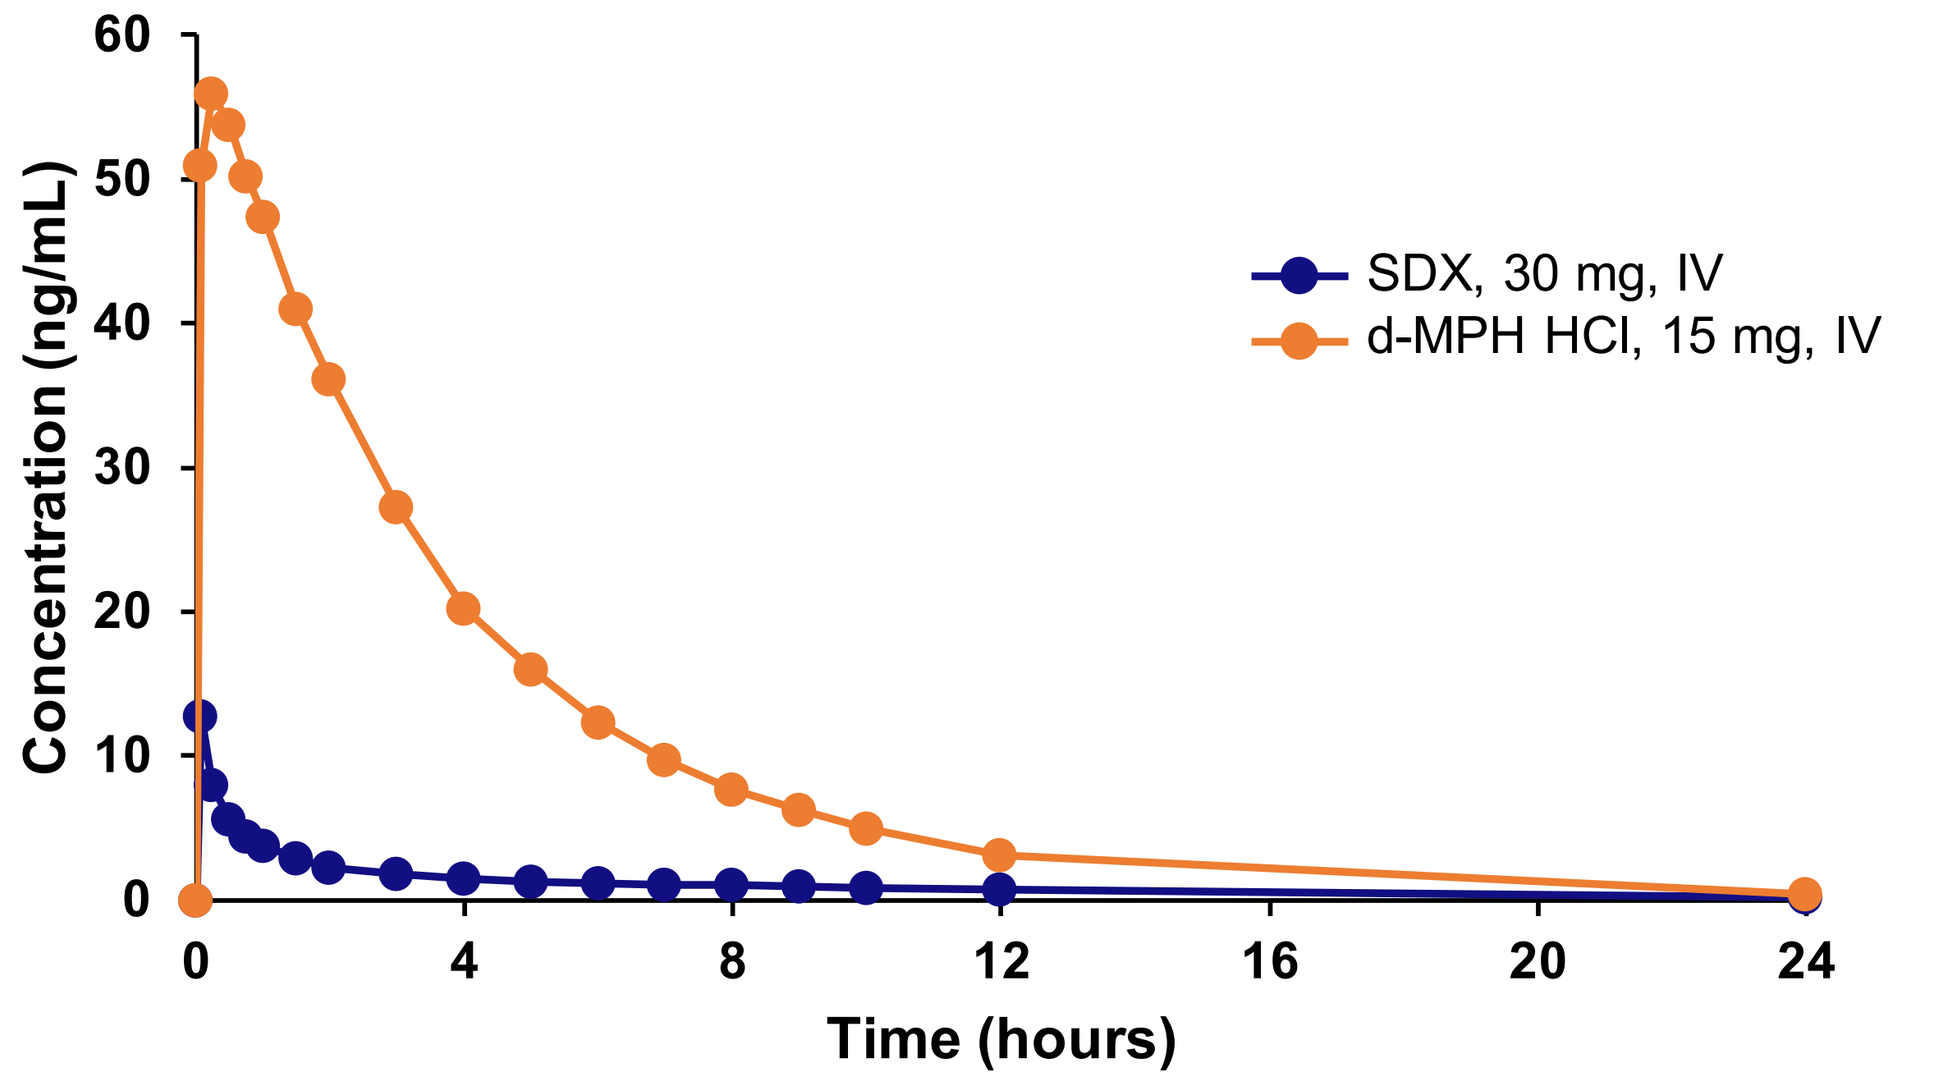

Concentration (ng/mL)
60
50
40
30
20
10
0
0
4
8
12
16
20
24
Time (hours)
SDX, 30 mg, IV
d-MPH HCl, 15 mg, IV

Pharmacodynamics

- **Figure 4** shows that d-MPH HCl, but not SDX or placebo, produced rapid and robust increases in Drug Liking that were apparent within 2 minutes of dosing
- Drug Liking E_{max} values for d-MPH HCl were significantly higher compared to placebo (LS mean difference [95% CI] = 30.5 [25.9, ∞], p<0.001), thus confirming study validity (**Figure 5**)
- Drug Liking E_{max} values for SDX were significantly lower compared to d-MPH HCl (median difference [95% CI] = 29.0 [22.5, ∞], p=0.001) and non-inferior to placebo (median difference [95% CI] = 0.5 [-∞, 5.5], p=0.001) (**Figure 5**)
- Similar treatment effects were observed for Overall Drug Liking (**Figure 5**) and Take Drug Again (key secondary endpoint), Feeling High, and Good Effects; Bad Effects were generally low for all treatments (**Figure 6**)

Figure 4. Drug Liking[a] VAS scores over time

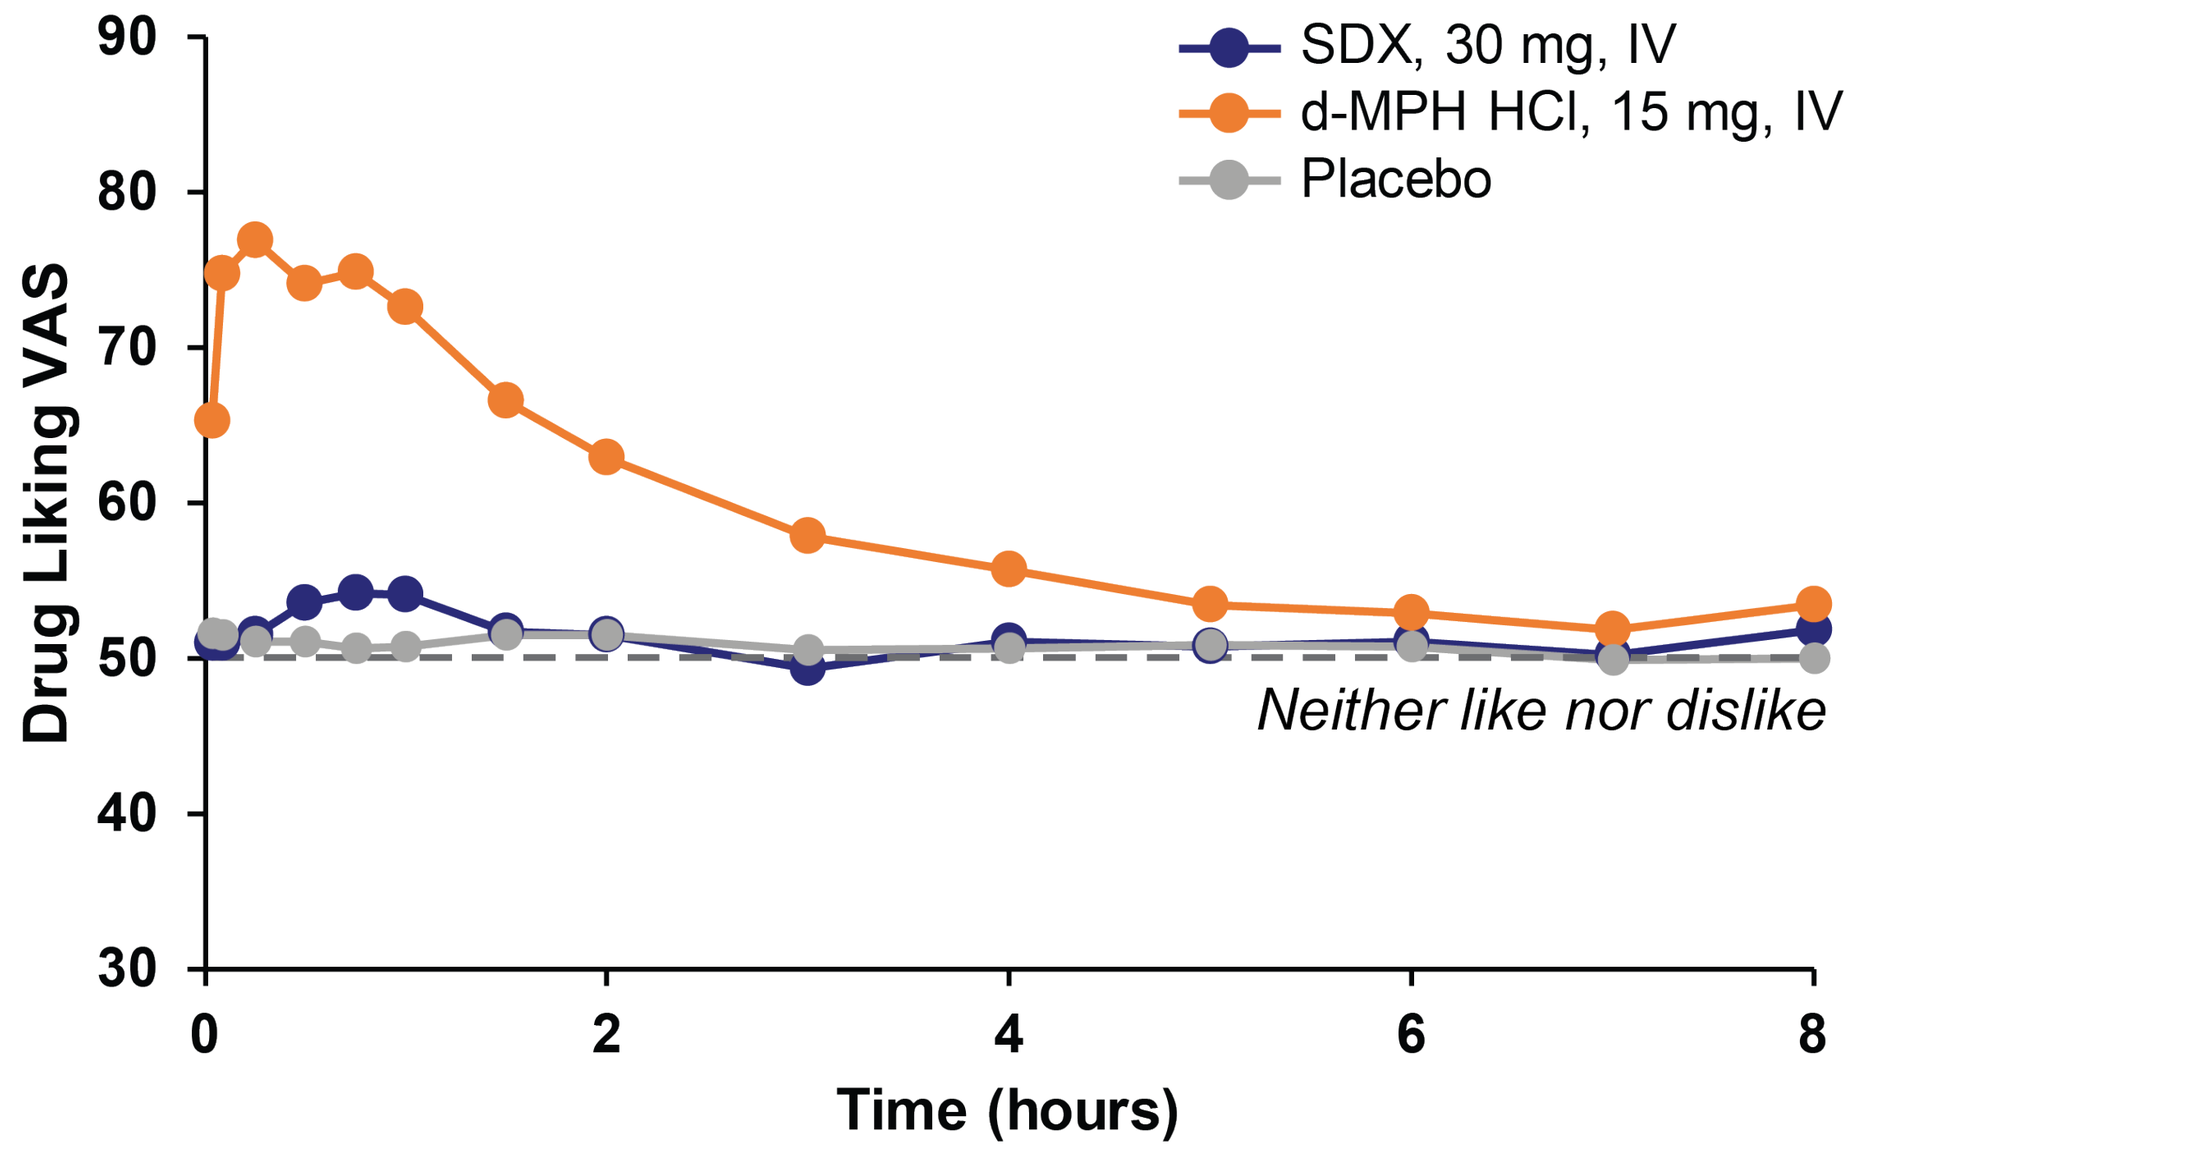

Drug Liking VAS
90
80
70
60
50
40
30
0
2
4
6
8
Time (hours)
SDX, 30 mg, IV
d-MPH HCl, 15 mg, IV
Placebo
Neither like nor dislike

[a] Subjects responded to the question: "At this moment, my liking for the drug is?", with 0=strong disliking, 50=neither like nor dislike, and 100=strong liking

Figure 5. "At the moment" Drug Liking and Overall Drug Liking VAS E_{max} scores for IV SDX, d-MPH HCl, and placebo

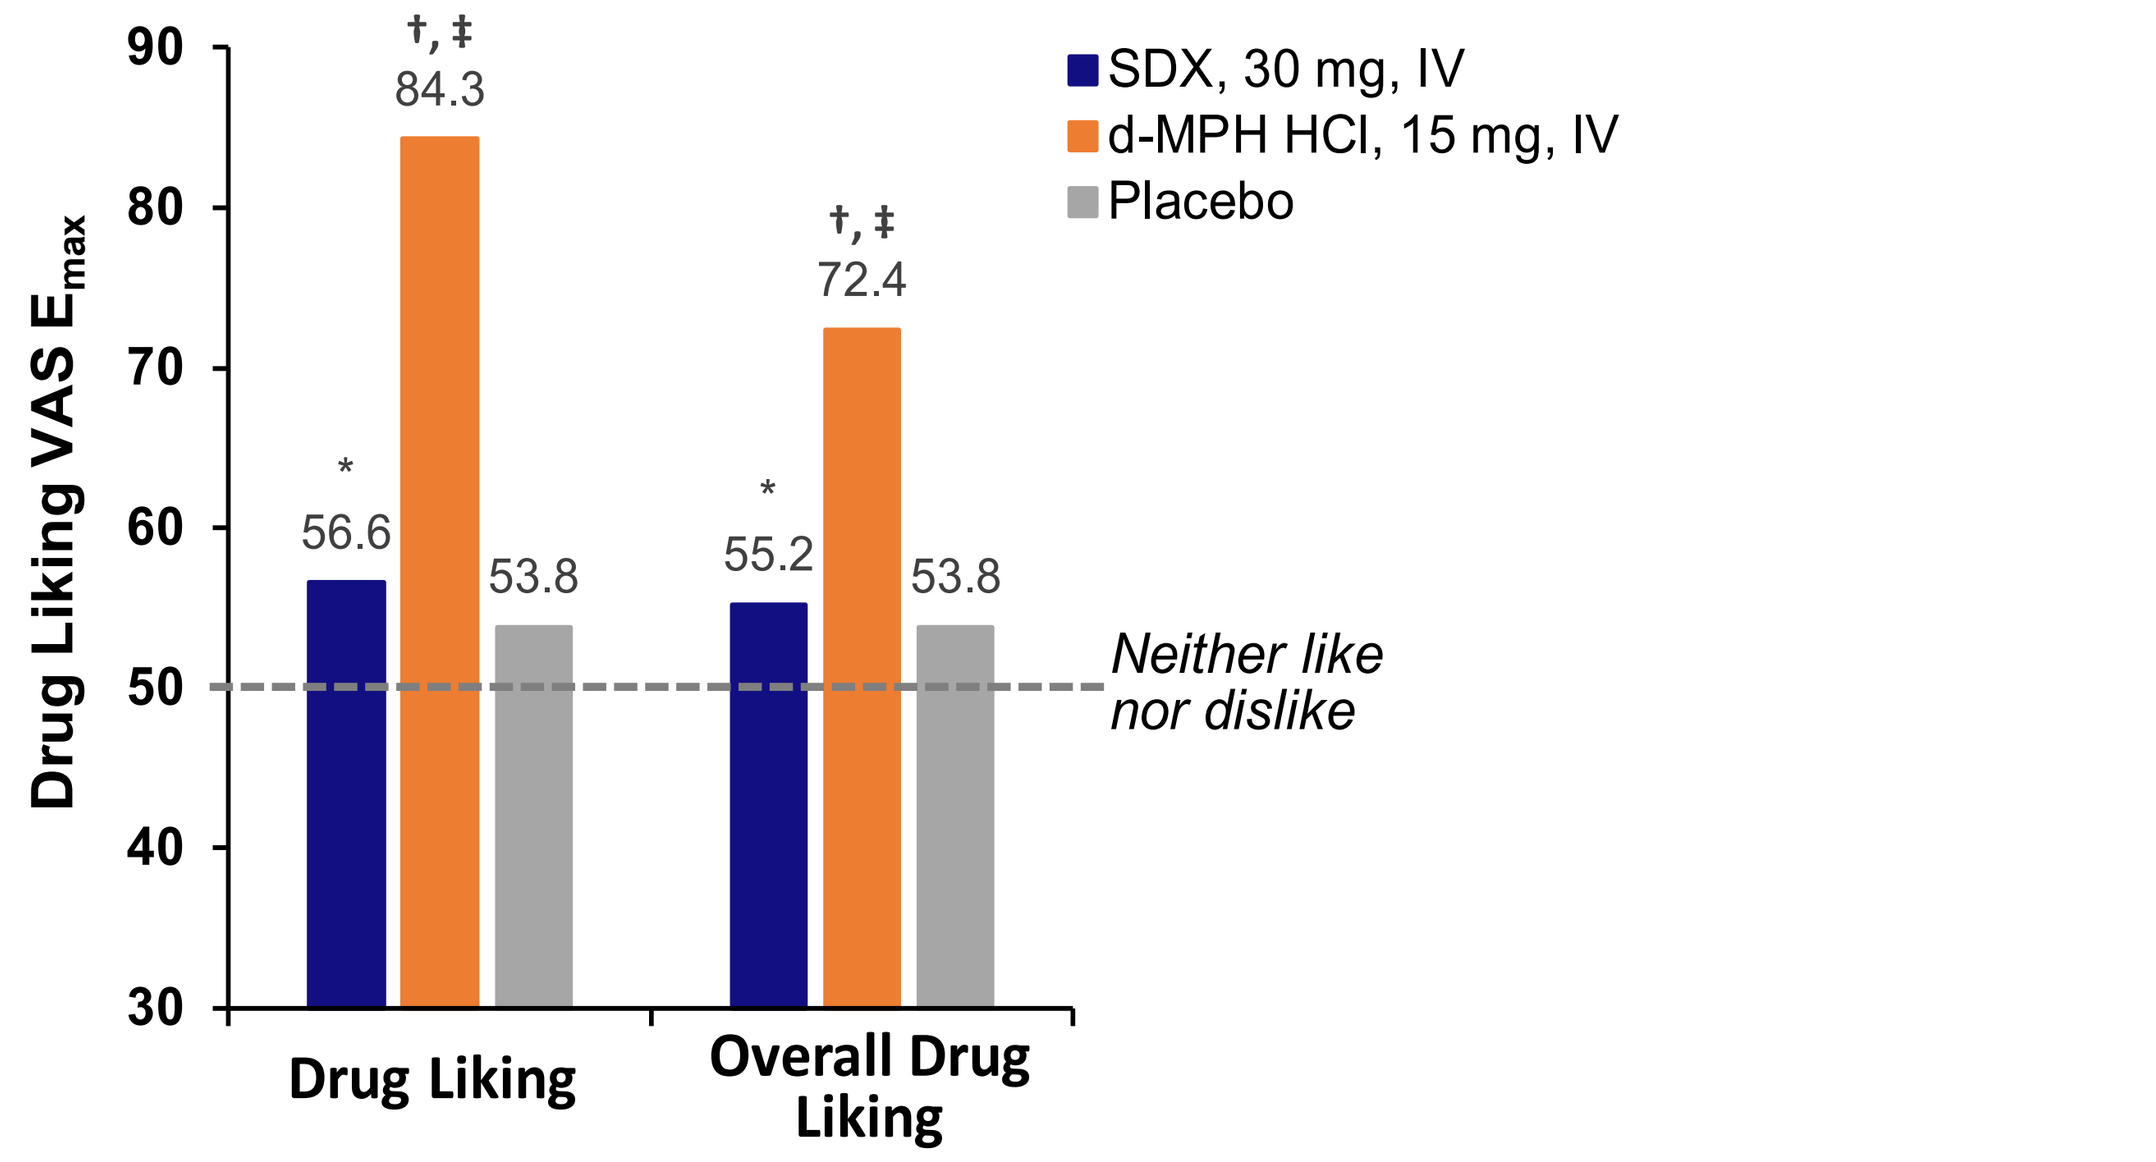

Drug Liking VAS Emax
90
80
70
60
50
40
30
†, ‡
84.3
*
56.6
53.8
†, ‡
72.4
*
55.2
53.8
Neither like nor dislike
Drug Liking
Overall Drug Liking
SDX, 30 mg, IV
d-MPH HCl, 15 mg, IV
Placebo

<u>Drug Liking</u>: * significantly non-inferior to placebo (p=0.001); † significantly higher vs. SDX (p=0.001); ‡ significantly higher vs. placebo (p<0.001). <u>Overall Drug Liking:</u> * significantly similar to placebo (p=0.658); † significantly higher vs. SDX (p=0.001); ‡ significantly higher vs. placebo (p<0.001).
Note: Assessed on a 0-100 point bipolar scale

Figure 6. E_{max} scores for Take Drug Again, Feeling High, Good Effects, and Bad Effects VAS

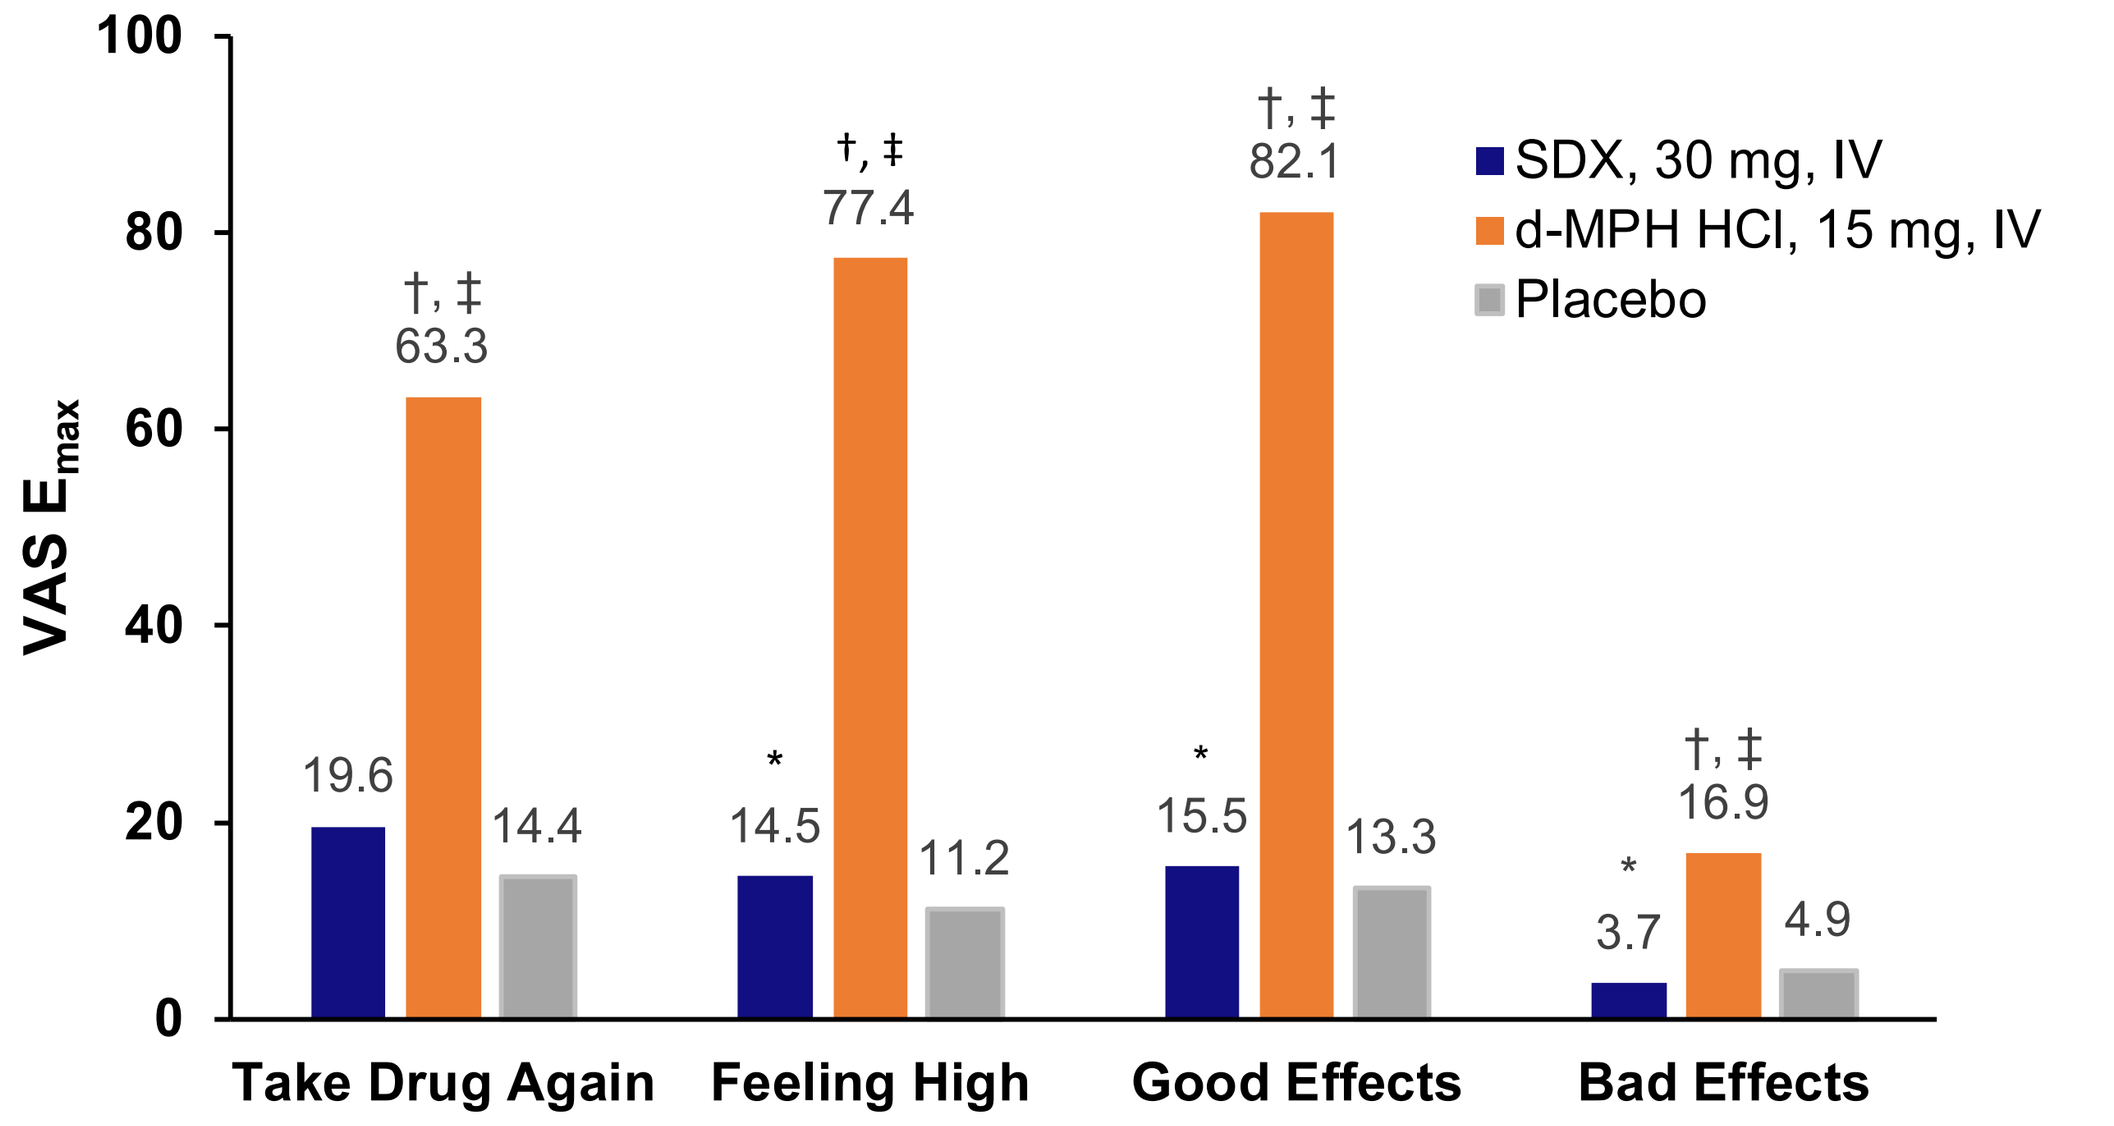

VAS Emax
100
80
60
40
20
0
19.6
†, ‡
63.3
14.4
*
14.5
†, ‡
77.4
11.2
*
15.5
†, ‡
82.1
13.3
*
3.7
†, ‡
16.9
4.9
Take Drug Again
Feeling High
Good Effects
Bad Effects
SDX, 30 mg, IV
d-MPH HCl, 15 mg, IV
Placebo

*statistically similar to placebo (p≥0.1), †significantly higher vs. SDX (p<0.05), ‡significantly higher vs. placebo (p<0.05)
Note: Assessed on a 0-100 point unipolar scale

Tolerability and Safety

- AEs typical of stimulants were more common for d-MPH HCl than SDX, most notably euphoric mood (d-MPH: 56.7% vs. SDX: 12.9%), hypervigilance (33.3% vs. 12.9%), dry mouth (20.0% vs. 0%), heart rate increased (16.7% vs. 0%), tachycardia (13.3% vs. 0%), and sinus tachycardia (13.3% vs. 0%)
- There were no clinically significant clinical laboratory values, ECG results, or out-of-range vital signs following IV SDX

CONCLUSIONS

- SDX had no appreciable pharmacological activity at monoaminergic transporters or other molecular targets when tested up to a concentration of 10μM
- SDX remained largely intact, with little conversion to d-MPH, in human whole blood, plasma, and liver S9 fractions
- IV administration of SDX to rats and humans yielded very low concentrations of d-MPH
- IV administration of SDX to recreational stimulant abusers produced pharmacodynamic effects that were comparable to placebo on multiple abuse-related endpoints, and significantly less than d-MPH HCl administered at an equimolar dose
- The performance of SDX in these studies provides converging evidence that the prodrug is unlikely to be attractive for IV abuse

Disclosures: MJS is an employee of Altreos Research Partners, Inc. SG, TCM, RG, AS, and ACB are employees and shareholders of KemPharm, Inc. This study was funded by KemPharm, Inc., Celebration, FL, USA. Design support was provided by Research Triangle Graphics, LLC.

References: 1. Braeckman R et al. Poster presentation at American Professional Society for ADHD and Related Disorders, January 12-14th, 2018, Washington, DC. **2.** Burtner J et al. Poster presentation at the APA 2017 Annual Meeting, May 20th-24th, 2017, San Diego, CA. **3.** Cassidy TA et al. J Atten Disord. 2015b;19(4):275-83.

American College or Neuropsychopharmacology (ACNP) • December 9-13, 2018 • Hollywood, FL